LUND UNIVERSITY

Early- versus Late-Onset Alzheimer’s Disease—Differences in Functional Impairment.

Wattmo, Carina; Wallin, Åsa

2016

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Wattmo, C., & Wallin, Å. (2016). *Early- versus Late-Onset Alzheimer’s Disease—Differences in Functional Impairment.*. Poster session presented at 9th Clinical Trials Conference on Alzheimer’s Disease, San Diego, CA, United States.

Total number of authors:
2

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
• Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
• You may not further distribute the material or use it for any profit-making activity or commercial gain
• You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

Download date: 14. May. 2024

Early- versus Late-Onset Alzheimer's Disease — Differences in Functional Impairment

Carina Wattmo & Åsa K. Wallin

Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, Malmö, Sweden

Conclusions

The present study highlights the clinical importance of functional evaluations for individuals with early-onset AD (EOAD). Patients in the late-onset AD (LOAD) group had significantly worse functional ability at baseline than those with EOAD; however, younger patients deteriorated faster in some individual items. Performance in instrumental activities of daily living (IADL), but not cognitive ability, predicted nursing home placement (NHP) in both groups. A similar need for NHP and survival time in nursing homes might be expected for both groups, which is important knowledge for community-based services. Among patients with EOAD, higher education or antihypertensives/cardiac therapy might predict less risk of institutionalization.

Background

Persons with clinical onset of Alzheimer's disease (AD) before 65 years of age are diagnosed with EOAD. The prevalence of EOAD is low, but varies among studies from 6% to 16%. Most individuals with EOAD are still working, have an active social life, and might have children living at home. Therefore, the consequences of being diagnosed early with a disease that implies progressive deterioration of cognitive performance and activities of daily living (ADL), and personality and behavior changes, are enormous. These individuals may also have a decreased average life expectancy of 15–18 years. Some studies suggest that EOAD might be a separate, more severe entity than LOAD. Neuropathological studies have found that younger patients exhibit a higher burden of AD pathology and a larger, more widespread cholinergic deficit than older patients. A faster cognitive progression among patients with EOAD has also been described. The clinical diagnosis of AD in younger persons can be difficult because of atypical symptoms and/or non-amnestic presentations. The present study aimed to investigate the functional outcomes in EOAD versus LOAD, and potential predictors of NHP.

Methods

The Swedish Alzheimer Treatment Study (SATS) is a 3-year, prospective, observational, multicenter study that investigated the long-term effectiveness of cholinesterase inhibitor (ChEI) treatment from various perspectives, e.g., cognition, ADL, and community-based service usage. Among the 1,258 outpatients clinically diagnosed with probable or possible AD, 1,021 had mild-to-moderate AD (Mini-Mental State Examination [MMSE] score, 10–26) at the start of ChEI therapy (baseline). Of these, 143 patients (14%) were defined as having EOAD (onset <65 years), 874 (86%) as having LOAD (onset ≥65 years), and age at onset was missing for 4; thus, 1,017 patients were enrolled in the present study. Participants were assessed for cognitive ability (MMSE) and functional capacity (IADL scale and Physical Self-Maintenance Scale [PSMS]). The NHP date was recorded if this occurred during the study. Independent-sample *t* tests were performed to compare the differences between the means obtained for two groups (Table 1 and Figure 3), and χ^2 tests were computed to analyze categorical variables (Table 1 and Figures 1 & 2). Binary logistic regression was used to determine the patient characteristics that affected NHP. Potential predictors were investigated, including: sex, apolipoprotein E ε4 carrier status, solitary living, years of education, duration of AD, age at baseline, specific concomitant medications, and cognitive and functional abilities at baseline and their rates of decline (Table 2).

Contact address:
Carina Wattmo, RN, BSc, PhD, Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, SE-205 02 Malmö, Sweden.
Tel +46 40 33 56 01, Fax +46 40 33 56 57, E mail: carina.wattmo@skane.se

Poster presented at the 9th Clinical Trials Conference on Alzheimer's Disease, San Diego, CA, USA; December 8-10, 2016.

Results

Table 1. Sociodemographic and clinical characteristics at baseline

	Early-onset AD (*n* = 143)	Late-onset AD (*n* = 874)	*p*
Female sex	57%	65%	0.091
APOE genotype			**<0.001**
No ε4 alleles	25%	33%	
One ε4 allele	46%	54%	
Two ε4 alleles	29%	13%	
Solitary living	21%	37%	**<0.001**
Antihypertensives/cardiac therapy	20%	44%	**<0.001**
NSAIDs/acetylsalicylic acid	10%	33%	**<0.001**
Anxiolytics/sedatives/hypnotics	4%	16%	**<0.001**
Estimated age at onset of AD, years[a]	58.6 ± 4.7	74.4 ± 4.9	**<0.001**
Estimated duration of AD, years[a]	4.1 ± 3.4	2.9 ± 1.7	**<0.001**
Age, years[a]	62.7 ± 5.4	77.3 ± 4.7	**<0.001**
Education, years[a]	10.1 ± 2.8	9.3 ± 2.5	**0.004**
MMSE score, range 0–30[a]	21.4 ± 3.8	21.4 ± 3.7	0.987
IADL score, range 8–31[a]	13.9 ± 5.3	16.3 ± 5.4	**<0.001**
PSMS score, range 6–30[a]	6.7 ± 1.2	7.6 ± 2.4	**<0.001**
Number of concomitant medications[a]	1.8 ± 1.7	3.1 ± 2.5	**<0.001**

[a]Mean ± standard deviation (SD)

Usage of antidiabetics (5%), asthma medication (4%), thyroid therapy (8%), lipid-lowering agents (12%), estrogens (7%), antidepressants (25%) and antipsychotics (4%) was similar between the groups.

AD, Alzheimer's disease; APOE, apolipoprotein E; IADL, Instrumental Activities of Daily Living scale; MMSE, Mini-Mental State Examination; NSAIDs, nonsteroidal anti-inflammatory drugs; PSMS, Physical Self-Maintenance Scale

Table 2. Binary logistic regression analysis of nursing home placement during the study according to age at onset group

	Early-onset AD		Late-onset AD	
Percent correctly classified	87.2%		77.2%	
	OR (95% CI)	***p***	**OR (95% CI)**	***p***
Solitary living (no = 0, yes = 1)	8.24 (2.19–31.04)	0.002	2.75 (1.93–3.93)	<0.001
Antihypertensives/cardiac therapy (no = 0, yes = 1)	0.08 (0.01–0.95)	0.045		ns
Education (years)	0.73 (0.56–0.93)	0.013		ns
IADL score at baseline	1.27 (1.11–1.45)	<0.001	1.12 (1.08–1.16)	<0.001
IADL score, rate of change per month	0.01 (0.0005–0.14)	0.001	0.28 (0.15–0.51)	<0.001

Sex, number of APOE ε4 alleles, age at baseline, duration of AD, other concomitant medications, MMSE and PSMS scores at baseline or their rates of change per month were not significant in the multivariate models.

AD, Alzheimer's disease; APOE, apolipoprotein E; CI, confidence interval; IADL, Instrumental Activities of Daily Living scale; MMSE, Mini-Mental State Examination; ns, not significant; OR, odds ratio; PSMS, Physical Self-Maintenance Scale

Figure 1.

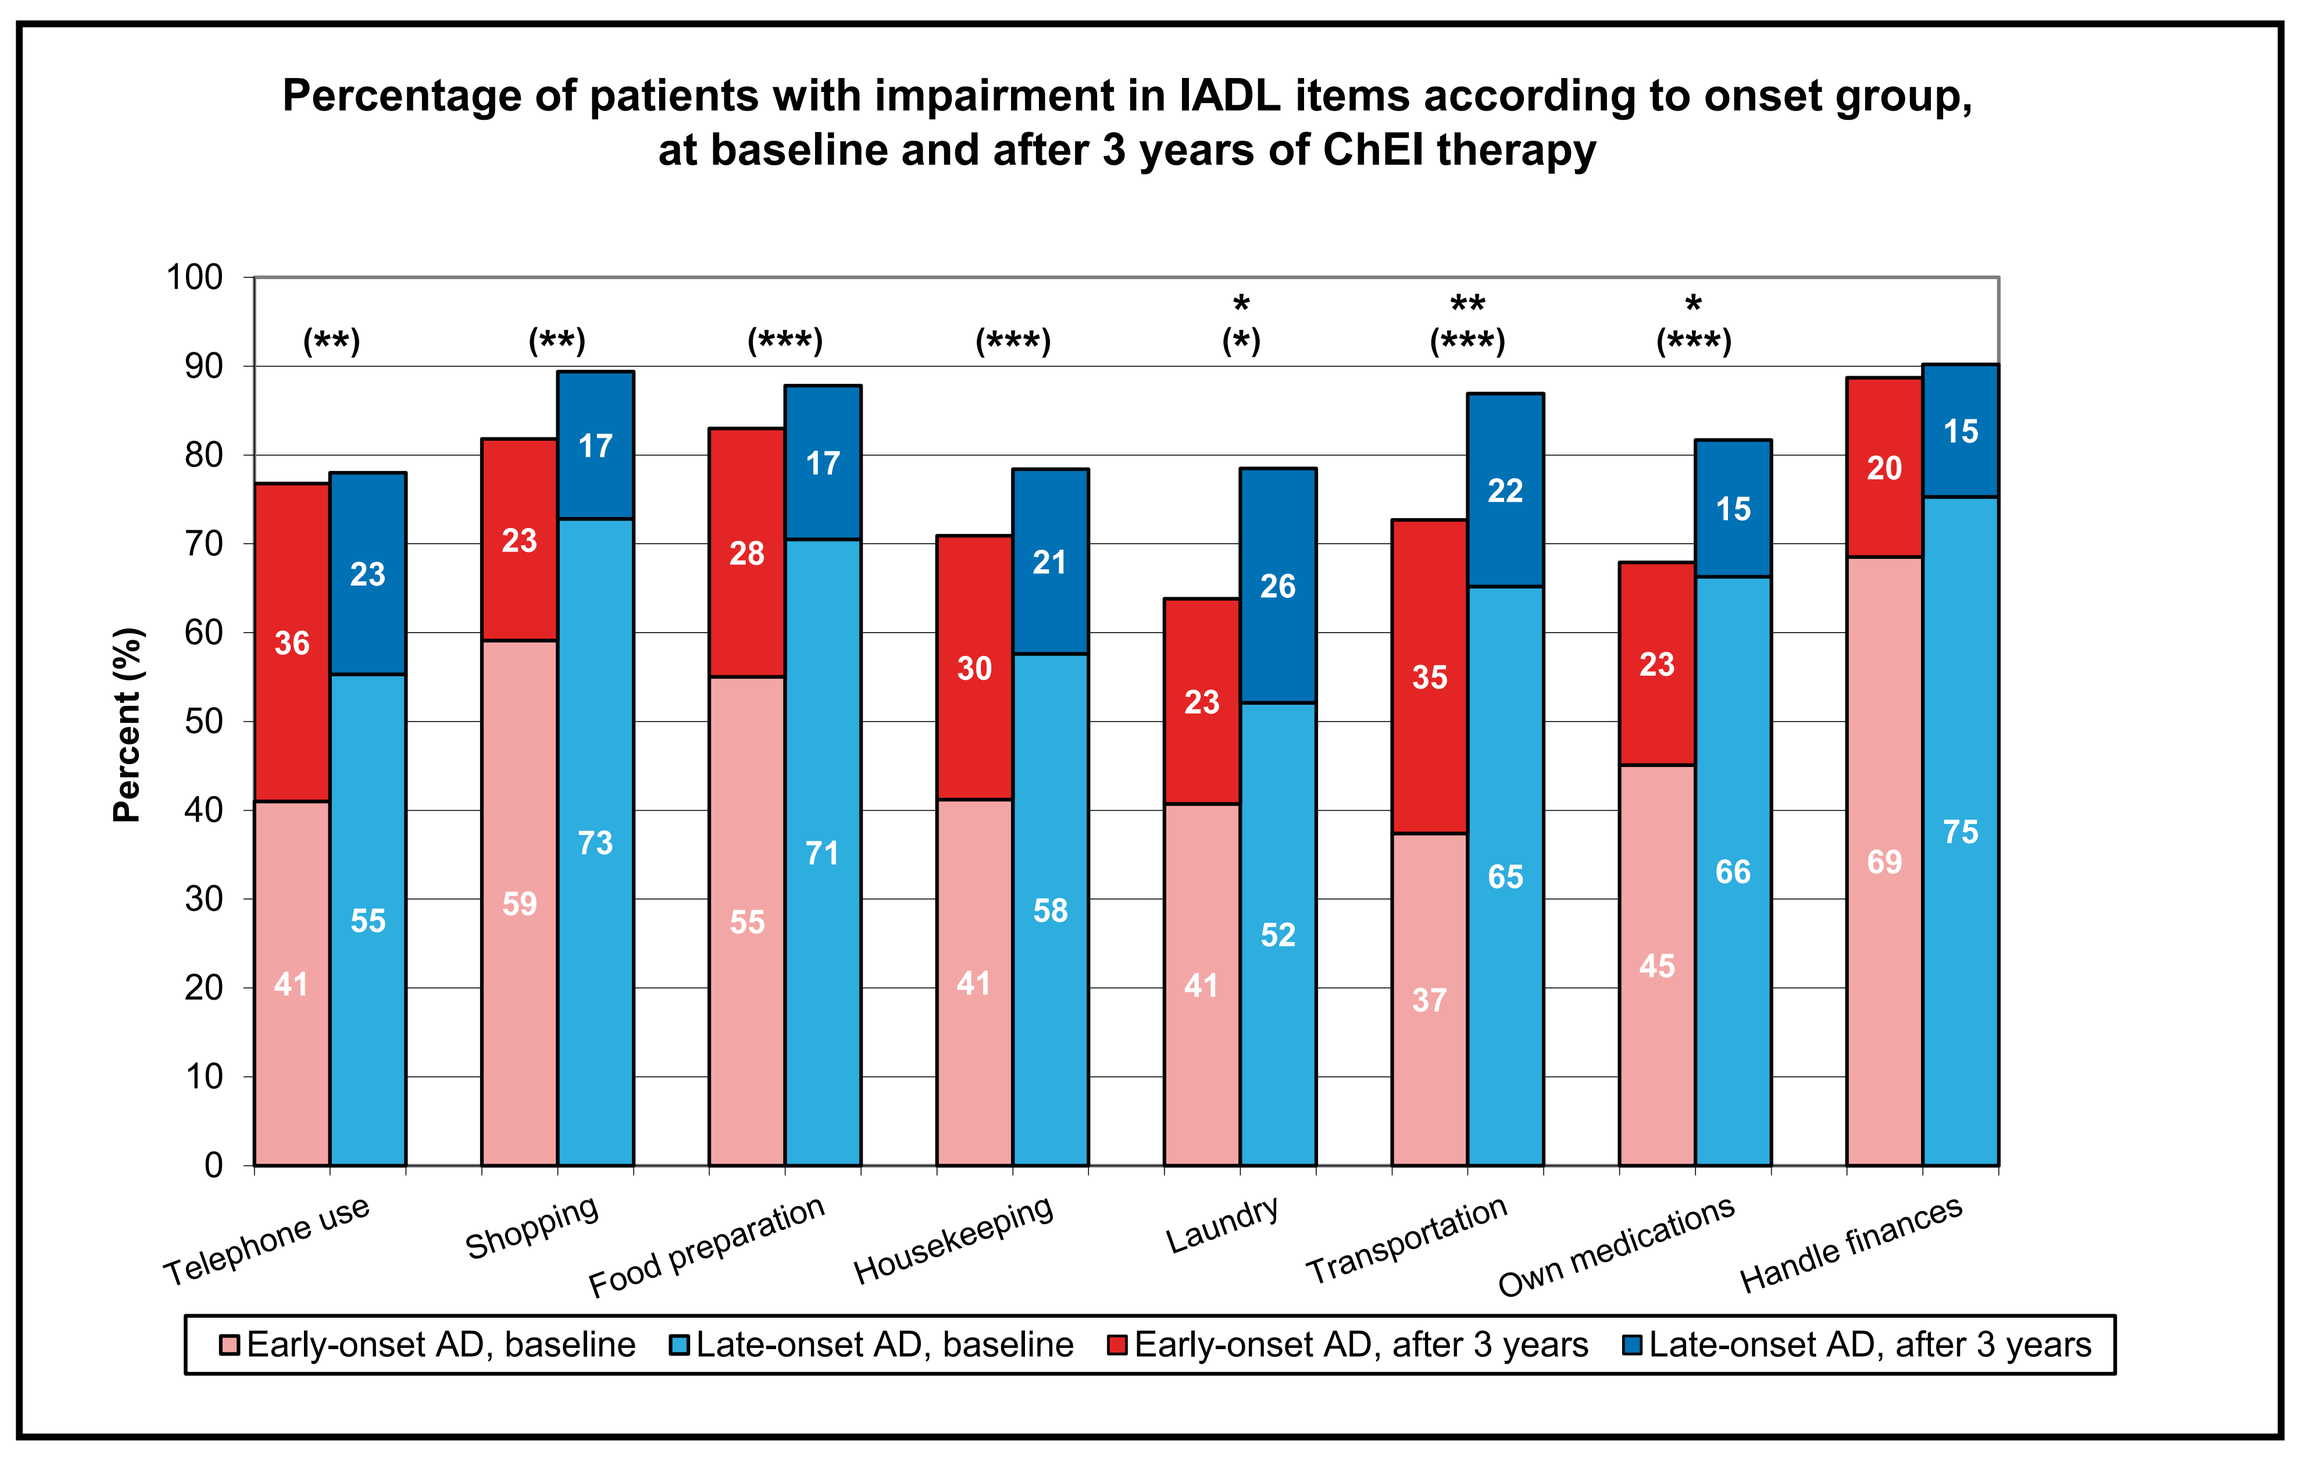

Percentage of patients with impairment in IADL items according to onset group, at baseline and after 3 years of ChEI therapy
Percent (%)
Telephone use
Shopping
Food preparation
Housekeeping
Laundry
Transportation
Own medications
Handle finances
41 55 36 23 (**)
59 73 23 17 (**)
55 71 28 17 (***)
41 58 30 21 (***)
41 52 23 26 * (*)
37 65 35 22 ** (***)
45 66 23 15 * (***)
69 75 20 15
Early-onset AD, baseline
Late-onset AD, baseline
Early-onset AD, after 3 years
Late-onset AD, after 3 years

The IADL capacity was already markedly impaired at the start of ChEI therapy (baseline); about 40–65% of the EOAD and 55–75% of the LOAD patients were dependent on assistance to perform these activities (IADL score, 2–5). The percentage of participants with impairment in the individual IADL items was significantly lower at baseline in the EOAD cohort, except for the "ability to handle finances" task. After 3 years, the IADL performance had deteriorated further; 70–90% of the remaining individuals in the SATS could not carry out these tasks independently. A larger proportion of the LOAD patients were impaired in the IADL items "laundry", "mode of transportation", and "responsibility for own medications" compared with the EOAD group (*** $p < 0.001$, ** $0.001 \leq p < 0.01$, * $p < 0.05$; baseline presented within parentheses in the figure).

Figure 2.

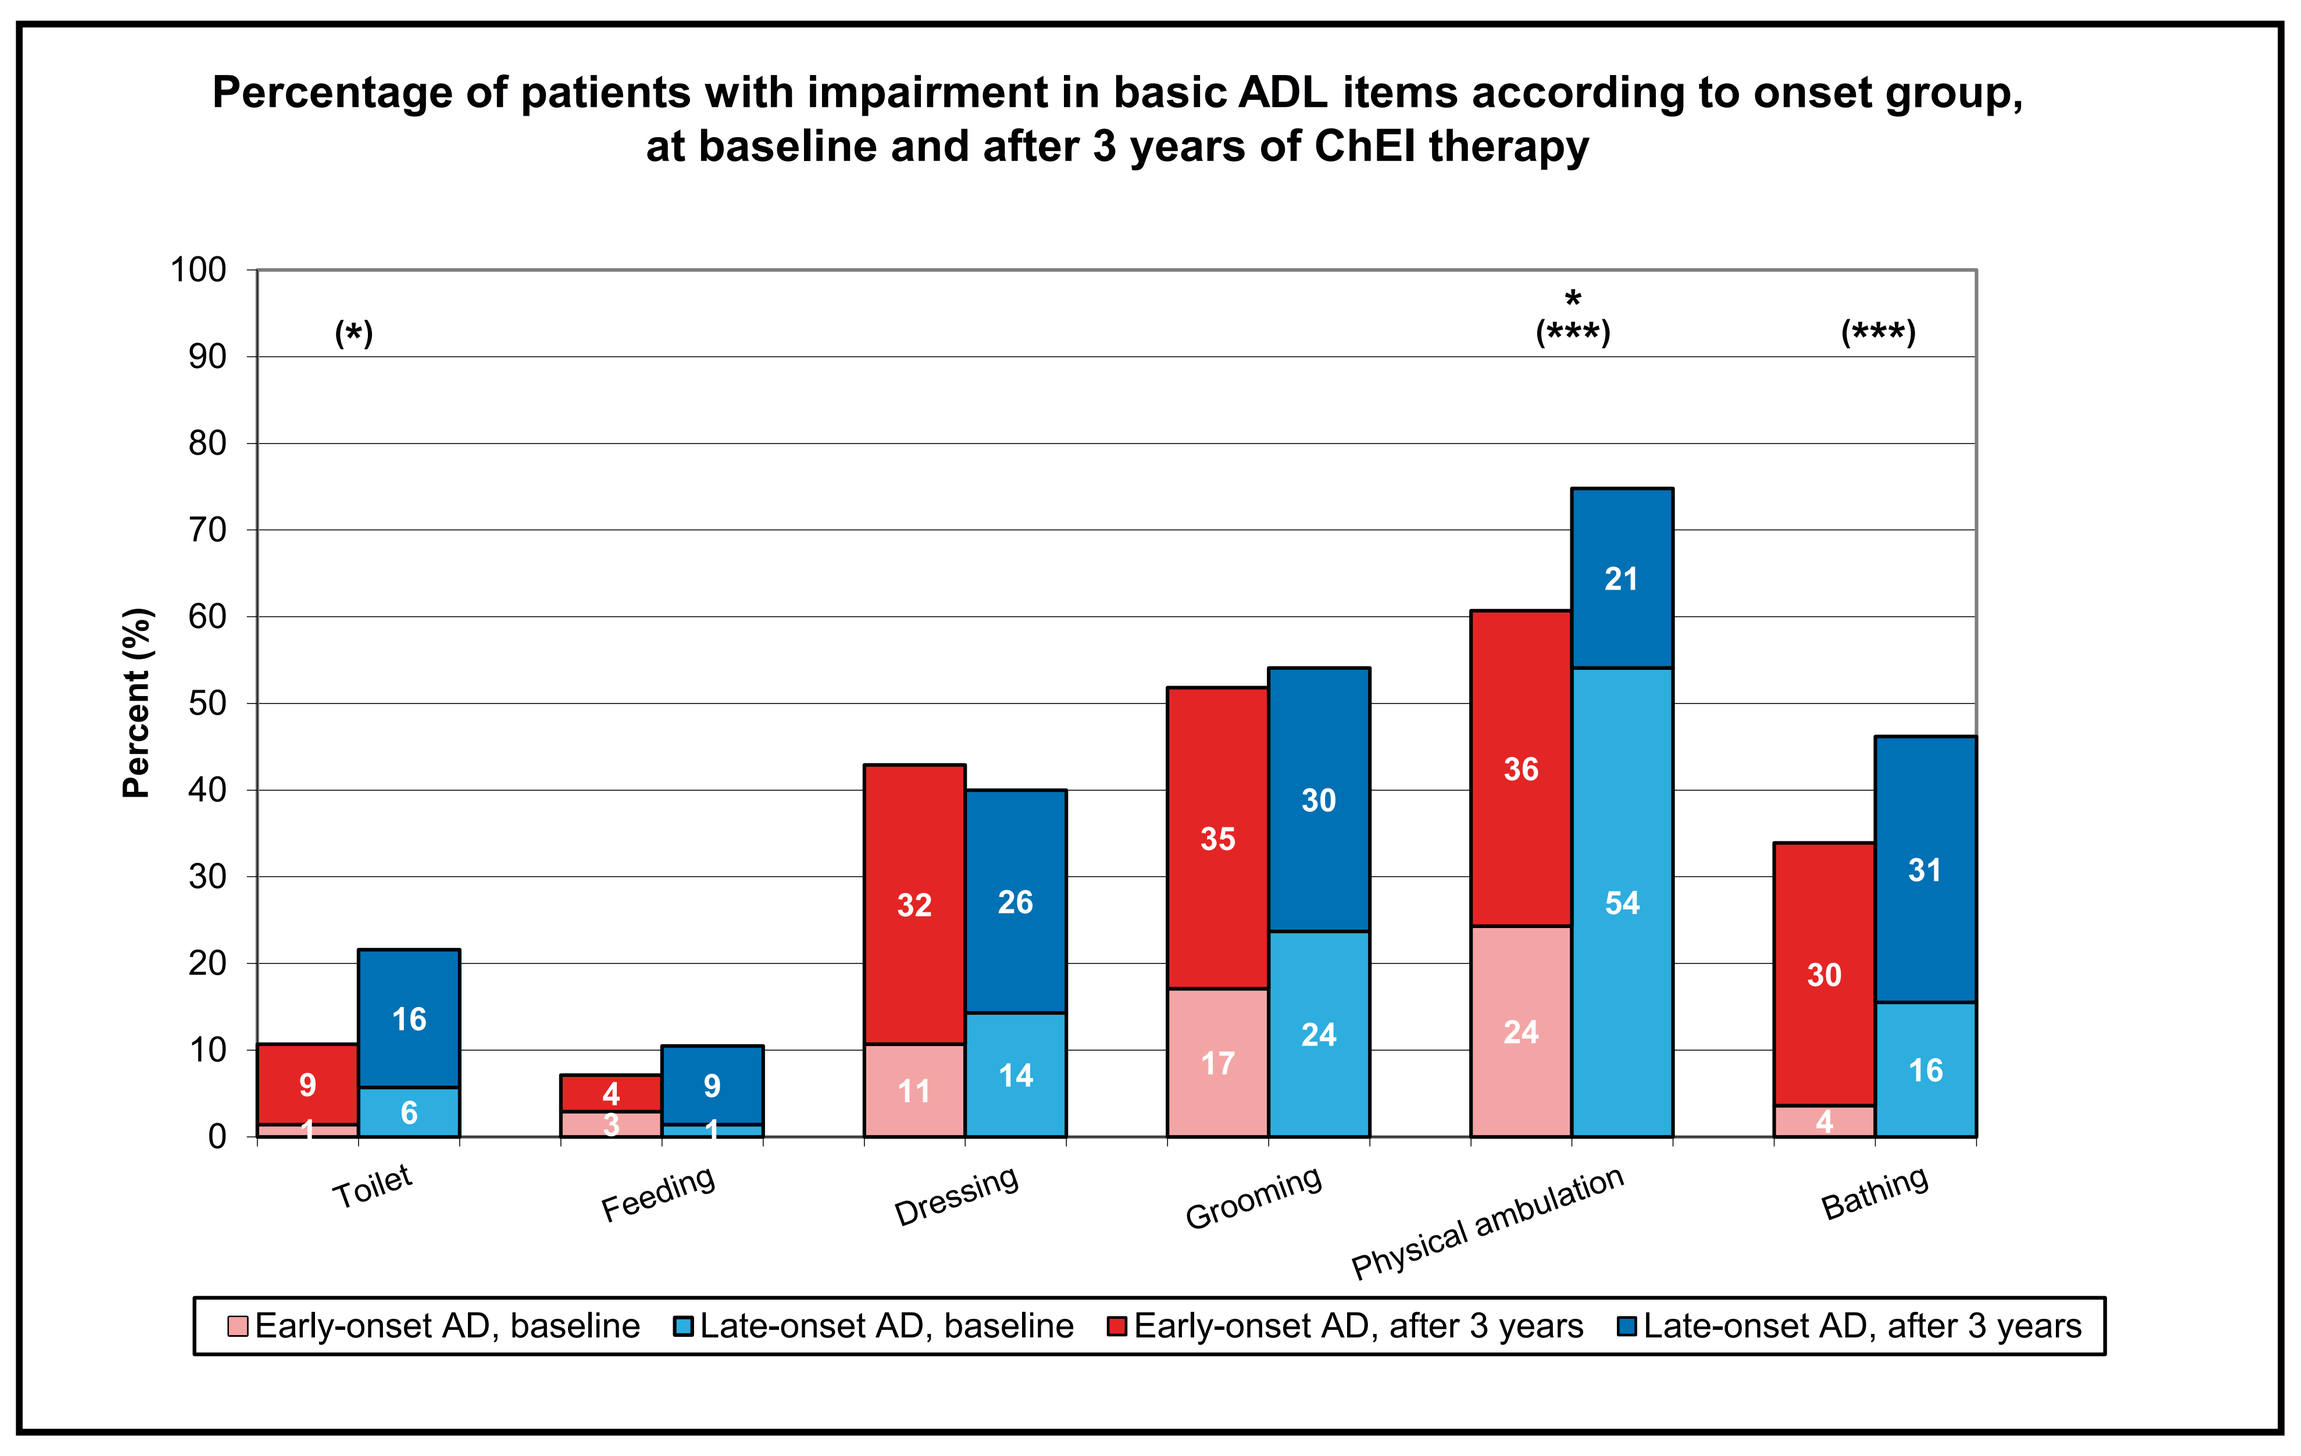

Percentage of patients with impairment in basic ADL items according to onset group, at baseline and after 3 years of ChEI therapy
Percent (%)
Toilet
Feeding
Dressing
Grooming
Physical ambulation
Bathing
1 6 9 16 (*)
3 1 4 9
11 14 32 26
17 24 35 30
24 54 36 21 * (***)
4 16 30 31 (***)
Early-onset AD, baseline
Late-onset AD, baseline
Early-onset AD, after 3 years
Late-onset AD, after 3 years

Regarding basic ADL, the majority of participants were able to manage themselves independently at baseline, with the exception of physical ambulation (more than 50% of the individuals with LOAD needed some assistance; PSMS score, 2–5). A significantly larger percentage of the LOAD patients were impaired in the ADL items "toilet", "physical ambulation", and "bathing". After 3 years, 35–55% of the remaining individuals needed assistance in performing the basic ADL items "dressing", "grooming", and "bathing" (*** $p < 0.001$, ** $0.001 \leq p < 0.01$, * $p < 0.05$; baseline presented within parentheses in the figure).

Figure 3.

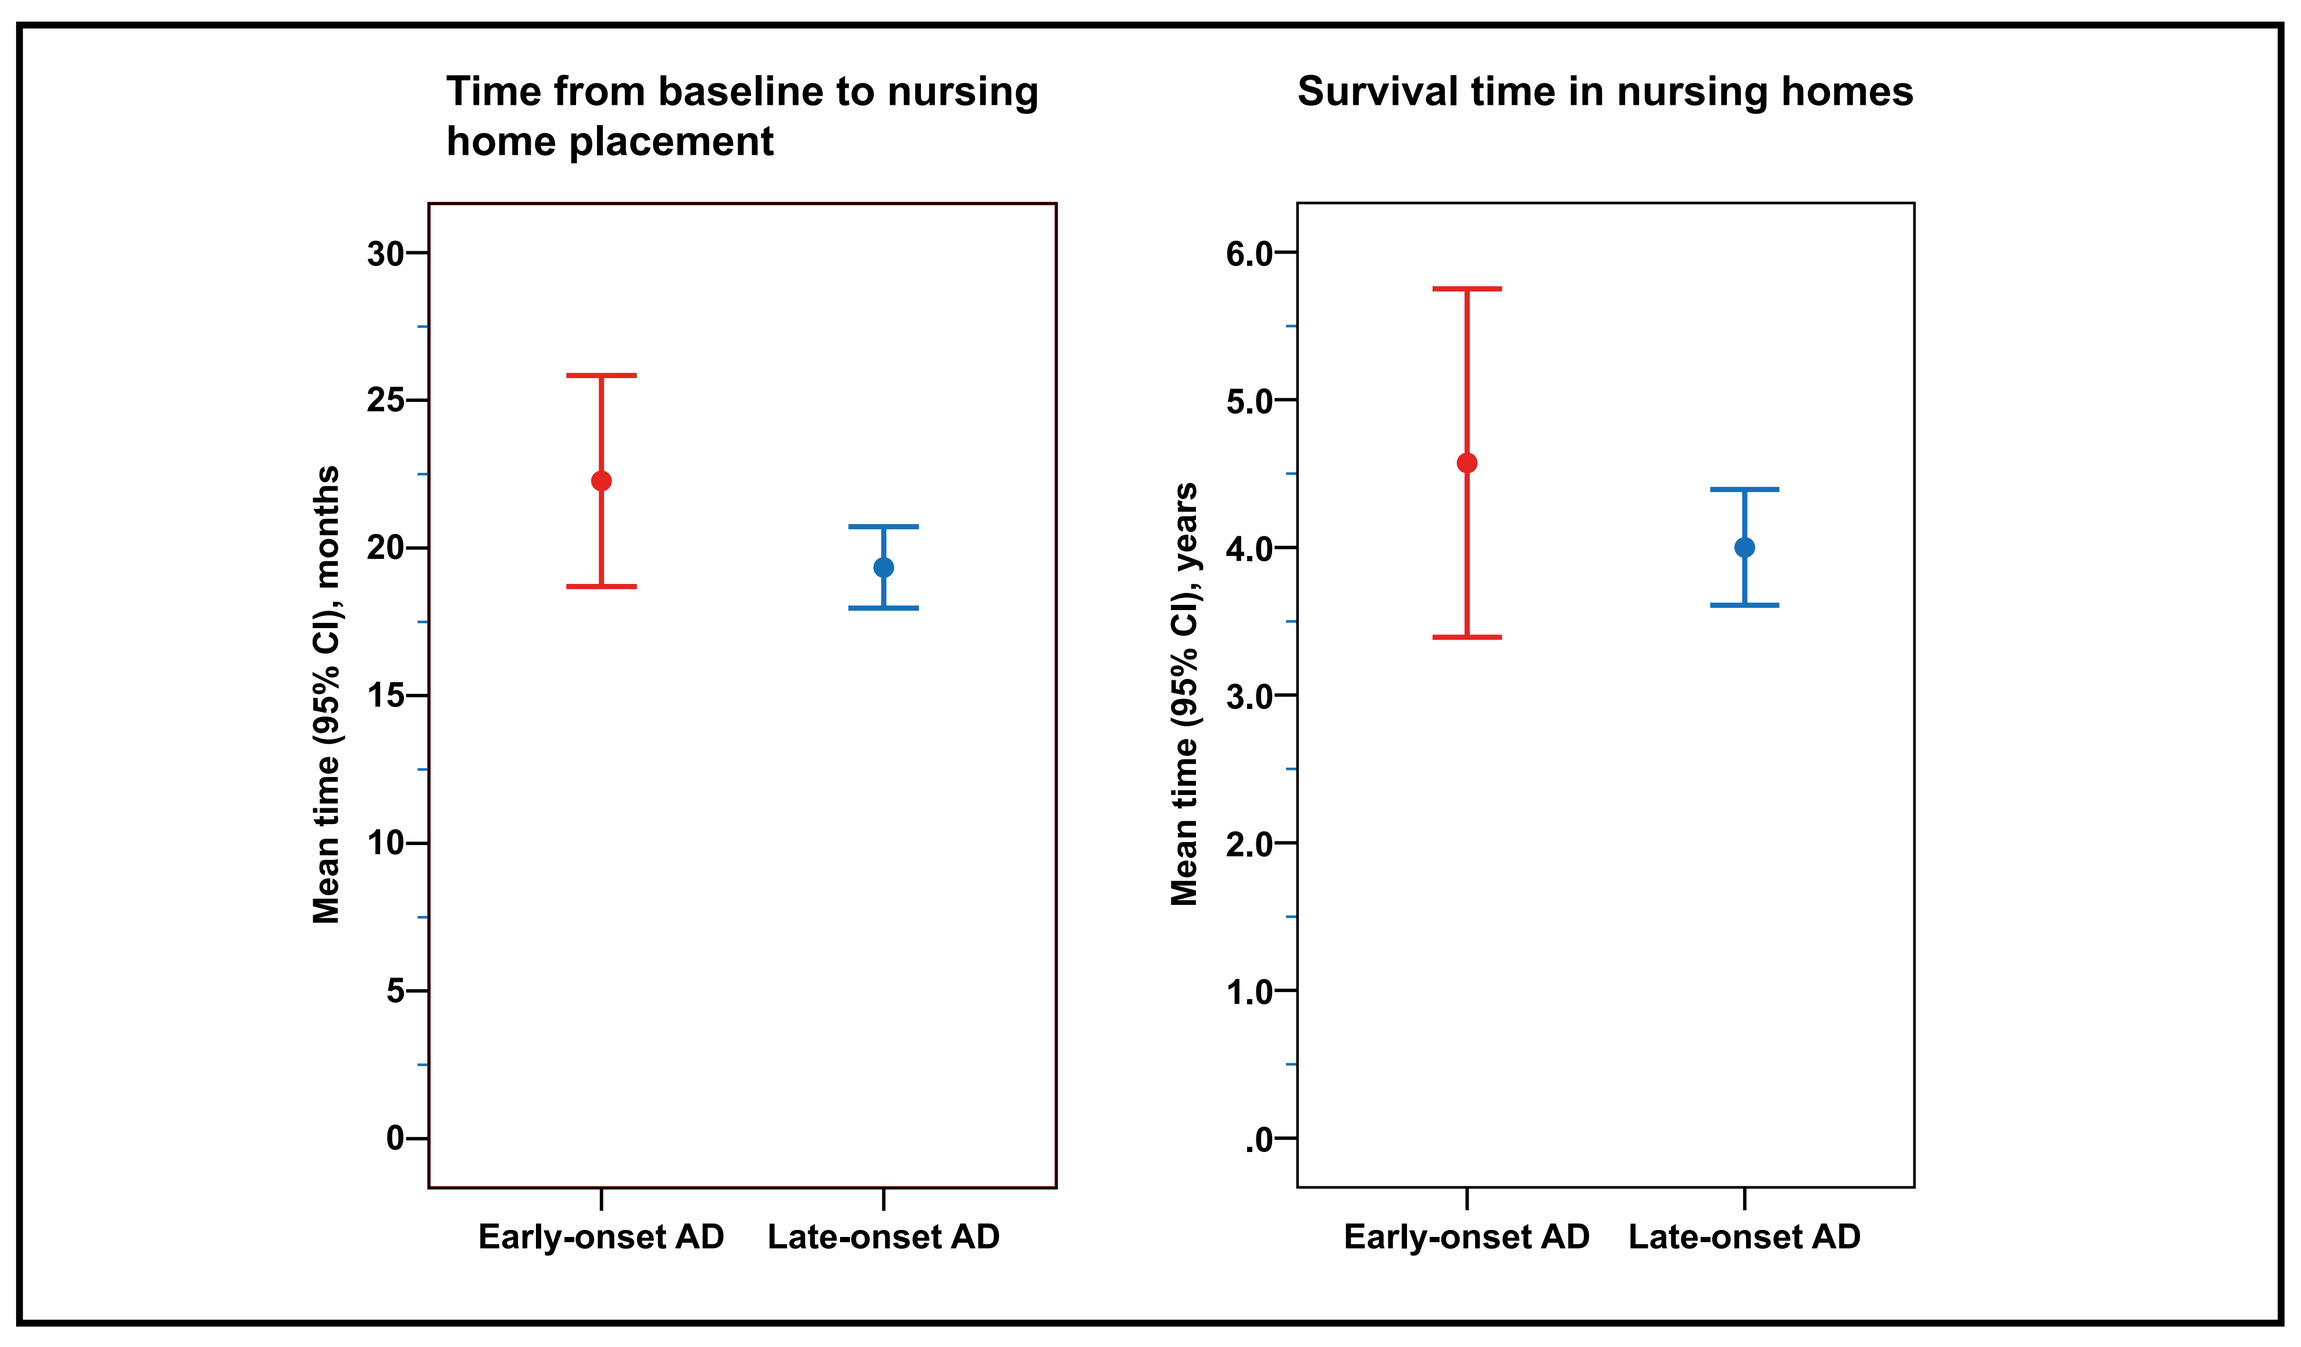

Time from baseline to nursing home placement
Mean time (95% CI), months
Survival time in nursing homes
Mean time (95% CI), years
Early-onset AD
Late-onset AD

During the SATS, 26 participants (18%) with EOAD and 205 (23%) with LOAD ($p = 0.196$) were admitted to nursing homes. The mean time from the start of ChEI treatment to institutionalization for patients with EOAD and LOAD was 22.3 (18.7–25.8) vs. 19.3 (18.0–20.7) months ($p = 0.156$), and the survival time in nursing homes was 4.6 (3.4–5.8) vs. 4.0 (3.6–4.4) years ($p = 0.352$), which were similar between the two groups.

Layout by Aletia Design, www.aletiadesign.com