LUND UNIVERSITY

Levels of cerebrospinal fluid biomarkers total tau and phosphorylated tau do not predict survival time after diagnosis of Alzheimer's disease – An 18-year follow-up.

Wattmo, Carina; Blennow, Kaj; Minthon, Lennart; Hansson, Oskar

2017

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Wattmo, C., Blennow, K., Minthon, L., & Hansson, O. (2017). *Levels of cerebrospinal fluid biomarkers total tau and phosphorylated tau do not predict survival time after diagnosis of Alzheimer's disease – An 18-year follow-up.*. Poster session presented at 10th Clinical Trials Conference on Alzheimer's Disease, Boston, Massachusetts, United States.

Total number of authors:
4

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
- Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
- You may not further distribute the material or use it for any profit-making activity or commercial gain
- You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

Levels of cerebrospinal fluid biomarkers total tau and phosphorylated tau do not predict survival time after diagnosis of Alzheimer's disease – An 18-year follow-up

Carina Wattmo[1], Kaj Blennow[2], Lennart Minthon[1], Oskar Hansson[1]

[1]Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, Malmö, Sweden.
[2]Institute of Neuroscience and Physiology, Department of Psychiatry and Neurochemistry, the Sahlgrenska Academy, University of Gothenburg, Mölndal, Sweden.

Conclusions

Mortality in Alzheimer's disease (AD) is complex and depends on many factors e.g., demographic and clinical. In this clinical-practice-based long-term study, almost half of the participants with AD had normal levels of cerebrospinal fluid (CSF) tau. We found no clear results that the levels of total tau (T-tau) and/or phosphorylated tau (P-tau) affect survival after diagnosis in AD. This observation does not support the theory that these patients have a more advanced disease. However, the individuals with pathological levels of tau had fewer years of education and worse cognitive status indicating a lower cognitive reserve capacity, which might influence life expectancy. These findings might be useful when considering new diagnostic criteria and when interpreting outcomes from future clinical trials of potentially disease-modifying AD therapies.

Background

The pathological process in AD probably starts decades before the onset of symptoms and the clinical AD diagnosis. In patients with AD, the level of CSF amyloid-β_{1-42} ($A\beta_{42}$) is usually lower, and the levels of T-tau and P-tau higher than in healthy elderly people. However, the cutoffs differ between studies and the predictive values are too low to diagnose AD using only CSF biomarkers. Several previous reports have shown that the levels of T-tau and P-tau become pathological later in the course of AD compared with $A\beta_{42}$, yet it is unclear if higher levels of tau shorten the individuals' life expectancy after diagnosis. The current study aims to investigate whether pathological levels of T-tau and/or P-tau can predict survival in AD.

Methods

The Swedish Alzheimer Treatment Study (SATS) is a prospective, observational, multicenter study for the longitudinal assessment of cholinesterase inhibitor treatment in a routine clinical setting. This presentation includes all 151 participants clinically diagnosed with AD, who underwent a lumbar puncture. Patients were evaluated regarding cognitive and functional abilities at baseline (time of diagnosis) and semi-annually over 3 years. Sociodemographic characteristics, concomitant medications and the date of death were recorded.

CSF was collected in polypropylene tubes, stored at −80 °C and analyzed after the clinical follow-up of the study was completed. The levels of T-tau, P-tau phosphorylated at Thr_{181} and $A\beta_{42}$ were determined using xMAP technology. Pathological levels of CSF biomarkers were defined as: T-tau >100 ng/ml, P-tau >51 ng/ml and $A\beta_{42}$ <209 ng/ml [1].

Independent-sample *t* tests were used to compare the differences between the means obtained for two groups, and χ^2 tests were computed to analyze categorical variables (Table 1). Cox proportional hazards regression was used to determine the patient characteristics that affected mortality. Potential predictors were investigated, including sex, age at baseline, apolipoprotein E (APOE) ε4 carrier status, years of education, the clinician's estimated duration of AD, cognitive and functional abilities at baseline, the number of concomitant medications, and levels of CSF biomarkers (Table 2). One-way analysis of variance (ANOVA) was performed to compare differences between the means obtained for the four groups (Figures 1 and 2).

Reference: 1. Hertze *et al.* Evaluation of CSF Biomarkers as Predictors of Alzheimer's Disease: A Clinical Follow-Up Study of 4.7 Years. *Journal of Alzheimer's Disease*, 2010;21:1119–1128.

Contact address: Carina Wattmo, RN, BSc, PhD, Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, SE-205 02 Malmö, Sweden. Tel +46 40 33 56 01, Fax +46 40 33 56 57, E mail: carina.wattmo@skane.se

Poster presented at the 10th Clinical Trials Conference on Alzheimer's Disease, Boston, MA, USA; November 1-4, 2017.

Results

Table 1. Sociodemographic and clinical characteristics at time of AD diagnosis (baseline)

	Tau– (*n* = 73)	Tau+ (*n* = 78)	*p* value
Female sex	46 (63%)	58 (74%)	0.132
APOE ε4 carrier	49 (67%)	58 (74%)	0.328
Number of deceased patients after 18 years of follow-up	65 (89%)	74 (95%)	0.186
Estimated age at onset of AD, years	73.5 ± 6.9	72.4 ± 6.9	0.325
Estimated duration of AD, years	3.0 ± 2.2	2.7 ± 1.8	0.483
Age, years	76.4 ± 5.8	75.1 ± 6.6	0.190
Education, years	10.2 ± 2.9	9.1 ± 2.0	**0.013**
MMSE score, range 0–30	22.8 ± 4.1	20.6 ± 4.6	**0.002**
IADL score, range 8–31	16.3 ± 5.8	15.7 ± 5.3	0.474
PSMS score, range 6–30	7.8 ± 2.7	7.7 ± 2.4	0.833
Number of concomitant medications	3.4 ± 2.6	2.8 ± 2.5	0.154
$A\beta_{42}$, ng/ml	124 ± 24	117 ± 15	**0.034**
T-tau, ng/ml	71 ± 16	136 ± 49	**<0.001**
P-tau, ng/ml	30 ± 13	69 ± 28	**<0.001**
Time from AD diagnosis to death, years (*n* = 139)	7.0 ± 3.6	6.5 ± 2.9	0.400
Age at death, years (*n* = 139)	83.6 ± 6.0	81.8 ± 5.9	0.068

Data are presented as number (%) or mean ± standard deviation.

$A\beta_{42}$, amyloid-β_{1-42}; AD, Alzheimer's disease; APOE, apolipoprotein E; IADL, Instrumental Activities of Daily Living scale; MMSE, Mini-Mental State Examination; PSMS, Physical Self-Maintenance Scale; P-tau, phosphorylated tau; T-tau, total tau; Tau–, normal T-tau and P-tau; Tau+, pathological T-tau and/or P-tau.

Table 2. Cox proportional hazards modeling of time to death

Independent variables	Hazard ratio (95% CI)	*p* value
Sex (male = 0, female = 1)	0.90 (0.60–1.34)	0.611
APOE ε4 carrier (no = 0, yes = 1)	0.94 (0.60–1.45)	0.764
Estimated duration of AD, years	0.92 (0.84–1.01)	0.087
Age at AD diagnosis, years	1.06 (1.03–1.10)	**0.001**
Education, years	0.99 (0.92–1.07)	0.870
MMSE score at AD diagnosis	0.90 (0.86–0.95)	**<0.001**
IADL score at AD diagnosis	1.02 (0.97–1.07)	0.513
PSMS score at AD diagnosis	1.00 (0.91–1.10)	0.955
Number of concomitant medications at AD diagnosis	1.01 (0.93–1.09)	0.813
$A\beta_{42}$[a]	1.006 (0.996–1.016)	0.258
T-tau[a]	1.003 (0.996–1.009)	0.379
P-tau[a]	1.002 (0.991–1.013)	0.700

Hazard ratios are expressed per 1 unit increase for continuous variables and for the condition present in categorized variables.

[a]Dichotomously coded CSF biomarkers (normal/pathological), instead of the continuous values, were not significant in a Cox regression model adjusted for the abovementioned predictors. Age and MMSE score at AD diagnosis were also significant predictors of survival time after diagnosis in that model.

$A\beta_{42}$, amyloid-β_{1-42}; AD, Alzheimer's disease; APOE, apolipoprotein E; CI, confidence interval; CSF, cerebrospinal fluid; IADL, Instrumental Activities of Daily Living scale; MMSE, Mini-Mental State Examination; PSMS, Physical Self-Maintenance Scale; P-tau, phosphorylated tau; T-tau, total tau.

- The number and frequency of SATS participants with pathological CSF biomarkers were: T-tau, *n* = 18 (12%); P-tau, *n* = 14 (9%); and both T-tau and P-tau, *n* = 46 (31%). All 151 patients had pathological $A\beta_{42}$.
- After 18 years of follow-up, 139 of the 151 participants (92%) had died; their mean (95% confidence interval [CI]) life-span after AD diagnosis was 6.7 (6.2–7.3) years. The numbers (%) of deceased individuals with pathological CSF biomarkers were: T-tau, *n* = 17 (12%); P-tau, *n* = 14 (10%); and both T-tau and P-tau, *n* = 43 (31%).
- No linear associations were found between life expectancy after AD diagnosis and $A\beta_{42}$ (r = −0.005, p = 0.957), T-tau (r = −0.127, p = 0.135), or P-tau (r = −0.020, p = 0.816). Moreover, no significant linear relationships were observed between survival time and any of the CSF biomarkers in the APOE ε4 noncarrier or in the ε4 carrier groups.

Figure 1. Normal/pathological CSF tau biomarkers

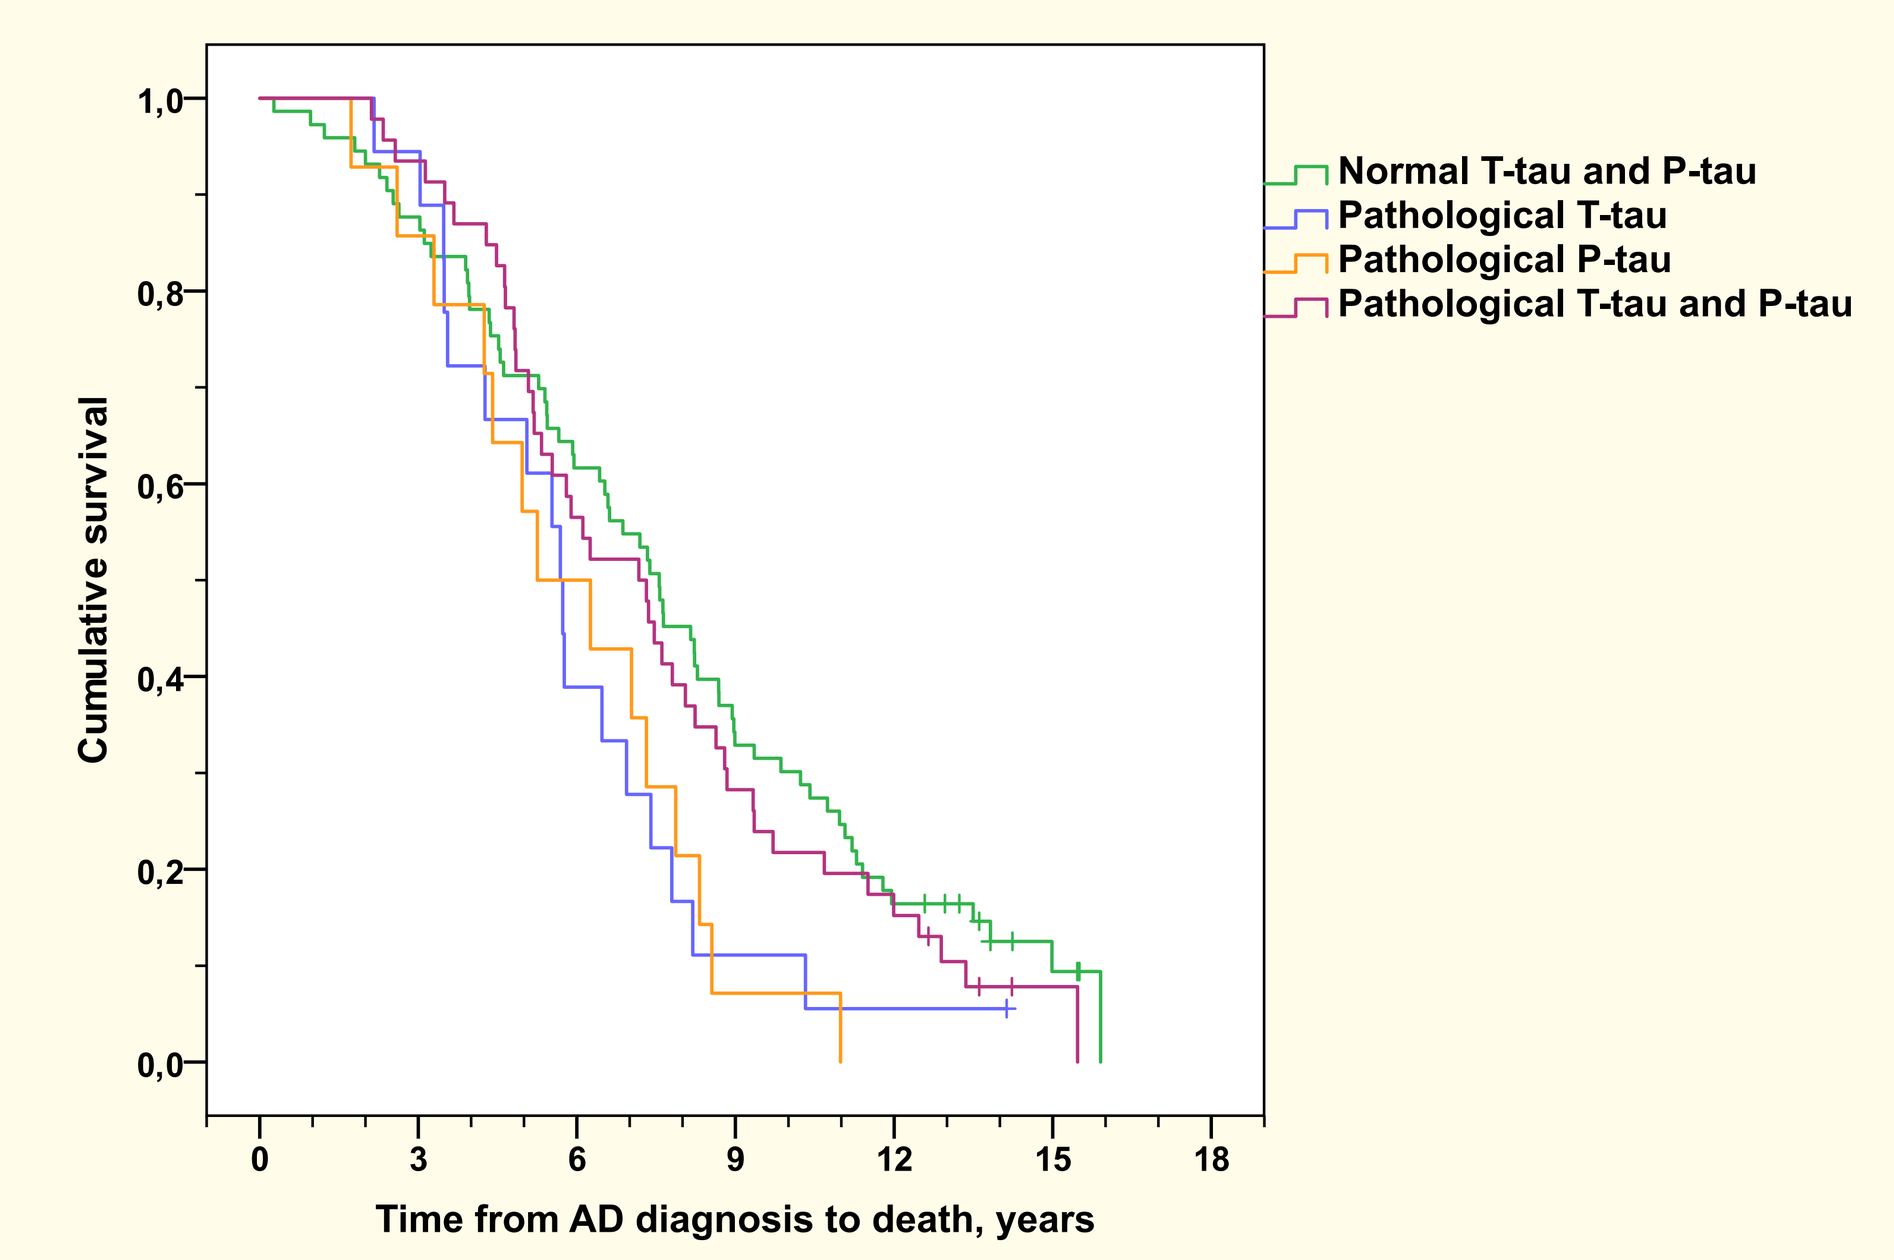
1,0
0,8
0,6
0,4
0,2
0,0
Cumulative survival
0
3
6
9
12
15
18
Time from AD diagnosis to death, years
Normal T-tau and P-tau
Pathological T-tau
Pathological P-tau
Pathological T-tau and P-tau

Kaplan–Meier graph of the distribution of time from the time of AD diagnosis to death for the four groups according to normal/pathological levels of CSF T-tau and P-tau. Using pairwise log-rank tests, the SATS patients with normal tau levels showed a longer life expectancy than those with pathological T-tau, >100 ng/ml (p = 0.044) and P-tau, >51 ng/ml (p = 0.025), but not if both tau biomarkers were pathological (p = 0.439). No difference in proportion of deaths was observed between the groups (p = 0.497).

For the 139 deceased individuals, the mean (95% CI) life-span after diagnosis was similar in the four AD groups when using an ANOVA: normal T-tau and P-tau, 7.0 (6.1–7.9) years; pathological T-tau, 5.6 (4.5–6.7) years; pathological P-tau, 5.9 (4.4–7.4) years; and both pathological T-tau and P-tau, 7.1 (6.1–8.1) years, p = 0.276.

Participants with the highest quartile and quintile of T-tau (≥126 and ≥129 ng/ml) and P-tau (≥65 and ≥70 ng/ml), respectively, were also examined; their survival time did not differ from the other individuals.

Figure 2. Normal/pathological CSF tau and APOE genotype

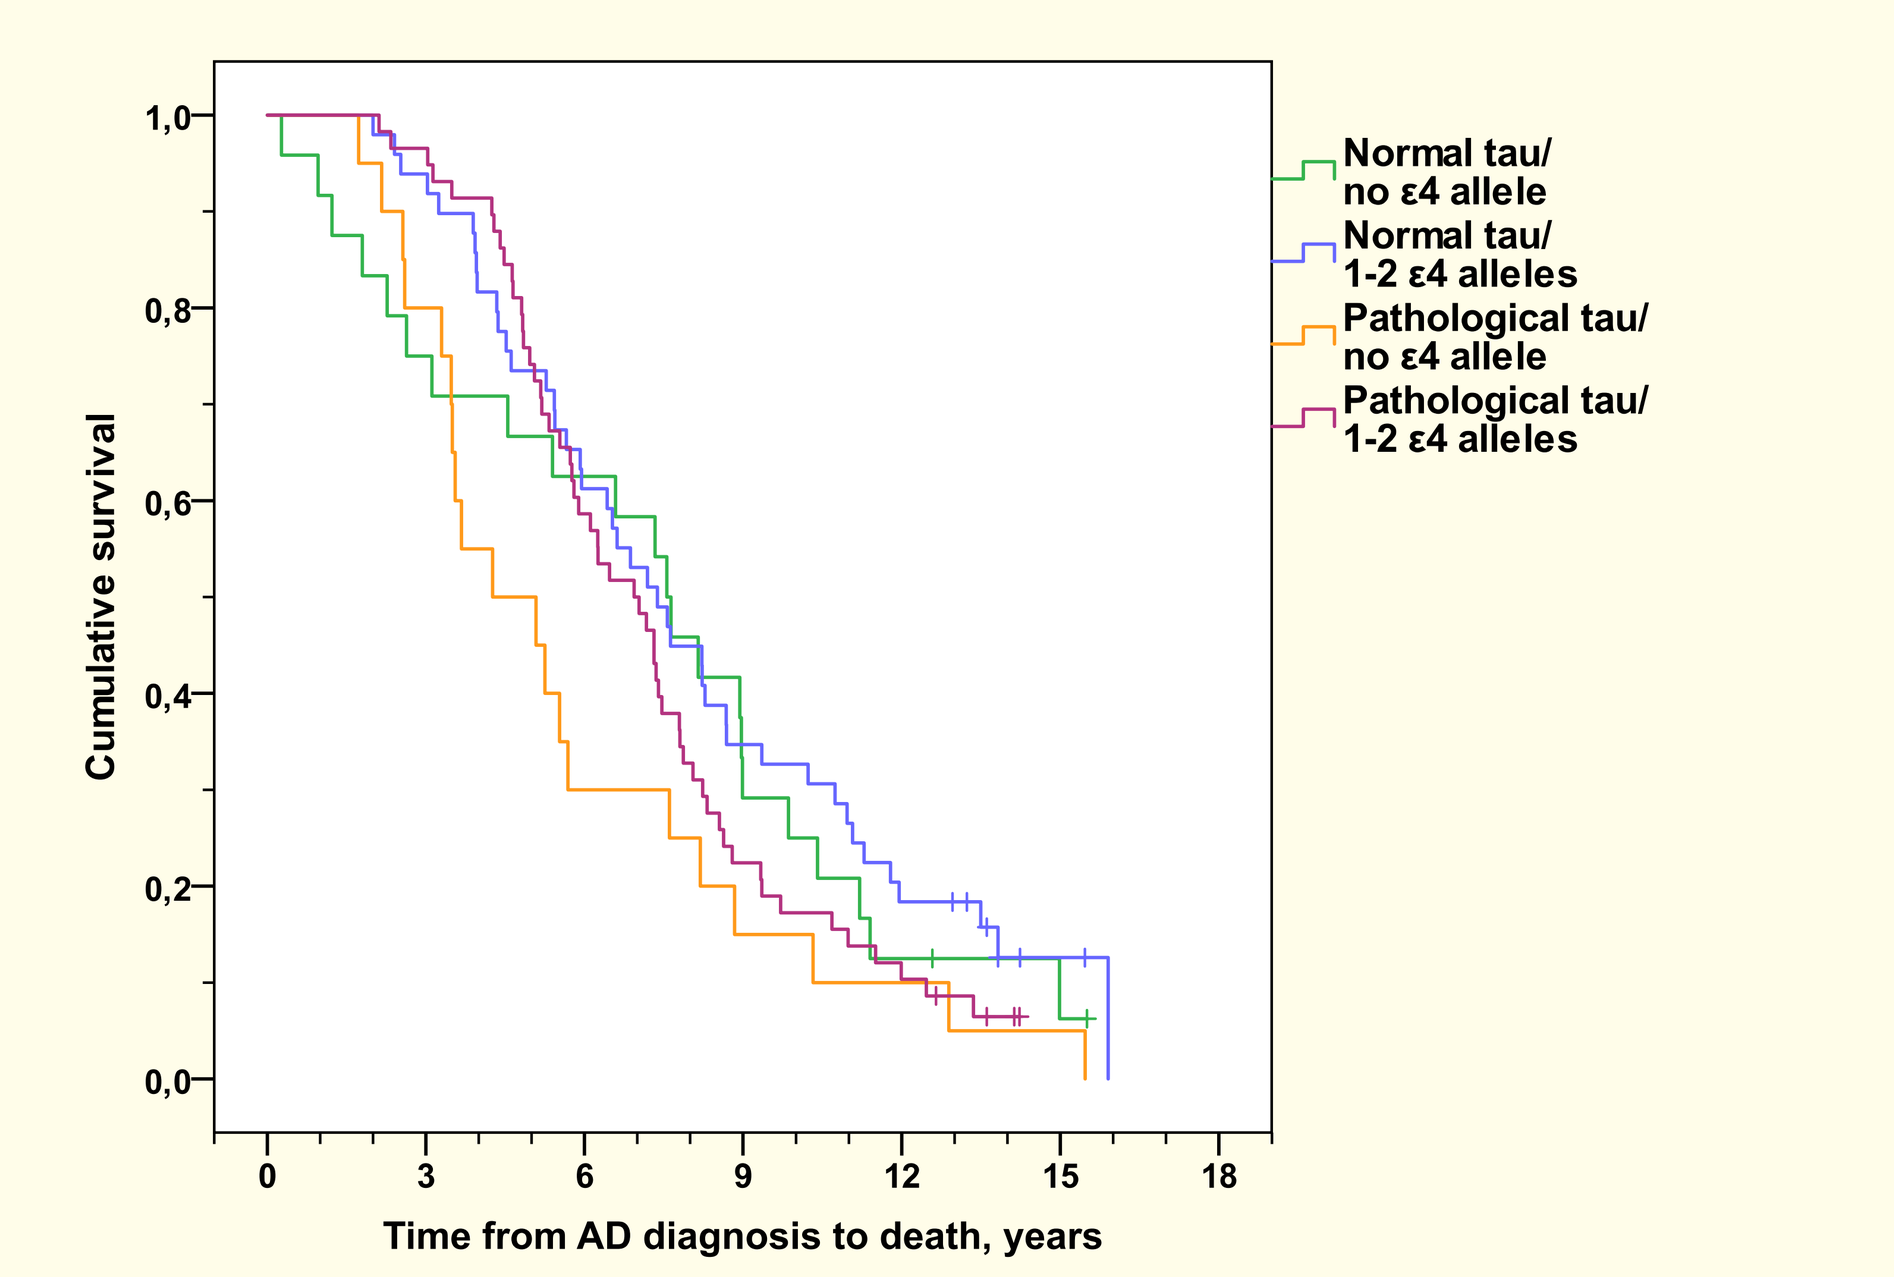
1,0
0,8
0,6
0,4
0,2
0,0
Cumulative survival
0
3
6
9
12
15
18
Time from AD diagnosis to death, years
Normal tau/ no ε4 allele
Normal tau/ 1-2 ε4 alleles
Pathological tau/ no ε4 allele
Pathological tau/ 1-2 ε4 alleles

Kaplan–Meier graph of the distribution of time from the time of AD diagnosis to death for the four groups according to the interaction effect of normal/pathological levels of CSF tau with presence/absence of the APOE ε4 allele. Using pairwise log-rank tests, the patients with normal tau/absence of APOE ε4 allele (n = 24) exhibited similar life expectancy to those with normal tau/presence of APOE ε4 allele (n = 49, p = 0.547), pathological tau/absence of APOE ε4 allele (n = 20, p = 0.199), and pathological tau/presence of APOE ε4 allele (n = 58, p = 0.514). However, the participants with normal tau/presence of APOE ε4 allele had a longer survival time compared with individuals with pathological tau/absence of APOE ε4 allele (p = 0.017). The proportion of deaths did not differ between the groups (p = 0.383).

For the 139 deceased AD patients, no difference in mean (95% CI) life-span after diagnosis was detected between the four groups: normal tau/absence of APOE ε4 allele, n = 22, 6.6 (4.8–8.3) years; normal tau/presence of APOE ε4 allele, n = 43, 7.2 (6.2–8.2) years; pathological tau/absence of APOE ε4 allele, n = 20, 5.8 (4.0–7.5) years; and pathological tau/presence of APOE ε4 allele, n = 54, 6.8 (6.1–7.5) years, (ANOVA, p = 0.451).

Layout: Maverick by Sigma | maverickbysigma.se