LUND UNIVERSITY

Cholinesterase inhibitor therapy does not affect time spent in nursing homes in patients with Alzheimer’s disease.

Wattmo, Carina; Londos, Elisabet; Minthon, Lennart

2016

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Wattmo, C., Londos, E., & Minthon, L. (2016). *Cholinesterase inhibitor therapy does not affect time spent in nursing homes in patients with Alzheimer’s disease.*. Poster session presented at Alzheimer’s Association International Conference 2016, Toronto, Canada.

Total number of authors:
3

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
• Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
• You may not further distribute the material or use it for any profit-making activity or commercial gain
• You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

CHOLINESTERASE INHIBITOR THERAPY DOES NOT AFFECT TIME SPENT IN NURSING HOMES IN PATIENTS WITH ALZHEIMER’S DISEASE

Carina Wattmo, Elisabet Londos, Lennart Minthon

Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, Sweden

CONCLUSIONS

Women cared for their spouses with Alzheimer’s disease (AD) at home longer than did men. The situation of these female informal caregivers needs attention and possibly support. There was no indication that any aspects of cholinesterase inhibitor (ChEI) therapy (drug agent, dosage, or duration of treatment) altered the survival time in nursing homes (NHs).

BACKGROUND

An increased knowledge of factors that affect the time spent in NHs for AD patients treated with ChEIs is important for clinicians and community-based services. The length of stay in NHs might be influenced by factors such as sociodemographic characteristics, rate of AD progression, impairment in activities of daily living (ADL), and concomitant disorders. Whether different aspects of ChEI therapy (drug agent, dose, or treatment duration) influence survival time in NHs has not been previously investigated. We aimed to study the relationship between the aforementioned potential predictors and time spent in NHs.

METHODS

The Swedish Alzheimer Treatment Study (SATS) is a prospective, observational, multicenter study for the long-term assessment of ChEI therapy in a routine clinical setting. This presentation includes information about 220 deceased SATS participants who had been clinically diagnosed with mild-to-moderate AD (Mini-Mental State Examination (MMSE) score, 10–26 at the start of ChEI treatment) who were admitted to NHs during the study. Cognitive status (MMSE) and functional performance (Instrumental Activities of Daily Living scale (IADL) and Physical Self-Maintenance Scale (PSMS)) were evaluated at the start of ChEI therapy (about the time of AD diagnosis) and semiannually over 3 years. Concomitant medications and dates of nursing home placement (NHP) and death were recorded. The choice of drug agent and all decisions regarding dosage for each individual patient were left entirely to the discretion and professional judgment of dementia specialists.

A *t*-test (Figure 1) and one-way analysis of variance (ANOVA) with Bonferroni correction (Figures 2–4) were used to compare differences between the means. A general linear model was used to identify the AD patients’ characteristics that independently affected the survival time from NHP to death (Table 2). The following potential predictors were investigated: sex, living alone (yes/no), age, years of education, cognitive ability, instrumental and basic ADL capacities at NHP, rates of decline in cognition and function, number of concomitant medications, and type of ChEI, dose, and duration of treatment.

Contact address:
Carina Wattmo, RN, BSc, PhD, Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, SE-205 02 Malmö, Sweden.
Tel +46 40 33 56 01, Fax +46 40 33 56 57, E mail: carina.wattmo@skane.se

Poster presented at the Alzheimer’s Association International Conference, Toronto, Canada; July 24-28, 2016.

RESULTS

Table 1. Sociodemographic and clinical characteristics (n = 220)

	At start of ChEI treatment	At nursing home placement
Female sex	161 (73%)	
Living alone	116 (53%)	126 (57%)
Estimated age at onset of AD, years	73.5 ± 6.9	
Estimated duration of AD, years	3.4 ± 2.6	5.1 ± 2.7
Age, years	76.9 ± 5.9	78.6 ± 5.8
Education, years	9.2 ± 2.3	
MMSE score	20.1 ± 4.0	17.5 ± 5.7
IADL score	18.3 ± 5.1	22.7 ± 4.7
PSMS score	8.2 ± 2.8	10.5 ± 3.9
Number of concomitant medications	3.0 ± 2.4	
Mean dose of ChEI during the SATS, mg		
Donepezil (n = 138)	6.8 ± 1.8	
Rivastigmine (n = 37)	5.7 ± 1.9	
Galantamine (n = 45)	15.1 ± 3.2	
Time from start of ChEI therapy to NHP, months	19.5 ± 10.0	
Age at death, years	82.6 ± 6.0	

The data are expressed as either number (percentage) or mean ± standard deviation.
AD, Alzheimer’s disease; ChEI, cholinesterase inhibitor; IADL, Instrumental Activities of Daily Living scale; MMSE, Mini-Mental State Examination; NHP, nursing home placement; PSMS, Physical Self-Maintenance Scale; SATS, Swedish Alzheimer Treatment Study.

Table 2. General linear model with time spent in nursing homes (years) as the dependent variable and significantly associated predictors

Independent variable	β	95% CI	*p*-value
Intercept	3.899	2.594, 5.204	<0.001
Sex by living status[a]			
Women living with family	2.549	1.488, 3.610	<0.001
Women living alone	2.281	1.326, 3.236	<0.001
Men living alone	1.834	0.436, 3.231	0.010
PSMS score at NHP	−0.111	−0.201, −0.021	0.016
Number of concomitant medications	−0.195	−0.337, −0.053	0.007

[a]Men living with family was the reference category.
β-Values are unstandardized and are expressed per 1-unit increase for continuous variables and for the condition present in dichotomous variables.
CI, confidence interval; NHP, nursing home placement; PSMS, Physical Self-Maintenance Scale.

The interaction effect of sex by living status, basic ADL impairment at NHP, and number of concomitant medications were independent predictors of survival time in NHs. Age, years of education, cognitive ability or instrumental ADL capacities at NHP, rate of decline in cognition or function, and type of ChEI, dose, and treatment duration were not significant variables in the multivariate model.

Figure 1.

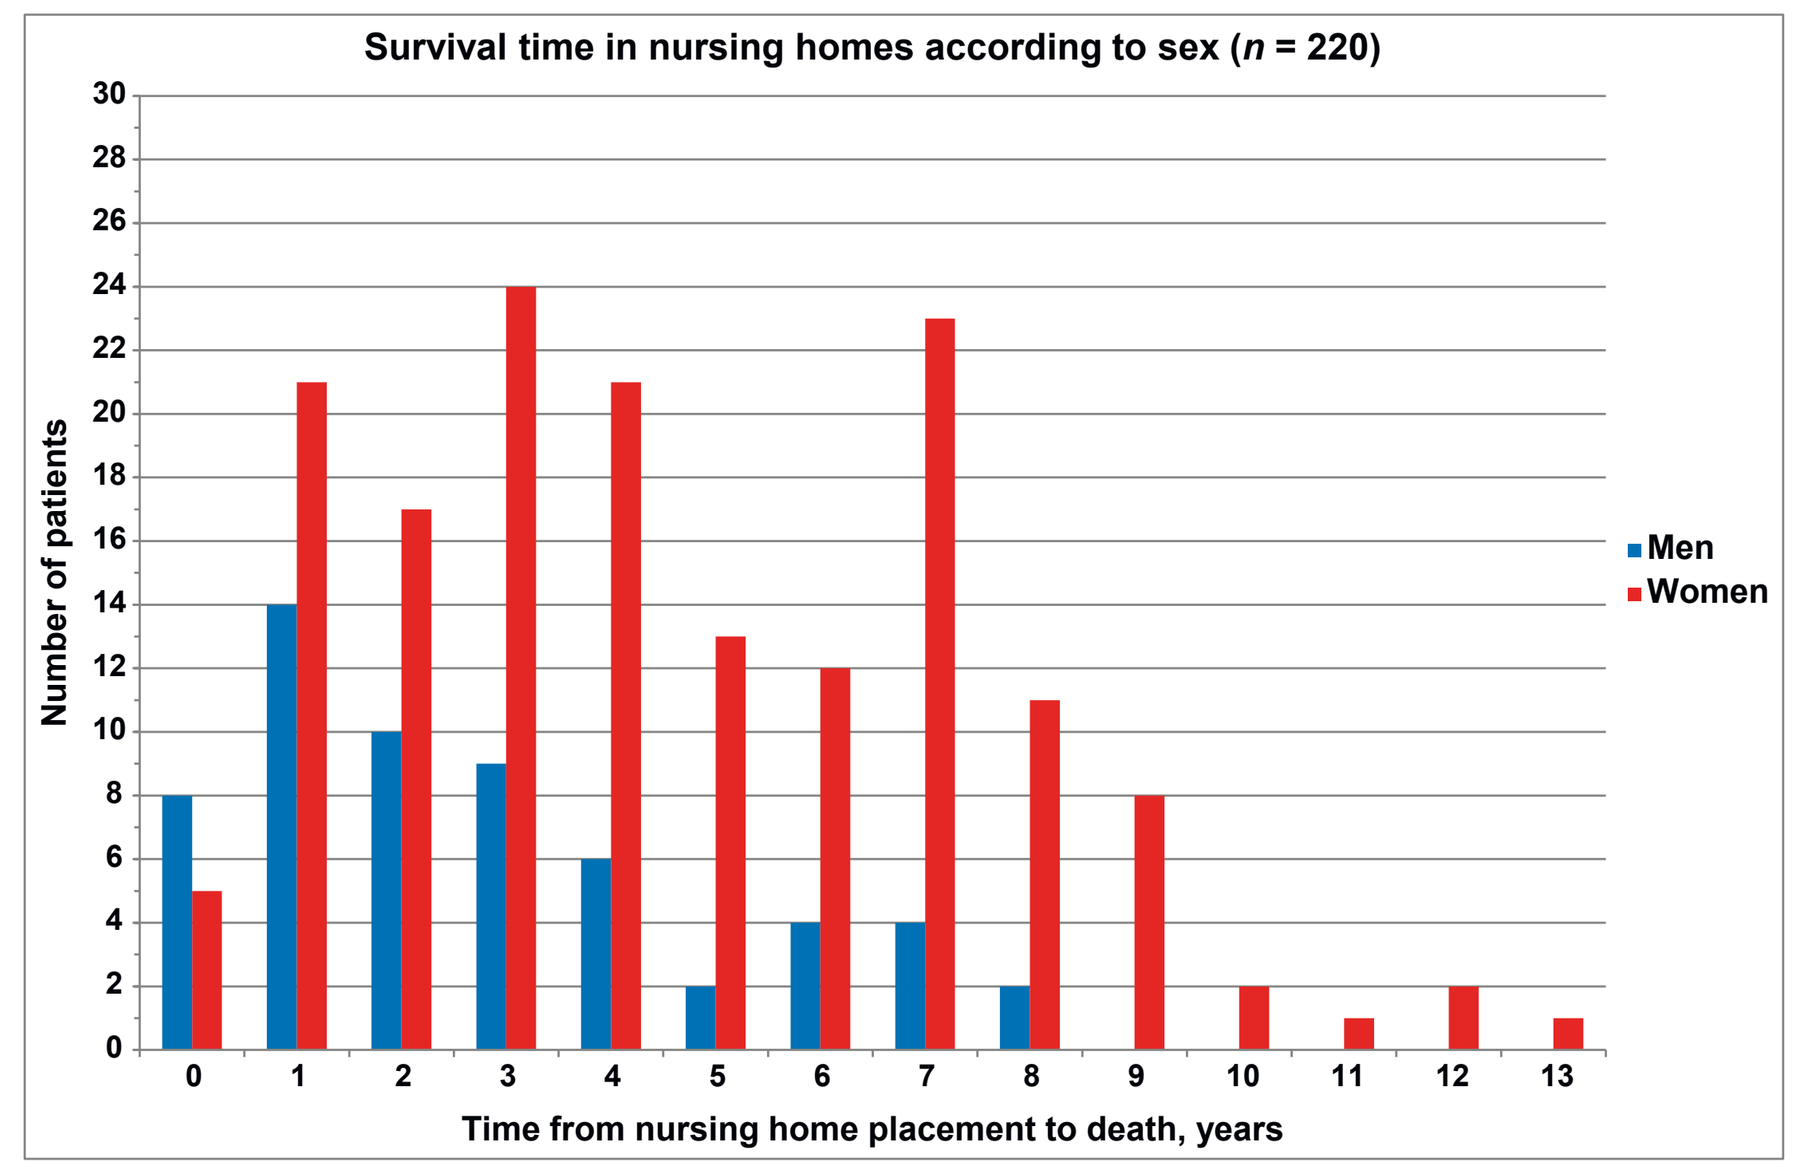

Survival time in nursing homes according to sex (n = 220)
Number of patients
Time from nursing home placement to death, years
Men
Women

The SATS patients’ time spent in NHs are shown separately for men and women. In the figure, time was rounded to the nearest integer year. The mean ± standard deviation (SD) length of time in NHs was 4.06 ± 2.77 years. Men with AD spent a shorter time in NHs than did women: 2.78 ± 2.29 years vs. 4.53 ± 2.79 years, respectively ($p < 0.001$). No sex difference was observed in age at AD onset, illness duration, age at diagnosis, time between diagnosis and NHP, age at NHP, and age at death.

Figure 2.

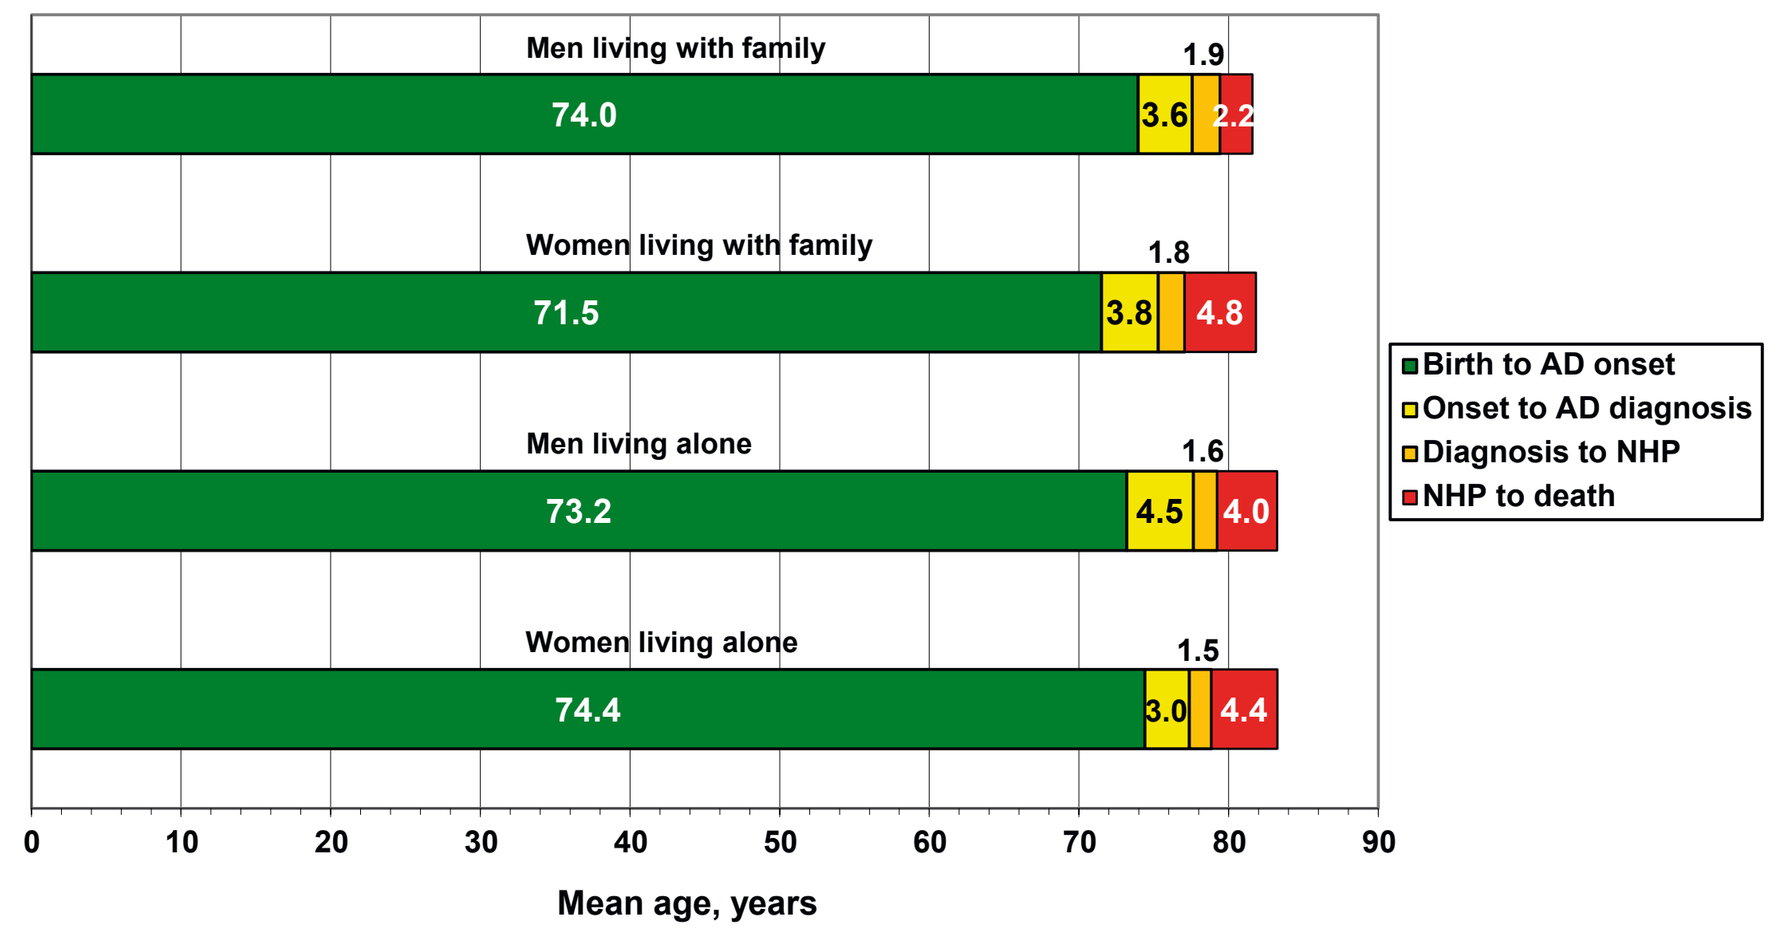

Time in nursing homes according to the interaction effect of sex with living status
Men living with family
74.0
3.6
1.9
2.2
Women living with family
71.5
3.8
1.8
4.8
Men living alone
73.2
4.5
1.6
4.0
Women living alone
74.4
3.0
1.5
4.4
Birth to AD onset
Onset to AD diagnosis
Diagnosis to NHP
NHP to death
Mean age, years

Mean age at AD onset, age at diagnosis, age at NHP, and age at death according to the interaction effect of sex by living status. Men living with a family member spent less time in NHs (mean ± SD, 2.2 ± 2.1 years) compared with the other groups: women living with family (4.8 ± 2.8 years); men living alone (4.0 ± 2.2 years); and women living alone (4.4 ± 2.8 years, $p < 0.001$). Moreover, men living with family had a longer mean time from AD diagnosis to NHP (1.9 ± 0.7 years) than did women living alone (1.5 ± 0.9 years, $p = 0.022$). No significant differences were detected in the age at AD onset, illness duration, age at diagnosis, age at NHP, and age at death.

Figure 3.

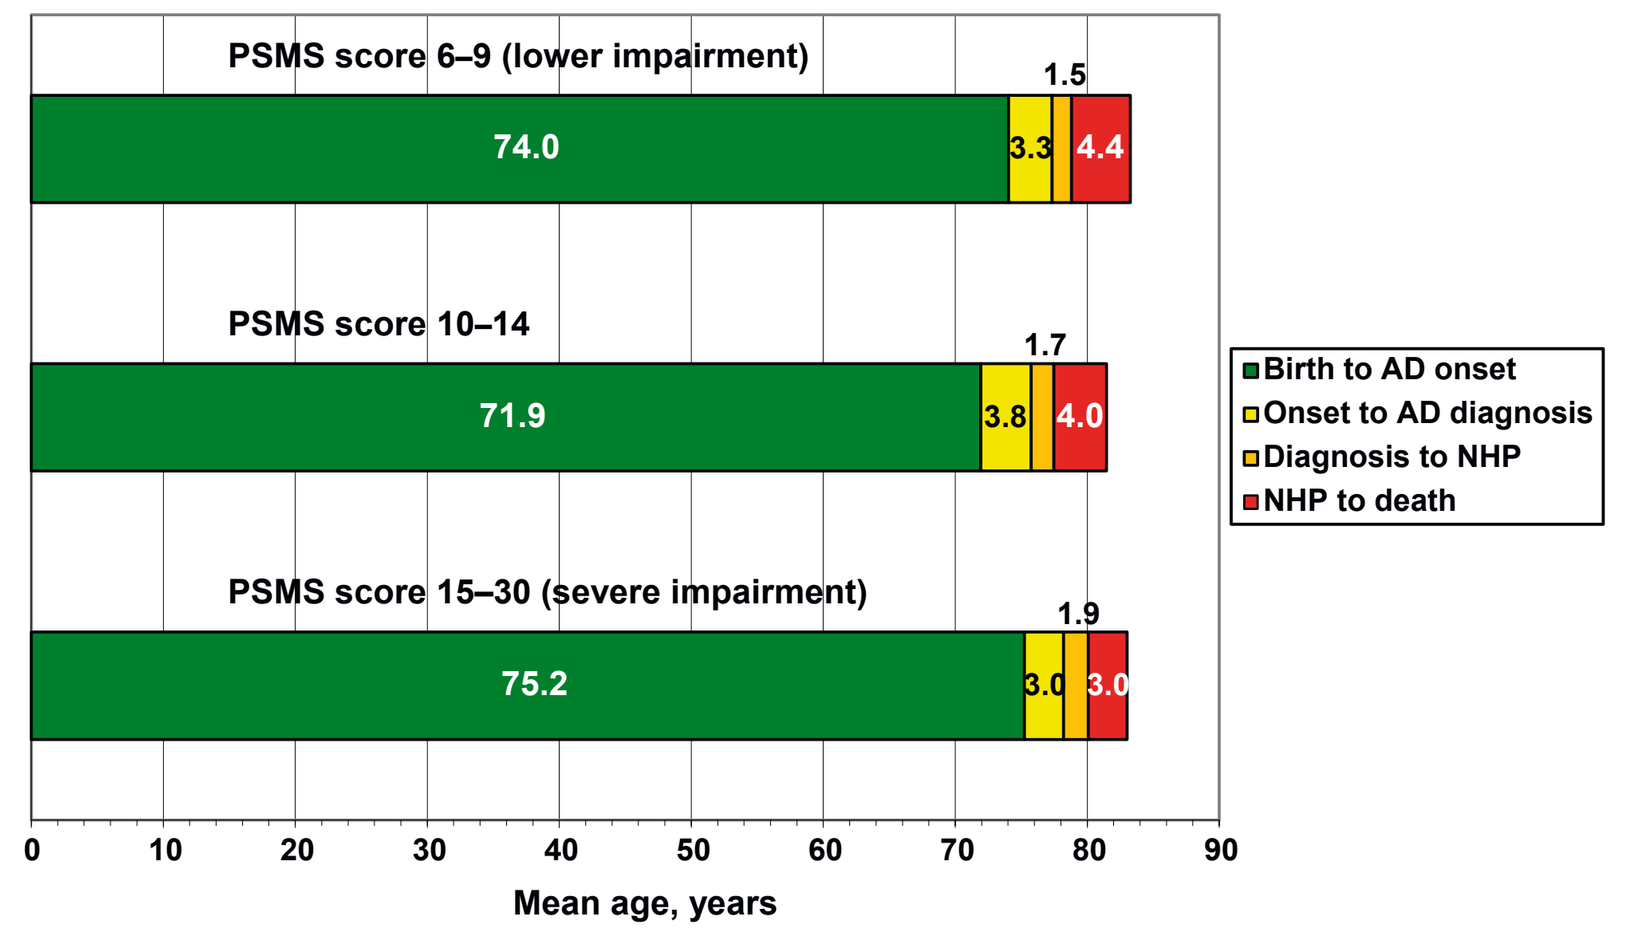

Time in nursing homes according to basic ADL at NHP
PSMS score 6–9 (lower impairment)
74.0
3.3
1.5
4.4
PSMS score 10–14
71.9
3.8
1.7
4.0
PSMS score 15–30 (severe impairment)
75.2
3.0
1.9
3.0
Birth to AD onset
Onset to AD diagnosis
Diagnosis to NHP
NHP to death
Mean age, years

Mean age at AD onset, age at diagnosis, age at NHP, and age at death in the three groups of SATS participants grouped according to their basic ADL performance at NHP. The individuals who exhibited a PSMS score of 6–9 at NHP spent a longer time in NHs (mean ± SD, 4.4 ± 2.9 years) than did those who had a PSMS score of 15–30 (3.0 ± 2.1 years, $p = 0.020$). No difference in survival time in NHs was observed between patients with no basic ADL impairment (PSMS score of 6) at NHP and those with a PSMS score of 7–9. Worse basic ADL capacity at NHP and a longer time between AD diagnosis and NHP also showed a significant relationship ($p = 0.021$). The group with a PSMS score of 10–14 was younger at AD onset than that with a PSMS score of 15–30 ($p = 0.034$). No differences were found in illness duration, age at diagnosis, age at NHP, and age at death between the three groups.

Figure 4.

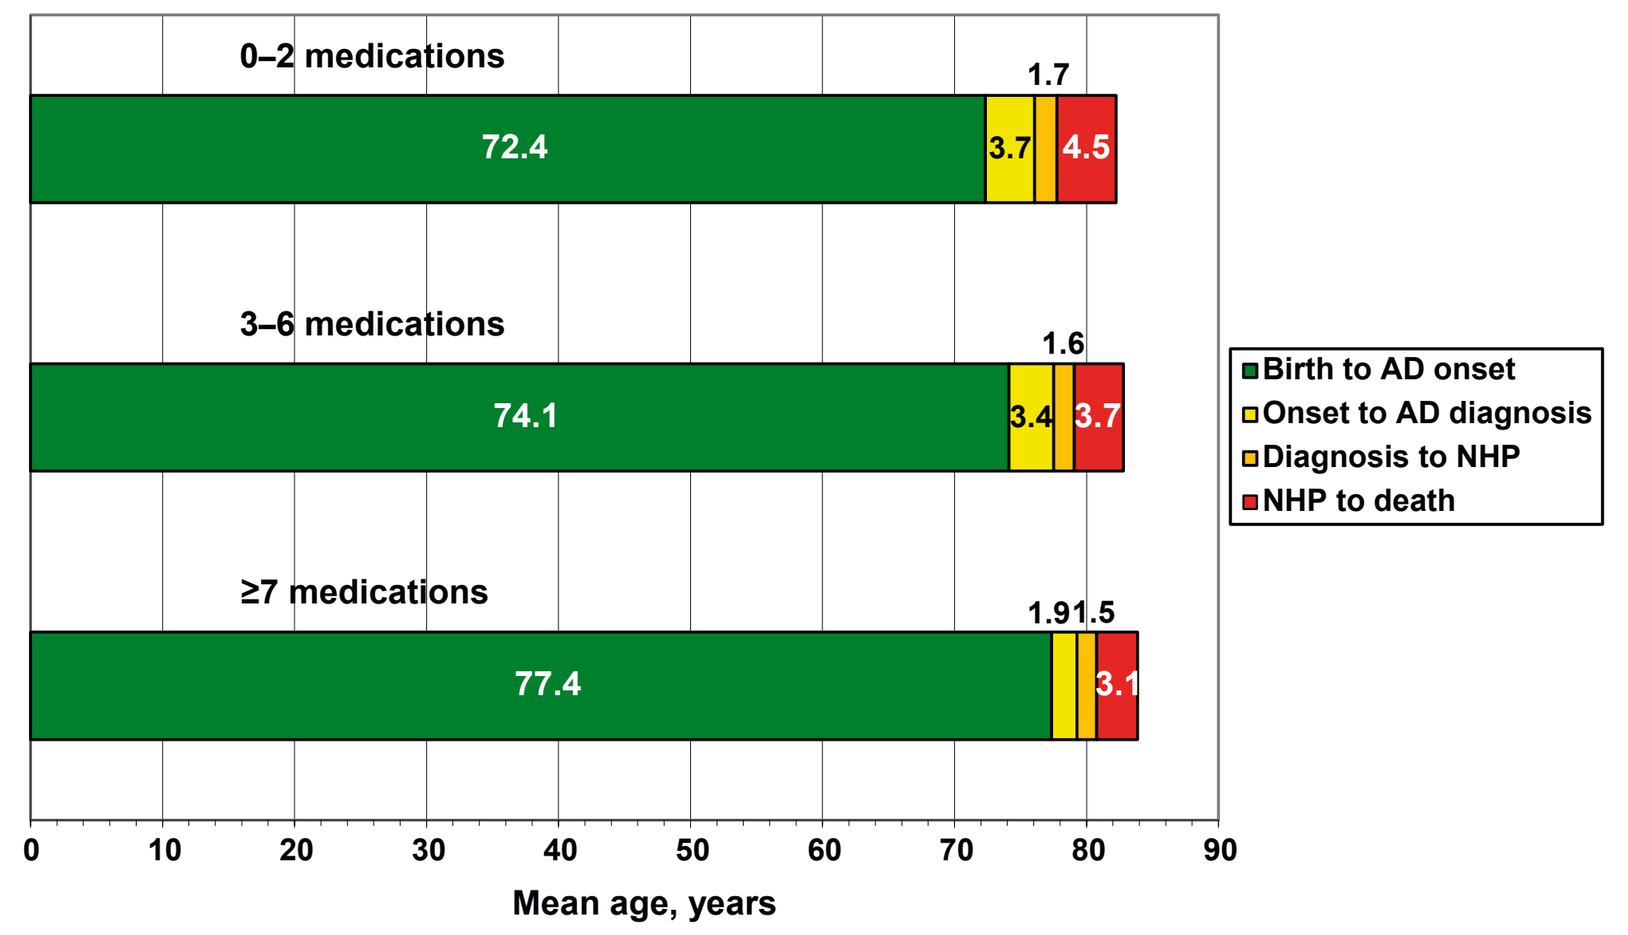

Time in nursing homes according to the number of medications
0–2 medications
72.4
3.7
1.7
4.5
3–6 medications
74.1
3.4
1.6
3.7
≥7 medications
77.4
1.9
1.5
3.1
Birth to AD onset
Onset to AD diagnosis
Diagnosis to NHP
NHP to death
Mean age, years

Mean age at AD onset, age at diagnosis, age at NHP, and age at death in the three groups of participants grouped according to the number of medications at the time of AD diagnosis (start of ChEI therapy). The patients who received 0–2 medications spent a longer time in NHs (mean ± SD, 4.5 ± 2.6 years) compared with those who received 3–6 or ≥7 medications (3.7 ± 3.0 years and 3.1 ± 2.4 years, respectively, $p = 0.044$). Taking more medications at the time of diagnosis was significantly associated with older age at AD onset ($p = 0.009$), shorter illness duration ($p = 0.022$), and older age at diagnosis ($p = 0.043$) and at NHP ($p = 0.043$). No differences were detected in the time between AD diagnosis and NHP, and age at death between the three groups.

Layout by Aletia Design, www.aletiadesign.com