LUND UNIVERSITY

Anticipating morphological and syntactic structures

investigating the pre-activation negativity

Söderström, Pelle; Horne, Merle; Mannfolk, Peter; van Westen, Danielle; Roll, Mikael

2017

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Söderström, P., Horne, M., Mannfolk, P., van Westen, D., & Roll, M. (2017). *Anticipating morphological and syntactic structures: investigating the pre-activation negativity*. Poster session presented at Cognitive Neuroscience Society Annual Meeting 2017, San Francisco, United States.

Total number of authors:
5

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
• Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
• You may not further distribute the material or use it for any profit-making activity or commercial gain
• You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

Anticipating morphological and syntactic structures

An analysis of the pre-activation negativity (PrAN)

Pelle Söderström, Merle Horne, Peter Mannfolk, Danielle van Westen & Mikael Roll

Department of Linguistics, Centre for Languages and Literature

Lund University, Sweden

Vetenskapsrådet

Knut and Alice Wallenberg Foundation

MARCUS AND AMALIA WALLENBERG FOUNDATION

Introduction

- Listeners constantly try to predict upcoming words when processing speech
- A brain potential – the 'pre-activation negativity' (PrAN) – has been suggested to reflect morphological pre-activation of likely word endings [1-4]
- We tested whether PrAN could be found in syntactically predictive contexts as well

PrAN

bå-… -ten/-tar/-t-hus…

boat-…-the/-s/-house

The present study

- Using a concurrent fMRI/ERP paradigm, we tested whether syntactic structure could be pre-activated based on strongly constraining tonal cues
- In Swedish, clause-initial tones (low/high) function as cues to syntactic structure
- Low tones are more predictively constraining (cueing only one type of structure), whereas high tones are less constraining (cueing a larger class of structures)

- More predictively useful tones gave rise to **left frontal ERP negativity (PrAN)** 140 ms after tone onset, as well as activity in **left insula and inferior frontal gyrus**
- Invalidly cued word orders elicited **P600** after low – but not high – tones, suggesting the disconfirmation of a syntactic prediction

PrAN

Jim hävdar att Cae**sar**$_{\textbf{Low tone}}$ inte intog Gallien 'Jim claims that Caesar **not conquered** Gaul'

Jim hävdar att Cae**sar**$_{\textbf{High tone}}$ intog inte Gallien 'Jim claims that Caesar **conquered not** Gaul'

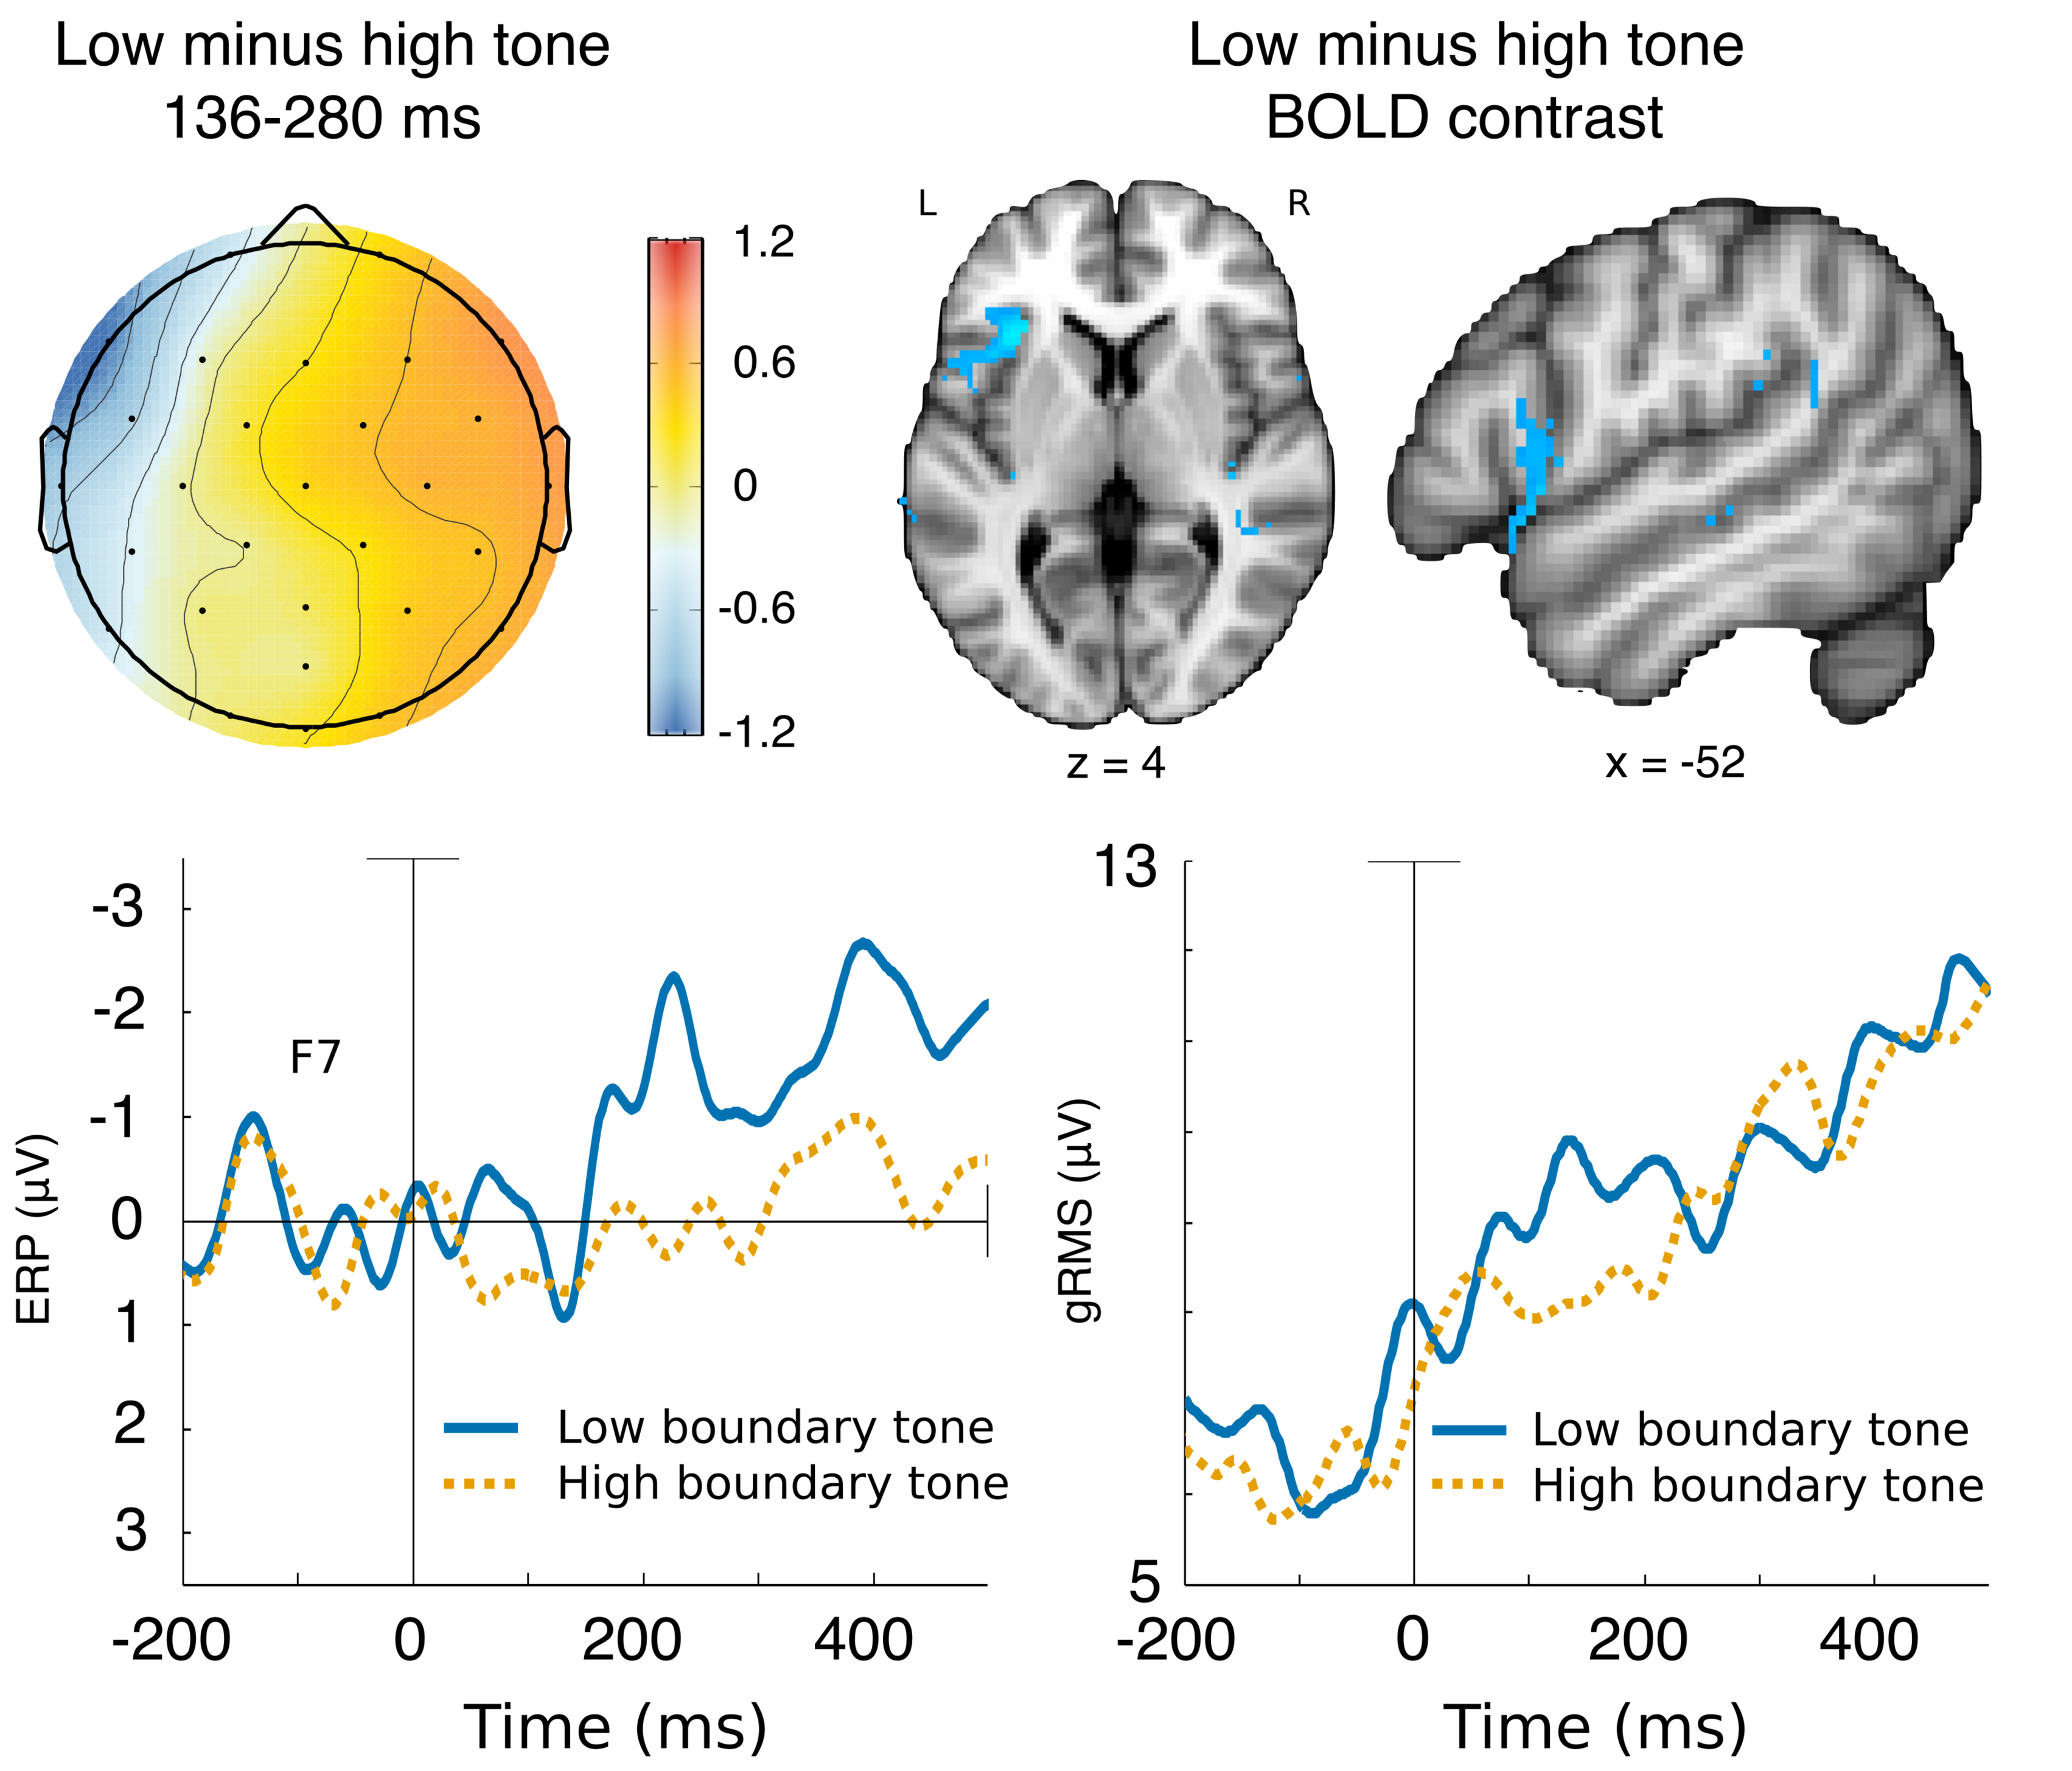
Low minus high tone
136-280 ms
1.2
0.6
0
-0.6
-1.2
Low minus high tone
BOLD contrast
L
R
z = 4
x = -52
F7
ERP (µV)
-3
-2
-1
0
1
2
3
Low boundary tone
High boundary tone
-200
0
200
400
Time (ms)
gRMS (µV)
13
5
Low boundary tone
High boundary tone
-200
0
200
400
Time (ms)

Top left: ERP topography (left frontal) for the ERP negativity found for more predictively constraining (low) tones. Top right: The low minus high tone contrast gave rise to activity in left insula and IFG. Bottom left: ERPs revealed a negativity for low tones beginning at 136 ms. Bottom right: A gRMS analysis suggested more neural activity for low tones in the same time window [5].

Method and results

- 19 native speakers of Swedish (11 female, mean age 24.5 years)
- Concurrent event-related fMRI/ERP (Brain Products GmbH)
- 50% of sentences had invalid word orders based on tonal cue (LoInvalid/HiInvalid)
- ERP data from 16 participants analysed
- Two time points: predictive tone onset, and word order disambiguation point
- Low tones gave rise to ERP negativity in 136-280 ms time window (cf. [3]) over left-lateralised electrodes ($F(1,15) = 7.252$, $p = 0.017$)
- A gRMS analysis revealed two peaks of neural activity at 100-150 ms ($F(1,15) = 5.691$, $p = 0.031$) and 150-230 ms ($F(1,15) = 5.264$, $p = 0.037$) for low tones
- P600 over left electrodes for LoInvalid ($F(1,15) = 5.354$, $p = 0.035$)
- Slower response times for LoInvalid as well ($F(1,15) = 5.944$, $p = 0.028$)
- A conjunction analysis (to isolate effects of tone) was performed on fMRI data (z threshold = 3.2, $p = 0.001$, GRF statistics)
- Largest cluster for the low minus high tone contrast spanned the left anterior insula and left inferior frontal gyrus
- Subject variability correlation between BOLD in prefrontal cluster and gRMS ($r = 0.609$, $p = 0.024$)

Conclusions

- Strong cues to syntactic structure elicited ERP negativity (PrAN) as early as 140 ms after cue onset
- Disconfirmed predictions gave rise to P600
- PrAN was found to mainly be underpinned by activity in left insula and IFG (cf. [6-9])
- Syntactic structures can be pre-activated based on a strongly constraining cue

LUND UNIVERSITY

References

[1] M. Roll *et al.*, "Word-stem tones cue suffixes in the brain." *Brain Research*, vol. 1520, pp. 116–120, 2013.
[2] P. Söderström *et al.*, "Stem tones pre-activate suffixes in the brain," *Journal of Psycholinguistic Research*, 2016.
[3] M. Roll *et al.*, "Word tones cueing morphosyntactic structure: neuroanatomical substrates and activation time course assessed by EEG and fMRI," *Brain & Language*, vol. 150, pp. 14–21, 2015.
[4] P. Söderström *et al.*, "Pre-activation negativity (PrAN) in brain potentials to unfolding words," *Frontiers in Human Neuroscience*, 10, 2016.
[5] W. Skrandies, "Global field power and topographic similarity," *Brain Topography*, vol. 3(1), pp. 137–141, 1990.
[6] J.M. Henderson *et al.*, "Language structure in the brain: A fixation-related fMRI study of syntactic surprisal in reading," *NeuroImage*, vol. 132, pp. 293–300, 2016.
[7] M. Jakuszeit *et al.*, "Generating predictions: Lesion evidence on the role of left inferior frontal cortex in rapid syntactic analysis," *Cortex*, vol. 49, pp. 2861–2874, 2013.
[8] A. Moro *et al.*, "Syntax and the Brain: Disentangling Grammar by Selective Anomalies," *NeuroImage*, vol. 13, pp. 110–118, 2001.
[9] W. Matchin *et al.*, "The role of the IFG and pSTS in syntactic prediction: Evidence from a parametric study of hierarchical structure in fMRI," *Cortex*, vol. 88, pp. 106–123, 2017.