LUND UNIVERSITY

The course and end-points of Alzheimer’s disease according to sociodemographics, apolipoprotein E genotype and cognitive ability.

Wattmo, Carina; Londos, Elisabet; Minthon, Lennart

2017

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Wattmo, C., Londos, E., & Minthon, L. (2017). *The course and end-points of Alzheimer’s disease according to sociodemographics, apolipoprotein E genotype and cognitive ability.*. Poster session presented at Alzheimer's Association International Conference (AAIC), 2017, London, United Kingdom.

Total number of authors:
3

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
• Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
• You may not further distribute the material or use it for any profit-making activity or commercial gain
• You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

THE COURSE AND END-POINTS OF ALZHEIMER'S DISEASE ACCORDING TO SOCIODEMOGRAPHICS, APOLIPOPROTEIN E GENOTYPE AND COGNITIVE ABILITY

Carina Wattmo, Elisabet Londos, Lennart Minthon

Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, Sweden

Contact address:
Carina Wattmo, RN, BSc, PhD, Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, SE-205 02 Malmö, Sweden.
Tel +46 40 33 56 01, Fax +46 40 33 56 57, E mail: carina.wattmo@skane.se

Poster presented at the Alzheimer's Association International Conference, London, England; July 16–20, 2017.

CONCLUSIONS

This study shows that interaction effects between sociodemographic characteristics and clinical factors affect the course and end-points of Alzheimer's disease (AD). Despite similar cognitive ability at AD diagnosis, younger patients exhibited longer time to nursing home placement (NHP) and lower cognitive status at the time of admission than older individuals. In the ≤ 77-year-old group, a significantly longer survival time in nursing homes (NHs) (~5 years), and thereby higher cost of care, might be expected in females, mild AD patients and apolipoprotein E (APOE) ε4-carriers. Younger persons with moderate AD showed remarkably low cognitive ability at NHP (mean Mini-Mental State Examination [MMSE] score 12); these individuals might need increased support.

BACKGROUND

The prognosis of AD might be influenced by many sociodemographic and clinical factors, for example, age at AD diagnosis, sex, APOE genotype, living alone, and cognitive performance. End-points, such as NHP and death, and the associated costs of care, may depend on these patient characteristics. Most earlier studies have investigated the main effects of various critical predictors that could affect the course of AD, but few have analysed potential interactions. This presentation aims to study long-term cognitive outcomes, time to NHP, survival time in NHs, and life expectancy by interactions between the above-mentioned factors in cholinesterase inhibitor (ChEI)-treated AD patients.

METHODS

The Swedish Alzheimer Treatment Study (SATS) is a prospective, observational, multicentre study for the longitudinal evaluation of ChEI therapy in clinical practice. This presentation includes all 224 deceased SATS participants diagnosed with mild-to-moderate AD (MMSE score 10–26 at the initiation of ChEI treatment, i.e., at the time of diagnosis) who were admitted to NHs during the study period. Sociodemographic characteristics, APOE genotype, dates of NHP and death were recorded. Cognitive abilities, e.g., MMSE scores, were assessed at the start of ChEI therapy (baseline) and semi-annually over 3 years. Chi-square tests (Table 1) were performed to analyse categorical variables. Independent-samples *t* tests (Table 1) and one-way analysis of variance (ANOVA) with Bonferroni correction (Figures 1–4) were used to compare differences between the means obtained for two and four groups, respectively.

RESULTS

Table 1. Sociodemographic and clinical characteristics by age at AD diagnosis, median cut-off 78 years ($n = 224$)

	Age group ≤ 77 years ($n = 112$)	**Age group ≥ 78 years** ($n = 112$)	***p* value**
	n / %		
Female sex	81 / 76%	84 / 71%	0.375
APOE ε4 carrier, ($n = 212$)	80 / 76%	75 / 70%	0.317
Living alone	49 / 46%	70 / 59%	0.061
	Mean ± standard deviation		
Estimated age at onset of AD, years	68.2 ± 6.1	78.0 ± 3.7	< 0.001
Estimated duration of AD, years	3.7 ± 3.0	3.2 ± 2.0	0.114
Age at baseline, years	71.9 ± 4.7	81.2 ± 2.8	< 0.001
Age at NHP, years	73.8 ± 4.7	82.6 ± 2.9	< 0.001
Education, years	9.2 ± 2.4	9.1 ± 2.2	0.650
MMSE score at baseline	20.0 ± 4.3	20.1 ± 3.8	0.907
MMSE score at NHP	16.2 ± 6.3	18.7 ± 4.9	0.002
Time from the start of ChEI therapy to NHP, months	22.2 ± 9.3	17.0 ± 9.9	< 0.001
Time from NHP to death, years	4.8 ± 3.1	3.7 ± 2.8	0.009
Time from AD diagnosis to death, years	6.6 ± 3.1	5.1 ± 2.9	< 0.001
Age at death, years	78.6 ± 5.4	86.3 ± 3.9	< 0.001
ChEI dose[a]	66% ± 18%	61% ± 17%	0.029

[a]Mean percentage of the maximum recommended dose, i.e., 10 mg for donepezil, 12 mg for rivastigmine and 24 mg for galantamine.
AD, Alzheimer's disease; APOE, apolipoprotein E; ChEI, cholinesterase inhibitor; MMSE, Mini-Mental State Examination; NHP, nursing home placement.

Figure 1.

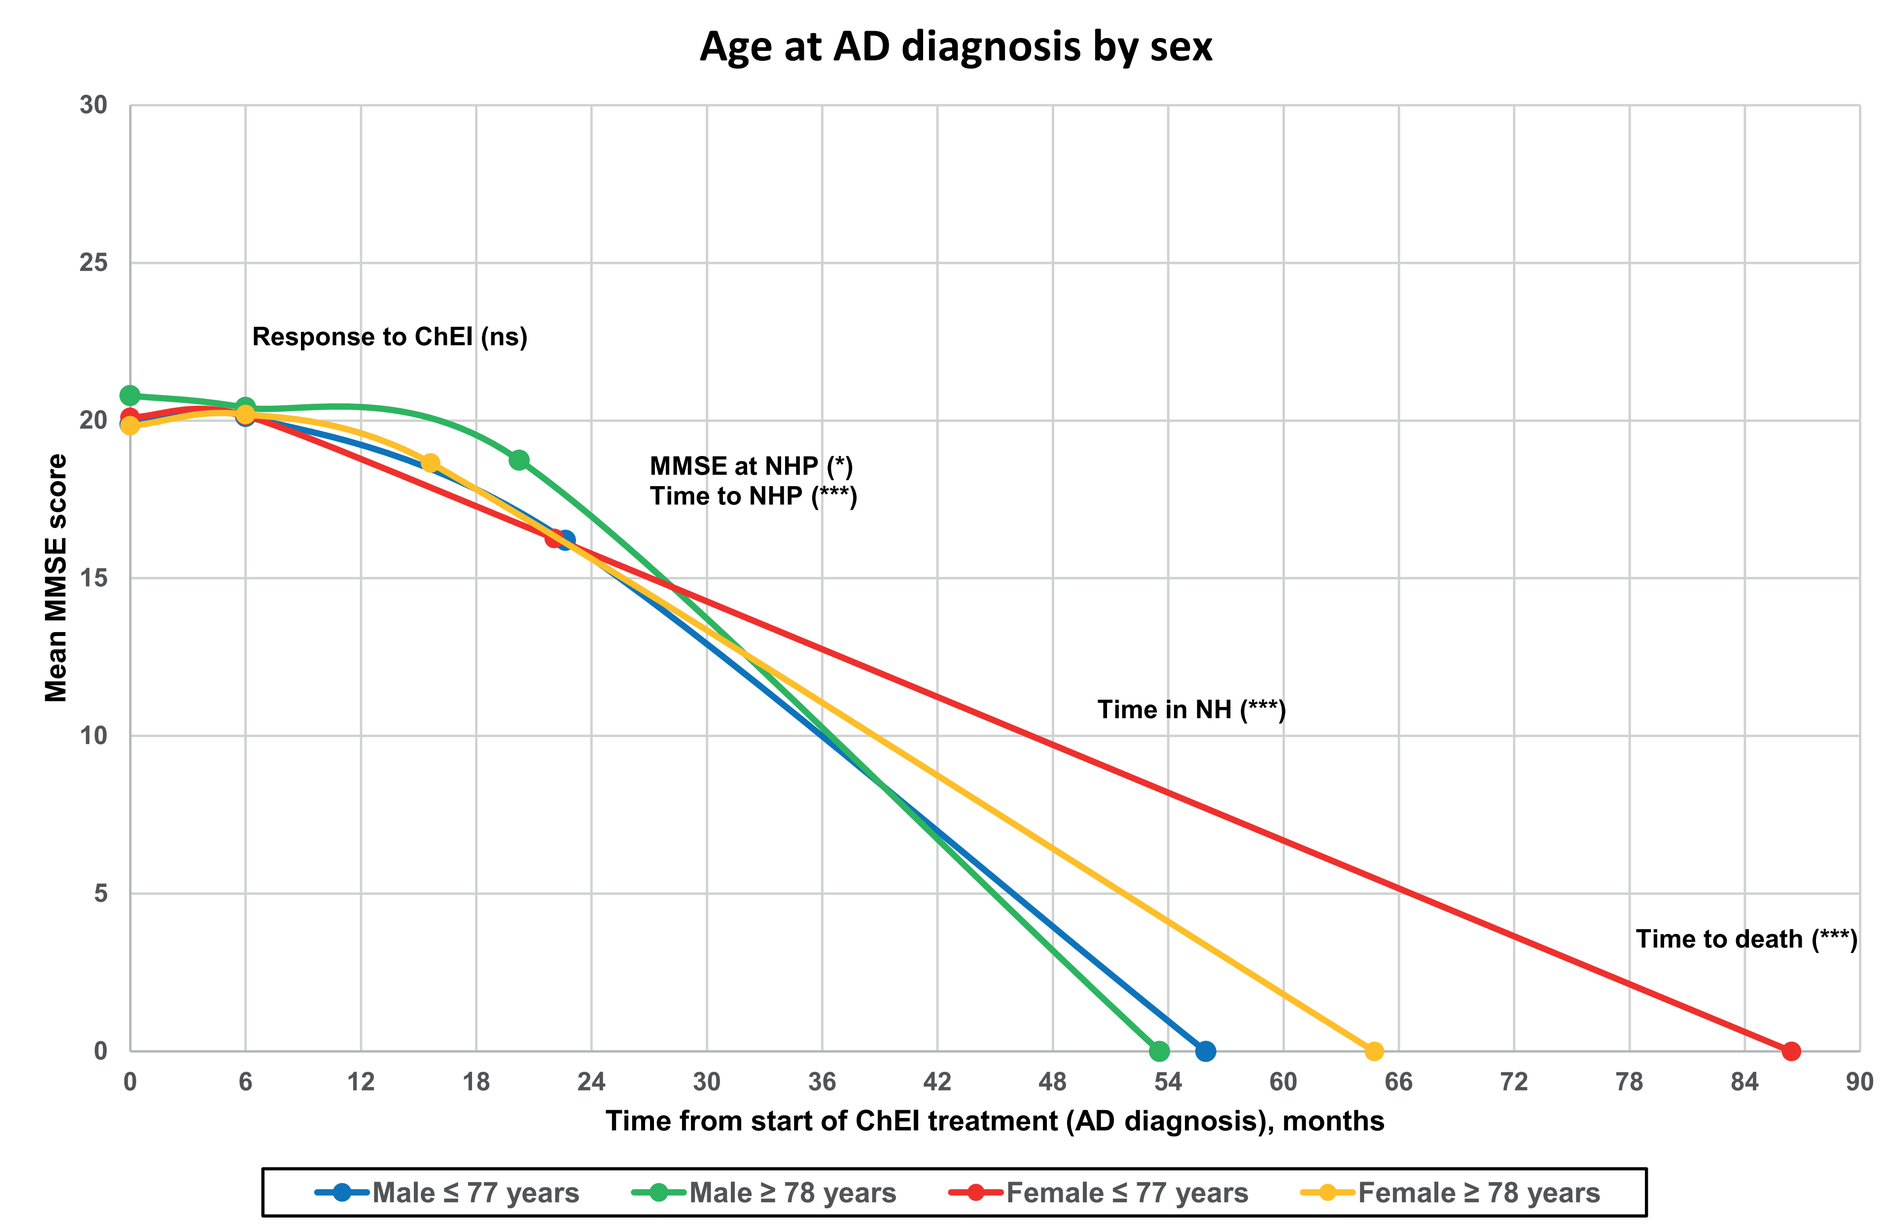
Age at AD diagnosis by sex
Mean MMSE score
Response to ChEI (ns)
MMSE at NHP (*)
Time to NHP (***)
Time in NH (***)
Time to death (***)
Time from start of ChEI treatment (AD diagnosis), months
Male ≤ 77 years
Male ≥ 78 years
Female ≤ 77 years
Female ≥ 78 years

Age was divided by median cut-off. * 0.01 ≤ p < 0.05, ** 0.001 ≤ p < 0.01, *** p < 0.001.
AD, Alzheimer's disease; ChEI, cholinesterase inhibitor; MMSE, Mini-Mental State Examination; NH, nursing home; NHP, nursing home placement.

No differences in cognitive status at AD diagnosis were found among the four groups. Female patients ≤77 years old at baseline had lower cognitive ability at NHP than their older counterparts: mean MMSE score (95% confidence interval [CI]), 16.3 (14.9–17.7) points vs. 18.7 (17.6–19.7) points, $p = 0.018$. Females ≥78 years old exhibited shorter time to NHP than their younger counterparts: 15.6 (13.6–17.7) months vs. 22.1 (19.9–24.2) months, $p < 0.001$. These differences were not observed among the males. A longer survival time in NHs was observed among females ≤77 years old (5.4 [4.7–6.0] years) compared with the other groups: females ≥78 years old, (4.1 [3.4–4.7] years), males ≤77 years old, (2.8 [1.7–3.8] years) and males ≥78 years old, (2.8 [2.0–3.5] years, $p < 0.001$). Younger females also demonstrated a longer life expectancy from AD diagnosis (7.2 [6.5–7.9] years) than the other groups: females ≥78 years old (5.4 [4.7–6.1] years), males ≤77 years old (4.7 [3.6–5.7] years) and males ≥78 years old (4.5 [3.7–5.2] years, $p < 0.001$).

Figure 2.

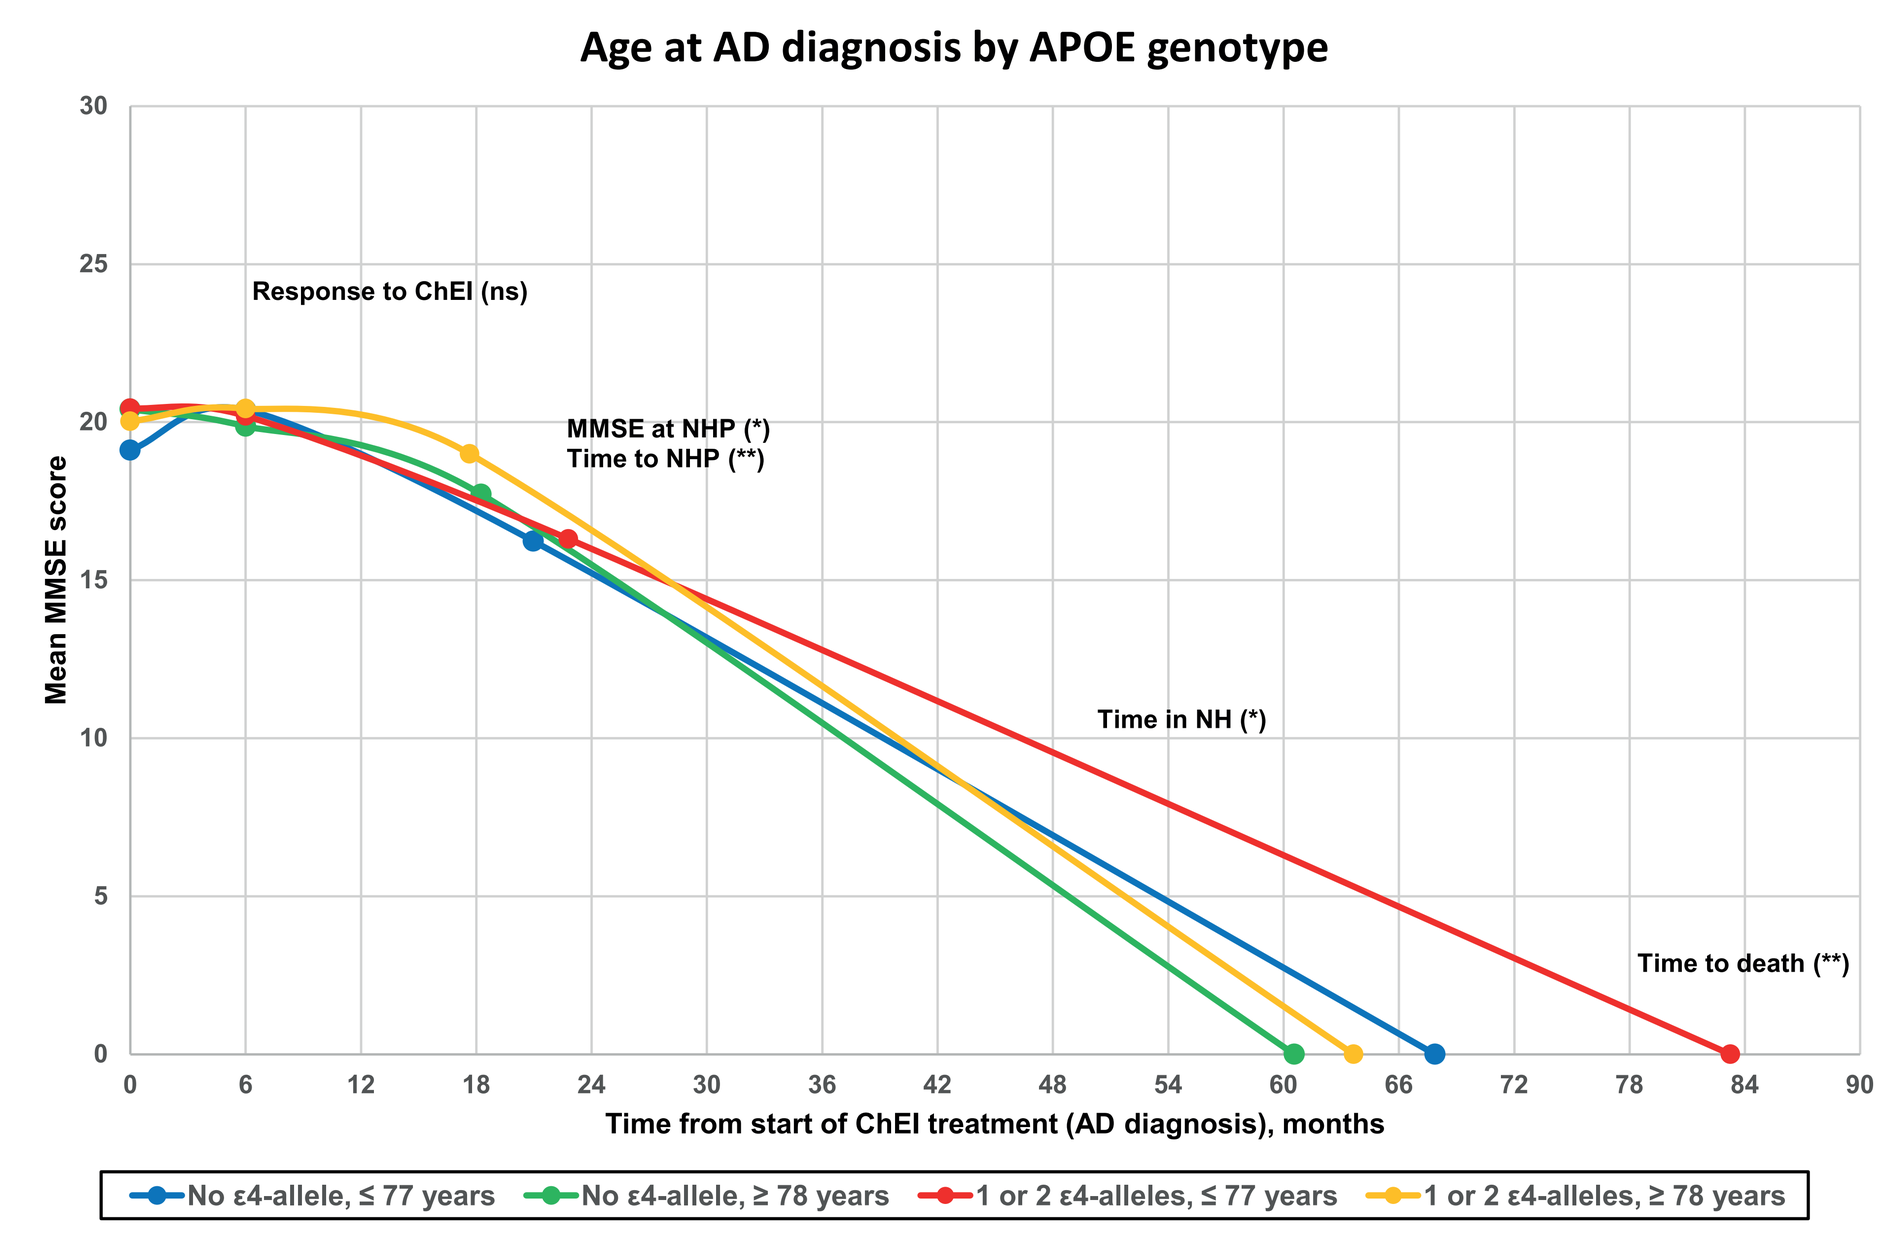
Age at AD diagnosis by APOE genotype
Mean MMSE score
Response to ChEI (ns)
MMSE at NHP (*)
Time to NHP (**)
Time in NH (*)
Time to death (**)
Time from start of ChEI treatment (AD diagnosis), months
No ε4-allele, ≤ 77 years
No ε4-allele, ≥ 78 years
1 or 2 ε4-alleles, ≤ 77 years
1 or 2 ε4-alleles, ≥ 78 years

Age was divided by median cut-off. * 0.01 ≤ p < 0.05, ** 0.001 ≤ p < 0.01, *** p < 0.001.
AD, Alzheimer's disease; APOE, apolipoprotein E; ChEI, cholinesterase inhibitor; MMSE, Mini-Mental State Examination; NH, nursing home; NHP, nursing home placement.

Regarding APOE ε4-carriers, the patients ≤77 years old had lower cognitive ability at NHP than those ≥78 years old: mean MMSE score (95% CI), 16.3 (14.9–17.7) points vs. 19.0 (17.9–20.1) points, $p = 0.021$. Younger ε4-carriers exhibited longer time to NHP than their older counterparts: 22.8 (20.8–24.8) months vs. 17.7 (15.4–19.9) months, $p = 0.006$. A longer survival time in NHs was also observed among the APOE ε4-carriers ≤77 years old compared with those ≥78 years old: 5.0 (4.4–5.7) years vs. 3.8 (3.2–4.5) years, $p = 0.030$. Moreover, younger ε4-carriers showed a longer life expectancy from AD diagnosis than their older counterparts: 6.9 (6.3–7.6) years vs. 5.3 (4.6–6.0) years, $p = 0.002$. The above-mentioned differences were not detected among the non-carriers of APOE ε4. No sex differences were observed among the four groups.

Figure 3.

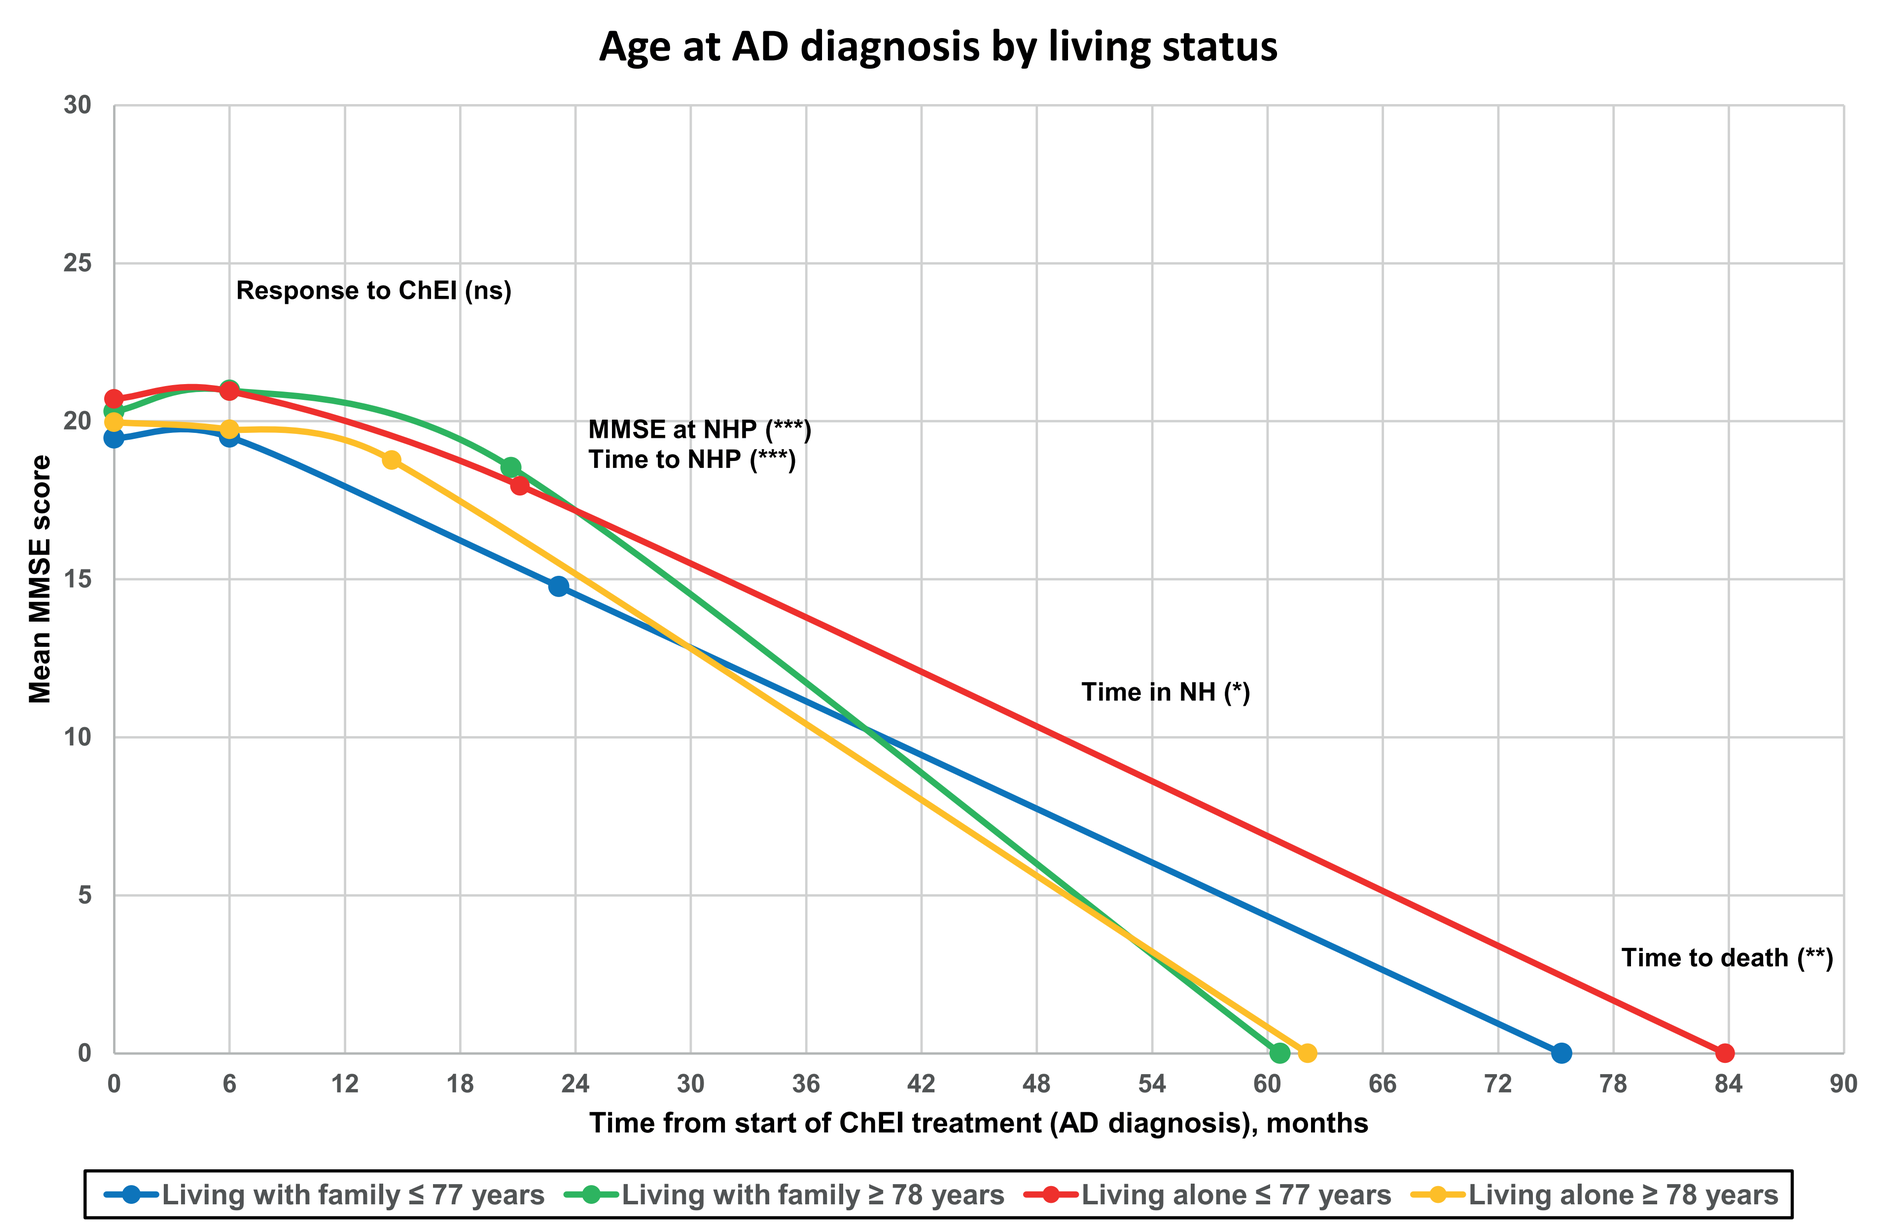
Age at AD diagnosis by living status
Mean MMSE score
Response to ChEI (ns)
MMSE at NHP (***)
Time to NHP (***)
Time in NH (*)
Time to death (**)
Time from start of ChEI treatment (AD diagnosis), months
Living with family ≤ 77 years
Living with family ≥ 78 years
Living alone ≤ 77 years
Living alone ≥ 78 years

Age was divided by median cut-off. * 0.01 ≤ p < 0.05, ** 0.001 ≤ p < 0.01, *** p < 0.001.
AD, Alzheimer's disease; ChEI, cholinesterase inhibitor; MMSE, Mini-Mental State Examination; NH, nursing home; NHP, nursing home placement.

Of the 119 participants living alone at baseline, 100 (84%) were women, and of the 105 patients living with family, 65 (62%) were women ($p < 0.001$). A lower cognitive ability at NHP was observed among individuals ≤77 years old living with family (mean MMSE score [95% CI], 14.8 [12.9–16.7] points) compared with the other groups: ≥78 years old living with family, (18.5 [17.1–19.9] points); ≤77 years old living alone (18.0 [16.6–19.3] points) and ≥78 years old living alone (18.8 [17.6–20.0] points, $p < 0.001$). The solitary-living patients ≥78 years old demonstrated a shorter time to NHP (14.4 [12.1–16.8] months) than the other groups: ≤77 years old living with family (23.1 [21.0–25.3] months), ≥78 years old living with family (20.6 [18.1–23.2] months) and ≤77 years old living alone (21.1 [18.1–24.1] months, $p < 0.001$). The younger solitary-living group exhibited a longer life expectancy from AD diagnosis than the older individuals living alone: 7.0 (6.1–7.9) years vs. 5.2 (4.5–5.9) years, $p = 0.002$; however, this difference was not detected among patients living with family.

Figure 4.

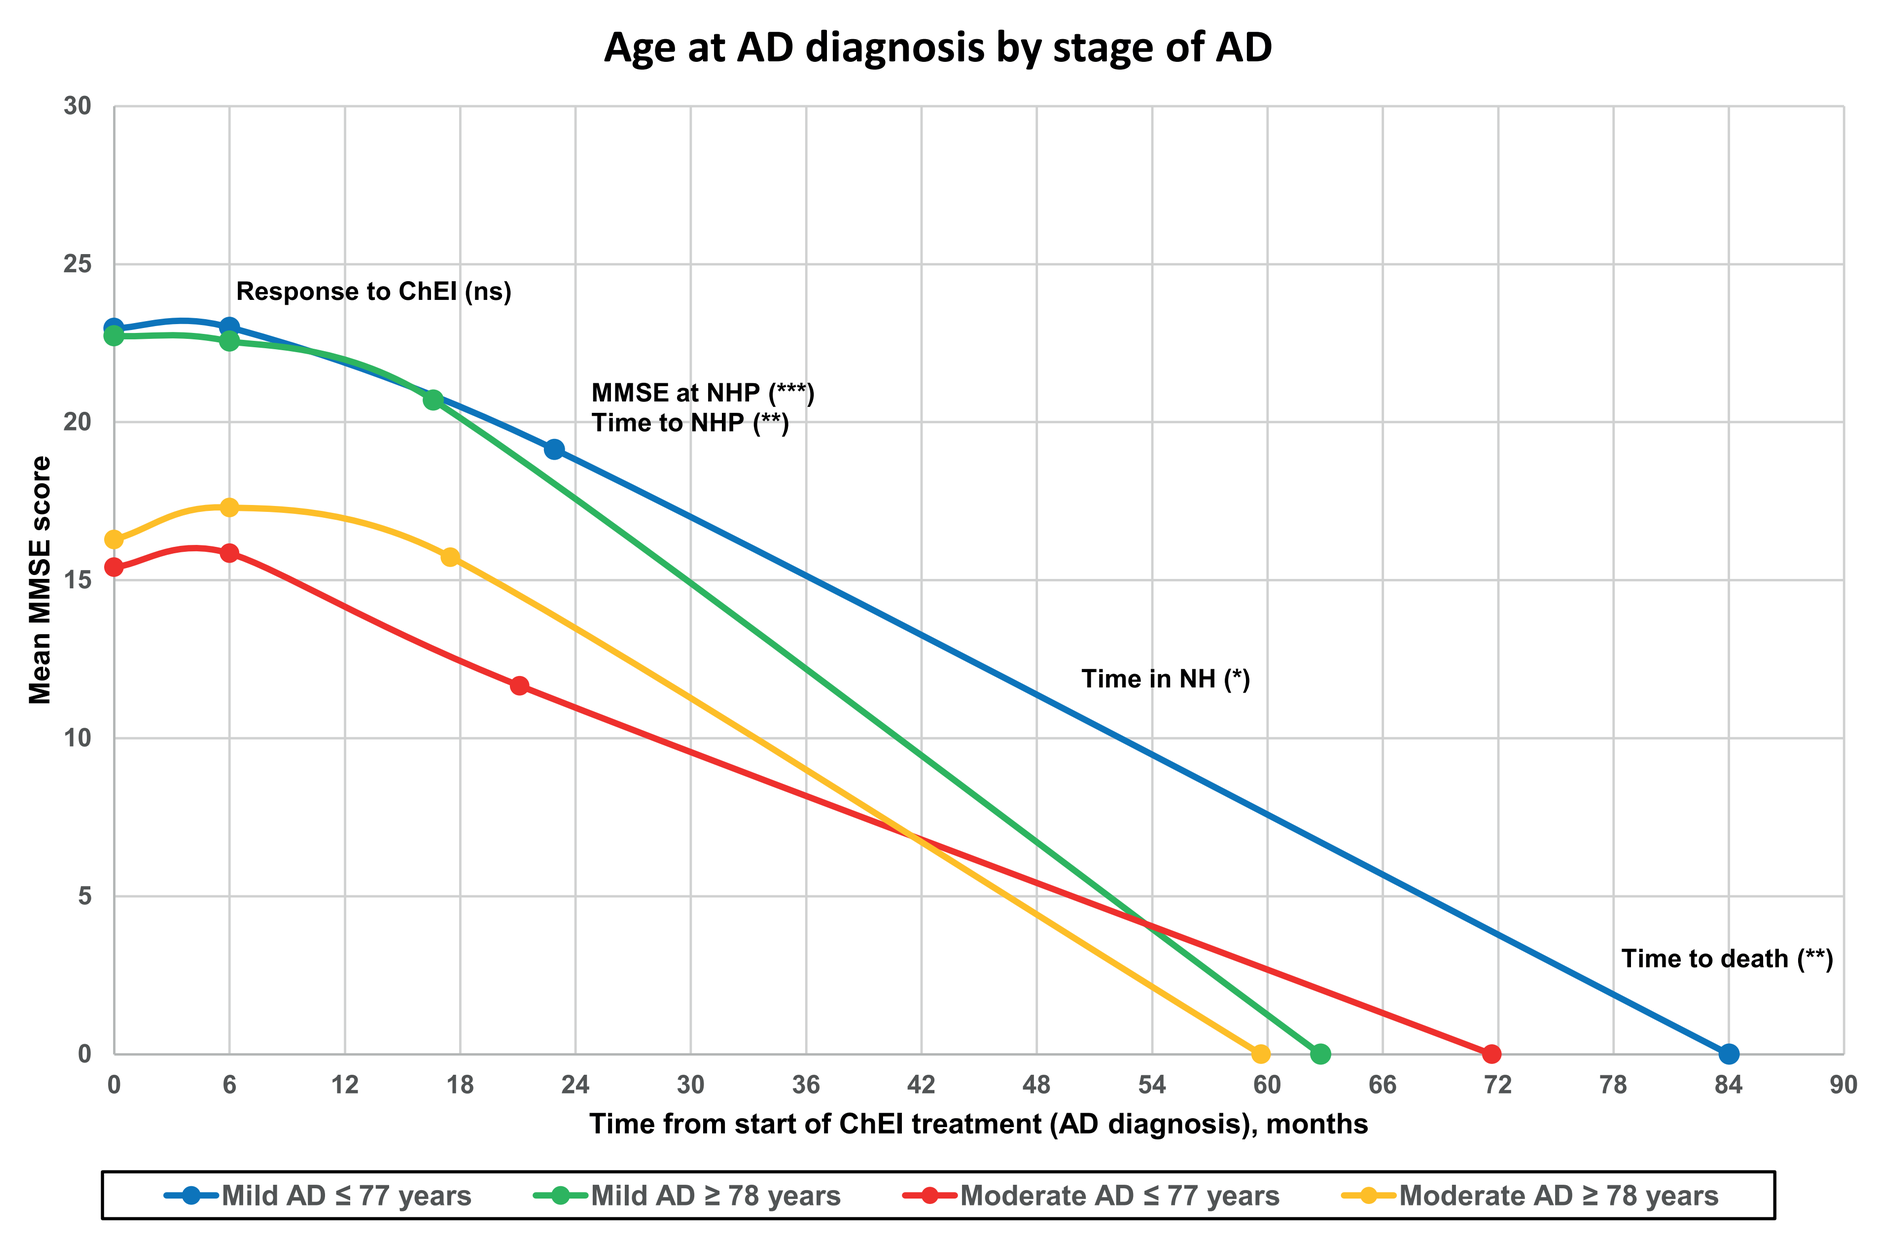
Age at AD diagnosis by stage of AD
Mean MMSE score
Response to ChEI (ns)
MMSE at NHP (***)
Time to NHP (**)
Time in NH (*)
Time to death (**)
Time from start of ChEI treatment (AD diagnosis), months
Mild AD ≤ 77 years
Mild AD ≥ 78 years
Moderate AD ≤ 77 years
Moderate AD ≥ 78 years

Age was divided by median cut-off. * 0.01 ≤ p < 0.05, ** 0.001 ≤ p < 0.01, *** p < 0.001.
AD, Alzheimer's disease; ChEI, cholinesterase inhibitor; MMSE, Mini-Mental State Examination; NH, nursing home; NHP, nursing home placement.

In patients with moderate AD (MMSE score 10–19), those ≤77 years old had lower cognitive ability at NHP than those ≥78 years old: (mean MMSE score [95% CI], 11.7 [9.8–13.5] points vs. 15.7 [14.2–17.2] points, $p < 0.001$); however, this difference was not detected among participants with mild AD (MMSE score 20–26). A longer time to NHP was observed for younger compared with older patients with mild AD: 22.9 (20.7–25.1) months vs. 16.6 (14.3–18.9) months, $p = 0.001$. In mild AD, individuals ≤77 years old also exhibited a longer survival time in NHs than those ≥78 years old: 5.1 (4.3–5.9) years vs. 3.9 (3.2–4.5) years, $p = 0.025$. Moreover, younger participants with mild AD had a longer life-span from AD diagnosis than their older counterparts: 7.0 (6.2–7.8) years vs. 5.2 (4.5–5.9) years, $p = 0.001$; however, these differences were not detected among patients with moderate AD. No sex differences were observed among the four groups.

Layout by Coure AB | www.coure.se