LUND UNIVERSITY

Prevalence of Lewy body dementia and neuroleptic treatment in nursing homes in Malmö, Sweden.

Zahirovic, Iris; Wattmo, Carina; Minthon, Lennart; Londos, Elisabet

DOI:
10.1016/j.jalz.2014.05.1224

2014

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Zahirovic, I., Wattmo, C., Minthon, L., & Londos, E. (2014). *Prevalence of Lewy body dementia and neuroleptic treatment in nursing homes in Malmö, Sweden.*. Poster session presented at Alzheimer's Association International Conference (AAIC), 2014, Copenhagen, Denmark. https://doi.org/10.1016/j.jalz.2014.05.1224

Total number of authors:
4

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
• Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
• You may not further distribute the material or use it for any profit-making activity or commercial gain
• You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

Prevalence of Lewy body dementia symptoms and Neuroleptic treatment in nursing homes in Malmö, Sweden

Lund University
Faculty of Medicine

Iris Zahirovic, Carina Wattmo, Lennart Minthon, Elisabet Londos,
Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, Sweden.

Dept of Clinical Sciences Malmö
Clinical Memory Research Unit

Background:

Dementia of Lewy body (DLB), is a neurocognitive disorder with hallmarks as Parkinsonism, visual hallucinations often with insight, fluctuating alertness and wakefulness, rapid eye movement (REM) sleep behavior disorder (RBD) with vivid dreams and violent movements and neuroleptic hypersensitivity. Nevertheless, DLB is often misdiagnosed and under-recognized.

A diagnosis of DLB is important because of the risk of hypersensitivity for neuroleptic drugs. Moreover, appropriate treatment of symptoms can improve quality of life considerably for both the individual with DLB and their caregivers.

This study aimed to investigate the prevalence of possible DLB symptoms and neuroleptic treatment in elderly care recipients in nursing homes.

Methods:

Information regarding prevalence of symptoms of DLB and concurrent medication was collected in 635 elderly living in 40 nursing homes in Malmö, Sweden during the time period 2012-13.

We used a questionnaire, electronic medication lists and interviewed the nursing staff. In the questionnaire, symptoms representing 4 of the DLB main characteristics were noted as present/absent; Parkinsonism, visual hallucinations, excessive daytime sleepiness and acting out dreams. Neuroleptic treatment was also noted as present or not, as well as the type of drug.

Conclusions:

Symptoms consistent with DLB are common in elderly in nursing homes.
Despite the recommendations of avoiding neuroleptic treatment this was a common finding among the studied individuals. To minimise inappropriate medical treatment recognizing symptoms of DLB are important for general practitioners.

► The Questionnaire

THE RESIDENT HAS THE FOLLOWING:	YES	NO
Parkinson or Parkinson´s like disorder		
Rigidity or stiffness. Tremor. Shuffling walk		
Weak voice		
Balance problems or repeated falls		
Visual hallucinations (seeing things that are not really present)		
Excessive daytime sleepiness		
Acting out dreams during sleep (sometimes violently) Nightmares.		
Antiparkinson medication, for example L-Dopa		
Antipsychotic treatment for example Haloperidol or Risperidon		
Earlier stroke? Brain tumour?		

► DLB symptoms in nursing home residents in Malmö, Sweden

DLB symptoms	All elderly N = 590-617 (%)
Parkinson diagnose	37 (6%)
Rigidity	202 (33%)
Weak voice	133 (22%)
Impaired balance	244 (41%)
Visual hallucinations	123 (20%)
Daytime sleepiness	232 (38%)
RBD like sleep disorder	55 (9%)

Results:

Men 160 (25%) women 475 (75%)
Mean age 86±7 years

In total 28% had ≥2 of 4 DLB symptoms

Neuroleptic treatment was reported in 22%

Neuroleptic treatment increased significantly ($p<0.002$) with increasing number of DLB symptoms

9% of the neuroleptics prescribed were of acceptable type such as klozapin and quetiapin

► Neuroleptic treatment in relation to number of DLB symptoms

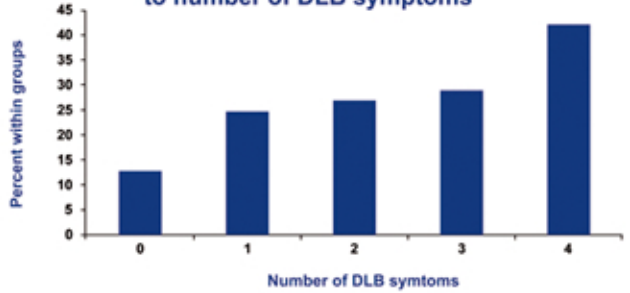
Percent within groups
45
40
35
30
25
20
15
10
5
0
0
1
2
3
4
Number of DLB symtoms