LUND UNIVERSITY

Dementia with Lewy Body (DLB) Symptoms Hidden within the Diagnosis “Dementia Not Otherwise Specified” a Cross-Sectional Study in 40 Swedish Nursing Homes.

Zahirovic, Iris; Londos, Elisabet; Torisson, Gustav; Wattmo, Carina

2017

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Zahirovic, I., Londos, E., Torisson, G., & Wattmo, C. (2017). *Dementia with Lewy Body (DLB) Symptoms Hidden within the Diagnosis “Dementia Not Otherwise Specified” a Cross-Sectional Study in 40 Swedish Nursing Homes.*. Poster session presented at Alzheimer's Association International Conference (AAIC), 2017, London, United Kingdom.

Total number of authors:
4

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
- Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
- You may not further distribute the material or use it for any profit-making activity or commercial gain
- You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

Download date: 25. Jul. 2024

LUND UNIVERSITY
Faculty of Medicine

Core Clinical Features of Dementia with Lewy Body (DLB) hidden within Different Dementia Diagnoses; a Cross-Sectional Study in 40 Swedish Nursing Homes

Iris Zahirovic, MD[1]; Carina Wattmo, RN, BSc, PhD[1]; Gustav Torisson, PhD, MD[2] and Elisabet Londos, MD, PhD[1]
Memory Clinic, Skåne University Hospital, Malmö Sweden, Clinical Memory Research Unit, Malmö, Sweden

Background

DLB is a neurocognitive disorder with core clinical features; such as Parkinsonism, visual hallucinations, fluctuating cognition/ excessive daytime sleepiness and rapid eye movement sleep behavior disorder (RBD) (1, 2). DLB is often misdiagnosed and unrecognized in elderly individuals. A diagnosis of DLB is important because of the risk of antipsychotic hypersensitivity, and the benefit of cholinesterase inhibitor treatment (3, 4). Moreover, appropriate treatment of symptoms can improve quality of life considerably for both the individual with DLB and their caregivers.

We hypothesized that we would find potential DLB signs in nursing home (NH) residents, hidden within different dementia diagnoses besides DLB. We also hypothesized a difference in treatment between these different diagnoses, mostly in the usage of antidementia medication.

Methods

Between 2012 and 2013, we applied a specially designed questionnaire that covered clinical DLB signs on the 40 Swedish NHs. **The questionnaire,** was designed to cover the main DLB core clinical features (visual hallucinations, Parkinsonism, fluctuations in attention and wakefulness and RBD), according to DLB consensus criteria from 2005.The nursing staffs completed the questionnaires after receiving specifically designed training. We also collected **computerized medical lists** from the Swedish National Medication Dispensing System & hospital medical records (5).

▶ Table 1: The Pharmacological treatment in the different diagnoses

Diagnoses N (%)	The Study Population * Nursing Home residents N (%)	DLB0-1 N (%)	DLB2-4 N (%)	Antidementia Medication N (%)	Antipsychotic Medication N (%)
AD	115 (26)	92 (80)	23 (20)	71 (64)	30 (26)
AD + AD Mix	97 (22)	83 (86)	14 (14)	66 (69)	25 (26)
VaD	85 (19)	69 (81)	16 (19)	5 (6)	20 (24)
DLB / PDD	22 (5)	2 (9)	20 (91)	13 (62)	5 (23)
Dementia NOS	121 (28)	102 (84)	19 (16)	37 (32)	22 (18)

* Nursing homes residents were: 610 (all residents) and 440 (residents with dementia diagnosis) Alzheimer's disease (AD), Alzheimer's mixed (AD-Mix),Vascular dementia (VaD), Dementia with Lewy bodies (DLB) and Parkinson's disease dementia (PDD).

Results

Participants: 650 nursing home residents, 610 (94%) were included (595 medical records).
The mean age was 86.0 ± 7.5 years; 467 (75%) were women.

2-4 of the main DLB signs were found in 20-29% of the elderly with different dementia diagnoses (beside the DLB/PDD diagnosis). In the residents with a DLB/PDD diagnosis, 91% had 2-4 main DLB signs (Fig.3).

The dementia diagnoses of the residents according to the medical records; AD in 115 (19%), AD-Mix in 97(16%), VaD in 85 (14%) and DLB/PDD in 22 (5%), Dementia NOS in 121 (20%) and no formal dementia diagnosis in 155 (26%).

The antidementia medication did not differ between residents with 0-1 DLB (32%) and 2-4 DLB (37%) signs. The antidementia medication among different dementia diagnoses varied. In the residents with a Dementia NOS diagnosis, the prevalence of cholinesterase inhibitor or memantine treatment was only about half (32%), compared to the other dementia diagnoses (62-69%) ($p<0.001$).

The antipsychotic treatment was found in 32% of the residents with 2-4 DLB signs and in 19% of those with 0-1 DLB signs ($p=0.003$). However, the antipsychotic treatment varied between 18-26% but did not differ between the dementia diagnoses.

▶ Fig. 1: The unsuitable Pharmacological treatment *

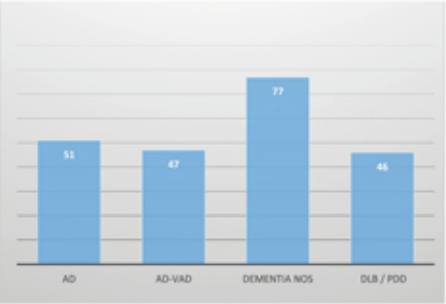
51
47
77
46
AD
AD-VAD
DEMENTIA NOS
DLB / PDD

▶ Fig. 2: The Study Population

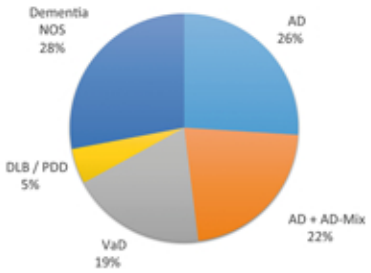
Dementia NOS 28%
AD 26%
AD + AD-Mix 22%
VaD 19%
DLB / PDD 5%

Alzheimer´s disease (AD), Alzheimer´s mixed (AD-Mix), Vascular dementia (VaD), Dementia with Lewy bodies (DLB) and Parkinson´s disease dementia (PDD).
* Unsuiatable pharmacological treatment definition; none anti-dementia medication and existing antipsychotic treatment.

Conclusions

We conclude that about one fourth of the residents with a dementia diagnosis, beside DLB/PDD, had two or more of the four main DLB signs.
Residents with increasing number of main DLB signs are at a higher risk of receiving antipsychotic treatment. Residents without a specified dementia diagnosis have the least beneficial treatment. This emphasizes the importance of identification of specific dementia diagnoses, recognition of DLB sign and suitable treatment.

▶ Fig. 3: Core Clinical DLB Features among Different Dementia Diagnoses inclusive DLB/PDD

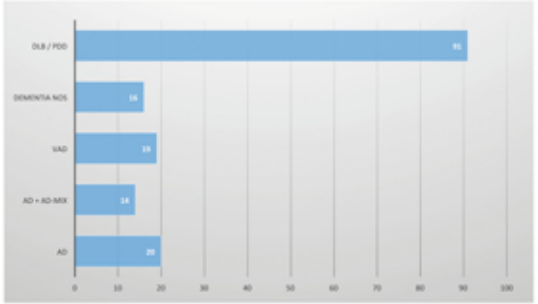
DLB / PDD
DEMENTIA NOS
VAD
AD + AD-MIX
AD
0
10
20
30
40
50
60
70
80
90
100

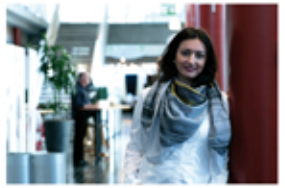

Iris Zahirovic, MD
iris.zahirovic@med.lu.se
Memory Clinic, Skåne University Hospital
SE-205 02, Malmö, Sweden

References

1. McKeith IG, Dickson DW, Lowe J, Emre M, O'Brien JT, Feldman H, et al. Diagnosis and management of dementia with Lewy bodies: third report of the DLB Consortium. Neurology. 2005;65(12):1863-72.
2. McKeith IG, Boeve BF, Dickson DW, Halliday G, Taylor JP, Weintraub D, et al. Diagnosis and management of dementia with Lewy bodies: Fourth consensus report of the DLB Consortium. Neurology. 2017.
3. McKeith I, Fairbairn A, Perry R, Thompson P, Perry E. Neuroleptic sensitivity in patients with senile dementia of Lewy body type. Bmj. 1992;305(6855):673-8.
4. McKeith I, Del Ser T, Spano P, Emre M, Wesnes K, Anand R, et al. Efficacy of rivastigmine in dementia with Lewy bodies: a randomised, double-blind, placebo- controlled international study. Lancet. 2000;356(9247):2031-6.
5. Zahirovic I, Wattmo C, Torisson G, Minthon L, Londos E. Prevalence of Dementia With Lewy Body Symptoms: A Cross-Sectional Study in 40 Swedish Nursing Homes. Journal of the American Medical Directors Association. 2016;17(8):706-11.