LUND UNIVERSITY

Prognostic regression models of life expectancy after diagnosis of Alzheimer’s disease – a 20-year follow-up.

Wattmo, Carina; Londos, Elisabet

2019

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Wattmo, C., & Londos, E. (2019). *Prognostic regression models of life expectancy after diagnosis of Alzheimer’s disease – a 20-year follow-up.*. Poster session presented at Alzheimer's Association International Conference (AAIC), 2019, Los Angeles, United States.

Total number of authors:
2

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
• Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
• You may not further distribute the material or use it for any profit-making activity or commercial gain
• You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

Prognostic regression models of life expectancy after diagnosis of Alzheimer's disease – a 20-year follow-up

Carina Wattmo and Elisabet Londos

Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, Sweden.

CONCLUSIONS

The survival time after a diagnosis of Alzheimer's disease (AD) could be predicted with a high degree of explanation using multivariate regression models including sociodemographic and clinical factors. The level of certainty in the models increased when including activities of daily living (ADL) performance at baseline, as well as rates of cognitive and functional deterioration, which underlines the importance of assessing those measures over time for a better AD prognosis. A longer life-span of up to ~2 years could be expected for individuals who received and tolerated long-term cholinesterase inhibitor (ChEI) treatment. Our clinically relevant prognostic models can be used to calculate estimated life expectancy in AD.

BACKGROUND

The mean length of life after AD diagnosis varies between 3 and 10 years, mostly depending on the patient's age; however, individuals with AD can live considerably longer, up to 15–20 years. Several sociodemographic, genetic, and disease-related characteristics have been shown to increase survival in AD, such as female sex, a lower education level, the absence of the apolipoprotein E (APOE) ε4 allele, better cognitive ability, and fewer cardiovascular disorders. Reports of potential differences in years of life expectancy according to various protective or risk factors in AD, as well as aspects of ChEI therapy, are scarce. We aimed to determine characteristics that might affect survival time in AD patients treated with ChEI, and create statistical models of estimated life-span after AD diagnosis.

METHODS

The Swedish Alzheimer Treatment Study (SATS) is a prospective, observational, multicenter study to assess the use of long-term ChEI therapy in a routine clinical setting. The SATS enrolled 1,021 participants with a clinical diagnosis of mild-to-moderate AD (Mini-Mental State Examination [MMSE] score, 10–26) at the start of ChEI treatment (shortly after AD diagnosis). Of these, 966 individuals (95%) had died after up to 20 years of follow-up and were included in this presentation (Table 1). Multivariate linear regression models were used to identify the patients' characteristics that independently affected the survival time from diagnosis to death (Tables 2 and 3).

The following potential predictors were investigated: sex, APOE genotype, solitary living, duration of AD, age at baseline, years of education, type of ChEI, dose of ChEI, length of ChEI therapy in the SATS, specific concomitant medications (antihypertensive/cardiac therapy, antidiabetic drugs, asthma medication, thyroid therapy, lipid-lowering agents, estrogens, nonsteroidal anti-inflammatory drugs/acetylsalicylic acid, antidepressants, antipsychotics, and anxiolytics/sedatives/hypnotics), cognition (MMSE), and ADL (Instrumental Activities of Daily Living scale [IADL] and Physical Self-Maintenance Scale [PSMS]) at baseline, and their rates of decline.

Cognitive status and instrumental and basic ADL were evaluated at baseline and semiannually over 3 years, and the date of death was recorded. The choice of ChEI agent and all decisions regarding dosage for each individual patient were left entirely to the discretion and professional judgment of dementia specialists.

Two types of multivariate regression models were created, one using only sociodemographic and clinical characteristics at baseline and another model that included baseline and longitudinal measures. Both main models included cognitive ability and the extended models also included ADL capacities, as well as aspects of ChEI treatment in the longitudinal model. *B* values were unstandardized and are expressed per 1 unit increase for continuous variables and for the condition present in categorized variables (Tables 2 and 3).

RESULTS

Table 1. Baseline characteristics (*n* = 966)

Female sex	620 (64%)
APOE ε4 carrier, (*n* = 946)	637 (67%)
Solitary living at baseline	343 (36%)
Antihypertensive/cardiac therapy	395 (41%)
Antidiabetics	50 (5%)
Asthma medication	41 (4%)
Thyroid therapy	82 (8%)
Lipid-lowering agents	104 (11%)
Estrogens	65 (7%)
NSAIDs/acetylsalicylic acid	290 (30%)
Antidepressants	246 (25%)
Antipsychotics	47 (5%)
Anxiolytics/sedatives/hypnotics	142 (15%)
	Mean ± standard deviation
Estimated age at onset, years	72.4 ± 7.2
Estimated duration of AD, years	3.0 ± 1.9
Age at the start of ChEI treatment (baseline), years	75.5 ± 6.8
Education, years	9.4 ± 2.5
MMSE score, range 0–30	21.3 ± 3.7
IADL score, range 8–31	16.1 ± 5.4
PSMS score, range 6–30	7.6 ± 2.3
Mean dose of ChEI during the SATS, mg	
Donepezil (*n* = 511)	6.9 ± 1.8
Rivastigmine (*n* = 202)	6.1 ± 2.1
Galantamine (*n* = 253)	15.2 ± 3.7
Length of ChEI therapy in the SATS, months	23.0 ± 13.0
Time from AD diagnosis to death, years	6.7 ± 3.5
Age at death, years	82.2 ± 6.7

AD, Alzheimer's Disease; APOE, apolipoprotein E; ChEI, cholinesterase inhibitor; IADL, Instrumental Activities of Daily Living scale; MMSE, Mini-Mental State Examination; NSAIDs, nonsteroidal anti-inflammatory drugs; PSMS, Physical Self-Maintenance Scale; SATS, Swedish Alzheimer Treatment Study.

• The arbitrary examples of a patient's estimated life-span after AD diagnosis presented in Tables 2 and 3 were based on a 76-year-old female with 10 years of education, who received antidiabetics and exhibited a MMSE score of 21, an IADL score of 16, and a PSMS score of 8 at baseline. Her mean decline in MMSE score was 3.0 points/year and in PSMS score 1.3 points/year. She received ChEI therapy during 2.2 years in the SATS.

Table 2. Baseline characteristics that affected the patients' survival time after AD diagnosis in years (linear regression models).

	Model 1 including MMSE			Model 2 including MMSE, IADL and basic ADL		
Degree of explained variance	R = 0.389, R^2 = 0.151, *P* < 0.001			R = 0.429, R^2 = 0.184, *P* < 0.001		
Significant predictors	*B* (SE)	*P* value	Example (*B*)	*B* (SE)	*P* value	Example (*B*)
Intercept	8.26 (0.57)	<0.001	8.26	9.11 (0.63)	<0.001	9.11
Sex, female[a]	1.25 (0.22)	<0.001	1.25	1.15 (0.22)	<0.001	1.15
Age at baseline[b]						
65–69 years	-1.48 (0.52)	0.005		-1.27 (0.51)	0.013	
70–74 years	-2.02 (0.45)	<0.001		-1.70 (0.45)	<0.001	
75–79 years	-2.68 (0.43)	<0.001	-2.68	-2.11 (0.43)	<0.001	-2.11
80–84 years	-3.22 (0.45)	<0.001		-2.46 (0.46)	<0.001	
≥85 years	-3.60 (0.59)	<0.001		-2.88 (0.60)	<0.001	
Education, ≥10 years[c]	-0.67 (0.25)	0.009	-0.67	-0.65 (0.25)	0.010	-0.65
Antihypertensive/cardiac therapy[d]	-0.65 (0.22)	0.003		-0.68 (0.22)	0.002	
Antidiabetics[e]	-1.26 (0.49)	0.011	-1.26	-1.08 (0.50)	0.031	-1.08
MMSE score at baseline[f]						
20–23 points	-0.56 (0.25)	0.027	-0.56		ns	0
17–19 points	-1.10 (0.32)	0.001			ns	
≤16 points	-1.81 (0.35)	<0.001		-0.72 (0.33)	0.029	
IADL score at baseline[g]	na					
9–12 points				-1.05 (0.42)	0.014	
13–19 points				-1.70 (0.41)	<0.001	-1.70
≥20 points				-2.01 (0.47)	<0.001	
PSMS score at baseline[h]	na					
7 points					ns	
8–10 points				-0.66 (0.30)	0.028	-0.66
≥11 points				-1.11 (0.42)	0.009	
Estimated life-span after AD diagnosis, years			**4.34**			**4.06**

Apolipoprotein E genotype, solitary living, duration of AD, and specific concomitant medications (asthma medication, thyroid therapy, lipid-lowering agents, estrogens, nonsteroidal anti-inflammatory drugs/acetylsalicylic acid, antidepressants, antipsychotics, and anxiolytics/sedatives/hypnotics) used at the time of AD diagnosis (baseline) were not significant.
Reference categories: [a] male sex, [b] age ≤64 years, [c] ≤9 years of education, [d] no antihypertensive/cardiac therapy, [e] no antidiabetics, [f] 24–26 MMSE points, [g] 8 IADL points (no impairment), [h] 6 PSMS points (no impairment).
AD, Alzheimer's disease; ADL, activities of daily living; IADL, Instrumental Activities of Daily Living scale; MMSE, Mini-Mental State Examination; na, not applicable; ns, not significant; PSMS, Physical Self-Maintenance Scale; SE, standard error.

Table 3. Baseline characteristics, rates of progression and aspects of ChEI therapy that affected the patients' survival time after AD diagnosis in years (linear regression models).

	Model 1 including MMSE			Model 2 including MMSE, IADL and basic ADL			Model 3 including MMSE, IADL, basic ADL and ChEI therapy		
Degree of explained variance	R = 0.447, R^2 = 0.200, *P* < 0.001			R = 0.479, R^2 = 0.229, *P* < 0.001			R = 0.536, R^2 = 0.287, *P* < 0.001		
Significant predictors	*B* (SE)	*P* value	Example (*B*)	*B* (SE)	*P* value	Example (*B*)	*B* (SE)	*P* value	Example (*B*)
Intercept	10.10 (0.45)	<0.001	10.10	9.69 (0.42)	<0.001	9.69	8.24 (0.44)	<0.001	8.24
Sex, female[a]	1.34 (0.22)	<0.001	1.34	1.31 (0.22)	<0.001	1.31	1.26 (0.21)	<0.001	1.26
Age at baseline[b]									
65–69 years	-1.53 (0.52)	0.003		-1.29 (0.51)	0.012		-1.28 (0.49)	0.009	
70–74 years	-2.14 (0.45)	<0.001		-1.72 (0.44)	<0.001		-1.83 (0.42)	<0.001	
75–79 years	-2.82 (0.43)	<0.001	-2.82	-2.14 (0.43)	<0.001	-2.14	-2.08 (0.41)	<0.001	-2.08
80–84 years	-3.45 (0.45)	<0.001		-2.71 (0.45)	<0.001		-2.69 (0.43)	<0.001	
≥85 years	-3.91 (0.59)	<0.001		-3.22 (0.60)	<0.001		-3.56 (0.57)	<0.001	
Education, ≥10 years[c]	-0.55 (0.25)	0.030	-0.55		ns	0		ns	0
Antihypertensive/cardiac therapy[d]	-0.77 (0.22)	<0.001		-0.81 (0.22)	<0.001		-0.78 (0.21)	<0.001	
Antidiabetics[e]	-1.37 (0.49)	0.005	-1.37	-1.29 (0.50)	0.010	-1.29	-1.07 (0.48)	0.025	-1.07
MMSE score at baseline[f]					ns			ns	
20–23 points	-0.57 (0.25)	0.025	-0.57			0			0
17–19 points	-1.07 (0.32)	0.001							
≤16 points	-1.55 (0.35)	<0.001							
MMSE score, rate of mean decline/year[g]									
≥3 to <5 points	-1.12 (0.30)	<0.001	-1.12	-0.68 (0.30)	0.025	-0.68		ns	0
≥5 points	-1.98 (0.31)	<0.001		-1.56 (0.32)	<0.001		-0.69 (0.31)	0.025	
IADL score at baseline[h]	na								
9–12 points					ns			ns	
13–19 points				-0.58 (0.26)	0.029	-0.58	-0.53 (0.25)	0.034	-0.53
≥20 points				-0.76 (0.34)	0.028		-1.06 (0.28)	<0.001	
PSMS score at baseline[i]	na							ns	
7 points					ns				
8–10 points				-0.66 (0.30)	0.026	-0.66			0
≥11 points				-1.14 (0.42)	0.007				
PSMS score, rate of mean decline/year[j]	na								
≥2 to <4 points				-0.61 (0.29)	0.036		-0.75 (0.28)	0.008	
≥4 points				-1.67 (0.37)	<0.001		-1.42 (0.35)	<0.001	
Length of ChEI therapy in the SATS[k]	na			na					
2 to <3 years							0.99 (0.27)	<0.001	0.99
3 years							2.26 (0.24)	<0.001	
Estimated life-span after AD diagnosis, years			**5.01**			**5.65**			**6.81**

Apolipoprotein E genotype, solitary living, duration of AD, specific concomitant medications (asthma medication, thyroid therapy, lipid-lowering agents, estrogens, nonsteroidal anti-inflammatory drugs/acetylsalicylic acid, antidepressants, antipsychotics, and anxiolytics/sedatives/hypnotics) used at the time of AD diagnosis (baseline), rate of IADL decline/year, type of ChEI, and dose of ChEI were not significant.
Reference categories: [a] male sex, [b] age ≤64 years, [c] ≤9 years of education, [d] no antihypertensive/cardiac therapy, [e] no antidiabetics, [f] 24–26 MMSE points, [g] <3 MMSE points/year, [h] 8 IADL points (no impairment), [i] 6 PSMS points (no impairment), [j] <2 PSMS points/year, [k] <2 years of ChEI therapy.
AD, Alzheimer's disease; ADL, activities of daily living; ChEI, cholinesterase inhibitor; IADL, Instrumental Activities of Daily Living scale; MMSE, Mini-Mental State Examination; na, not applicable; ns, not significant; PSMS, Physical Self-Maintenance Scale; SATS, Swedish Alzheimer Treatment Study; SE, standard error.

CONTACT ADDRESS:

Carina Wattmo, RN, BSc, PhD, Clinical Memory Research Unit,
Department of Clinical Sciences, Malmö,
Lund University, SE-205 02 Malmö, Sweden.
Tel +46 40 33 56 01, Fax +46 40 33 56 57
E mail: carina.wattmo@skane.se

Poster presented at the Alzheimer's Association International Conference,
Los Angeles, CA, USA; July 14-18, 2019.