LUND UNIVERSITY

Predictors of mortality in early- versus late-onset Alzheimer’s disease – an 18-year follow-up.

Wattmo, Carina; Londos, Elisabet

2018

Document Version:
Publisher's PDF, also known as Version of record

Link to publication

Citation for published version (APA):
Wattmo, C., & Londos, E. (2018). *Predictors of mortality in early- versus late-onset Alzheimer’s disease – an 18-year follow-up.*. Poster session presented at Alzheimer’s Association International Conference (AAIC), 2018, Chicago, Illinois, United States.

Total number of authors:
2

General rights
Unless other specific re-use rights are stated the following general rights apply:
Copyright and moral rights for the publications made accessible in the public portal are retained by the authors and/or other copyright owners and it is a condition of accessing publications that users recognise and abide by the legal requirements associated with these rights.
- Users may download and print one copy of any publication from the public portal for the purpose of private study or research.
- You may not further distribute the material or use it for any profit-making activity or commercial gain
- You may freely distribute the URL identifying the publication in the public portal

Read more about Creative commons licenses: https://creativecommons.org/licenses/

Take down policy
If you believe that this document breaches copyright please contact us providing details, and we will remove access to the work immediately and investigate your claim.

LUND UNIVERSITY

PO Box 117
221 00 Lund
+46 46-222 00 00

Download date: 24. Sep. 2024

PREDICTORS OF MORTALITY IN EARLY- VERSUS LATE-ONSET ALZHEIMER'S DISEASE – AN 18-YEAR FOLLOW-UP

Carina Wattmo and Elisabet Londos

Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, Sweden

CONCLUSIONS

Predictors of mortality differed between patients with early-onset Alzheimer's disease (EOAD) and late-onset Alzheimer's disease (LOAD). More impaired instrumental activities of daily living (IADL), but not cognitive performance, was a risk factor for worse prognosis in EOAD. Solitary-living younger males exhibited nearly a threefold risk of death compared with corresponding males living with a family. In LOAD, demographic factors (male sex irrespective of living status and older age), comorbidities (cardiovascular and diabetes), lower cognitive status at baseline, and more pronounced progression rate in cognition had independent significant impact on shorter survival time. Faster annual deterioration in basic activities of daily living (ADL) and hence the consequences of loss of essential functions predicted a shorter life expectancy in both age groups.

BACKGROUND

Patients with Alzheimer's disease (AD) have a higher risk of death than the general population, with a reported mean life-span between 5 and 10 years after AD diagnosis. The prognostic factors of survival may be different for individuals with EAOD, who are younger and usually have less comorbidity and disabilities, but might be prone to a more hereditary and aggressive course of the disease. Few studies have focused on predictors of life expectancy in patients with EOAD compared with LOAD. We aimed to investigate the effect of genetic, sociodemographic, and clinical factors on mortality in the two age groups.

METHODS

The Swedish Alzheimer Treatment Study (SATS) is a prospective, observational, multicenter study for longitudinal assessment of cholinesterase inhibitor (ChEI) therapy in clinical practice involving 1,021 participants diagnosed with mild-to-moderate AD (Mini-Mental State Examination score, 10–26) at the start of ChEI treatment (time of AD diagnosis). Of these, 143 were defined as having EOAD (onset <65 years), 874 LOAD (onset ≥65 years), and four missing age-at-onset; thus, 1,017 patients were included. As shown in Tables 1 and 2, *t*-tests were performed to analyze two independent groups and χ^2 tests were conducted to analyze categorical variables. Cox proportional hazards regression was used to determine characteristics that affected the time from AD diagnosis to death: sex, apolipoprotein E genotype, solitary living, duration of AD, age at baseline, years of education, specific concomitant medications (antihypertensive/cardiac therapy, antidiabetic drugs, asthma medication, thyroid therapy, lipid-lowering agents, estrogens, nonsteroidal anti-inflammatory drugs/acetylsalicylic acid, antidepressants, antipsychotics, and anxiolytics/sedatives/hypnotics), cognition (Alzheimer's Disease Assessment Scale–cognitive subscale [ADAS-cog]), and ADL (Instrumental Activities of Daily Living scale [IADL] and Physical Self-Maintenance Scale [PSMS]) at baseline, and rate of decline (Table 3). In Figure 2, a Kaplan–Meier graph with a log–rank test was used to illustrate the differences in survival time. One-way analysis of variance (ANOVA) with Bonferroni correction was used to compare the differences between the means (Figures 2–4).

Contact address:
Carina Wattmo, RN, BSc, PhD, Clinical Memory Research Unit, Department of Clinical Sciences, Malmö, Lund University, SE-205 02 Malmö, Sweden.
Tel +46 40 33 56 01, Fax +46 40 33 56 57, E mail: carina.wattmo@skane.se

Poster presented at the Alzheimer's Association International Conference, Chicago, IL, USA; July 22-26, 2018.

RESULTS

Table 1. Early-onset AD, baseline characteristics (n = 143)

	Deceased	Still alive	*P* value
Number of patients (n / %)	115 / 80%	28 / 20%	
Female sex	59%	50%	0.381
APOE genotype			0.607
Two ε4 alleles	28%	32%	
One ε4 allele	45%	50%	
Solitary living	22%	18%	0.651
Antihypertensives/cardiac therapy	17%	29%	0.181
Antidiabetics	3%	4%	0.981
Asthma medication	8%	0%	0.126
Thyroid therapy	7%	4%	0.508
Lipid-lowering agents	9%	21%	0.055
Estrogens	7%	4%	0.508
NSAIDs/acetylsalicylic acid	10%	14%	0.465
Antidepressants	28%	32%	0.651
Antipsychotics	0%	7%	**0.004**
Anxiolytics/sedatives/hypnotics	3%	11%	0.055
Estimated age at onset, years[a]	58.9 ± 4.4	57.3 ± 5.6	0.112
Age at first assessment (baseline), years[a]	63.1 ± 5.0	61.4 ± 6.6	0.133
Duration of AD, years[a]	4.2 ± 2.9	4.0 ± 5.0	0.866
Education, years[a]	10.0 ± 2.8	10.3 ± 2.8	0.599
ADAS-cog score (range, 0–70)[a]	20.5 ± 9.7	15.5 ± 8.2	**0.013**
IADL score (range, 8–31)[a]	14.3 ± 5.4	11.8 ± 4.6	**0.027**
PSMS score (range, 6–30)[a]	6.7 ± 1.2	6.7 ± 1.5	0.860

[a]Mean ± standard deviation (SD)

AD, Alzheimer's disease; ADAS-cog, Alzheimer's Disease Assessment Scale-cognitive subscale; APOE, apolipoprotein E; IADL, Instrumental Activities of Daily Living scale; NSAIDs, nonsteroidal anti-inflammatory drugs; PSMS, Physical Self-Maintenance Scale.

Table 2. Late-onset AD, baseline characteristics (n = 874)

	Deceased	Still alive	*P* value
Number of patients (n / %)	797 / 91%	77 / 9%	
Female sex	64%	75%	**0.046**
APOE genotype			0.295
Two ε4 alleles	13%	13%	
One ε4 allele	53%	61%	
Solitary living	37%	31%	0.280
Antihypertensives/cardiac therapy	44%	39%	0.357
Antidiabetics	5%	4%	0.602
Asthma medication	4%	3%	0.539
Thyroid therapy	9%	8%	0.768
Lipid-lowering agents	11%	22%	**0.002**
Estrogens	7%	8%	0.736
NSAIDs/acetylsalicylic acid	33%	30%	0.547
Antidepressants	25%	21%	0.415
Antipsychotics	5%	1%	0.124
Anxiolytics/sedatives/hypnotics	16%	19%	0.403
Estimated age at onset, years[a]	74.6 ± 4.9	72.6 ± 4.7	**0.001**
Age at first assessment (baseline), years[a]	77.5 ± 4.7	75.3 ± 4.6	**<0.001**
Duration of AD, years[a]	2.9 ± 1.7	2.7 ± 1.4	0.323
Education, years[a]	9.3 ± 2.4	9.9 ± 2.8	0.075
ADAS-cog score (range, 0–70)[a]	21.5 ± 8.8	15.3 ± 6.1	**<0.001**
IADL score (range, 8–31)[a]	16.6 ± 5.4	13.3 ± 4.2	**<0.001**
PSMS score (range, 6–30)[a]	7.7 ± 2.4	6.7 ± 1.0	**<0.001**

[a]Mean ± standard deviation (SD)

AD, Alzheimer's disease; ADAS-cog, Alzheimer's Disease Assessment Scale-cognitive subscale; APOE, apolipoprotein E; IADL, Instrumental Activities of Daily Living scale; NSAIDs, nonsteroidal anti-inflammatory drugs; PSMS, Physical Self-Maintenance Scale.

Table 3. Cox proportional hazards modelling of time to death, multivariate analyses

	Early-onset AD		Late-onset AD	
Significant predictors	**Hazard ratio (95% CI)**	***P* value**	**Hazard ratio (95% CI)**	***P* value**
Sex (male = 1, female = 0)[a]	na		1.64 (1.41–1.92)	<0.001
Sex by living status[b]			na	
Males living alone	2.71 (1.18–6.22)	0.019		
Females living with family	0.75 (0.48–1.17)	0.207		
Females living alone	1.20 (0.66–2.16)	0.553		
Antihypertensives/cardiac therapy (no = 0, yes = 1)		ns	1.26 (1.09–1.47)	0.002
Antidiabetics (no = 0, yes = 1)		ns	1.51 (1.06–2.14)	0.021
Age at first assessment (baseline), years		ns	1.04 (1.03–1.06)	<0.001
ADAS-cog score at baseline		ns	1.02 (1.01–1.03)	<0.001
IADL score at baseline	1.07 (1.02–1.11)	0.002		ns
PSMS score at baseline		ns	1.05 (1.02–1.09)	0.004
ADAS-cog score, rate of change per year		ns	0.99 (0.98–0.99)	0.004
PSMS score, rate of change per year	0.87 (0.77–0.98)	0.026	0.92 (0.89–0.95)	<0.001

Number of apolipoprotein E ε4 alleles, duration of AD, years of education, IADL score rate of change per year, and specific concomitant medications, with the exception of antihypertensives/cardiac therapy and antidiabetics, were not significant factors in the models.
[a]The interaction effect of sex with solitary living and the variable solitary living were not significant for the late-onset AD group.
[b]Male living with a family member was the reference category.
Hazard ratios are expressed per 1 unit increase for continuous variables and for the condition present for categorized variables.

AD, Alzheimer's disease; ADAS-cog, Alzheimer's Disease Assessment Scale-cognitive subscale; CI, confidence interval; IADL, Instrumental Activities of Daily Living scale; na, not applicable; ns, not significant; PSMS, Physical Self-Maintenance Scale.

Figure 1.

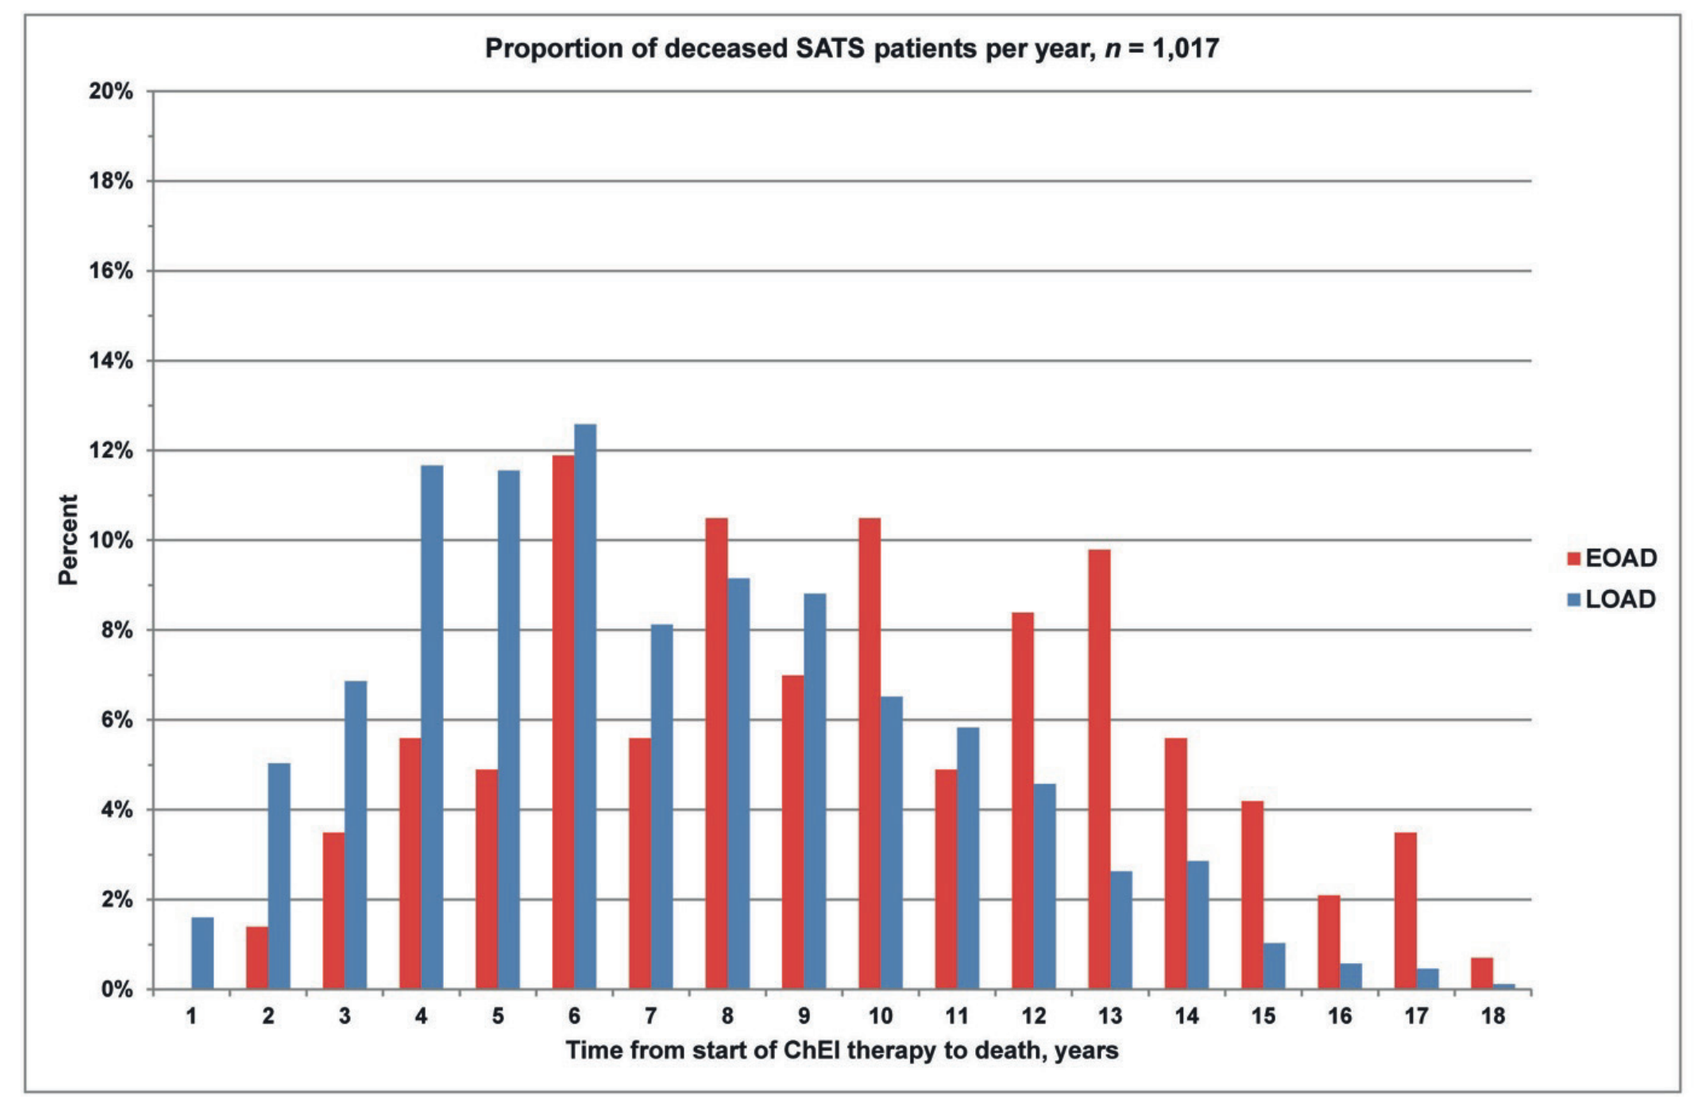

Proportion of deceased SATS patients per year, n = 1,017
Percent
EOAD
LOAD
Time from start of ChEI therapy to death, years

Proportion of deceased participants with EOAD and LOAD per year after the initiation of ChEI treatment (time of AD diagnosis). After 18 years of follow-up, 115 (80%) of the EOAD and 797 (91%) of the LOAD patients had died (P <0.001). The mean ± standard deviation time from diagnosis to death differed between individuals with EOAD and LOAD, 8.0 ± 3.4 years vs. 6.2 ± 3.1 years, (P < 0.001). Year 1 indicates a life-span after baseline of up to 1 year, year 2 indicates >1 to ≤2 years, year 3 indicates >2 to ≤3 years, etc.

AD, Alzheimer's disease; ChEI, cholinesterase inhibitor; EOAD, early-onset Alzheimer's disease; LOAD, late-onset Alzheimer's disease; SATS, Swedish Alzheimer Treatment Study.

Figure 2.

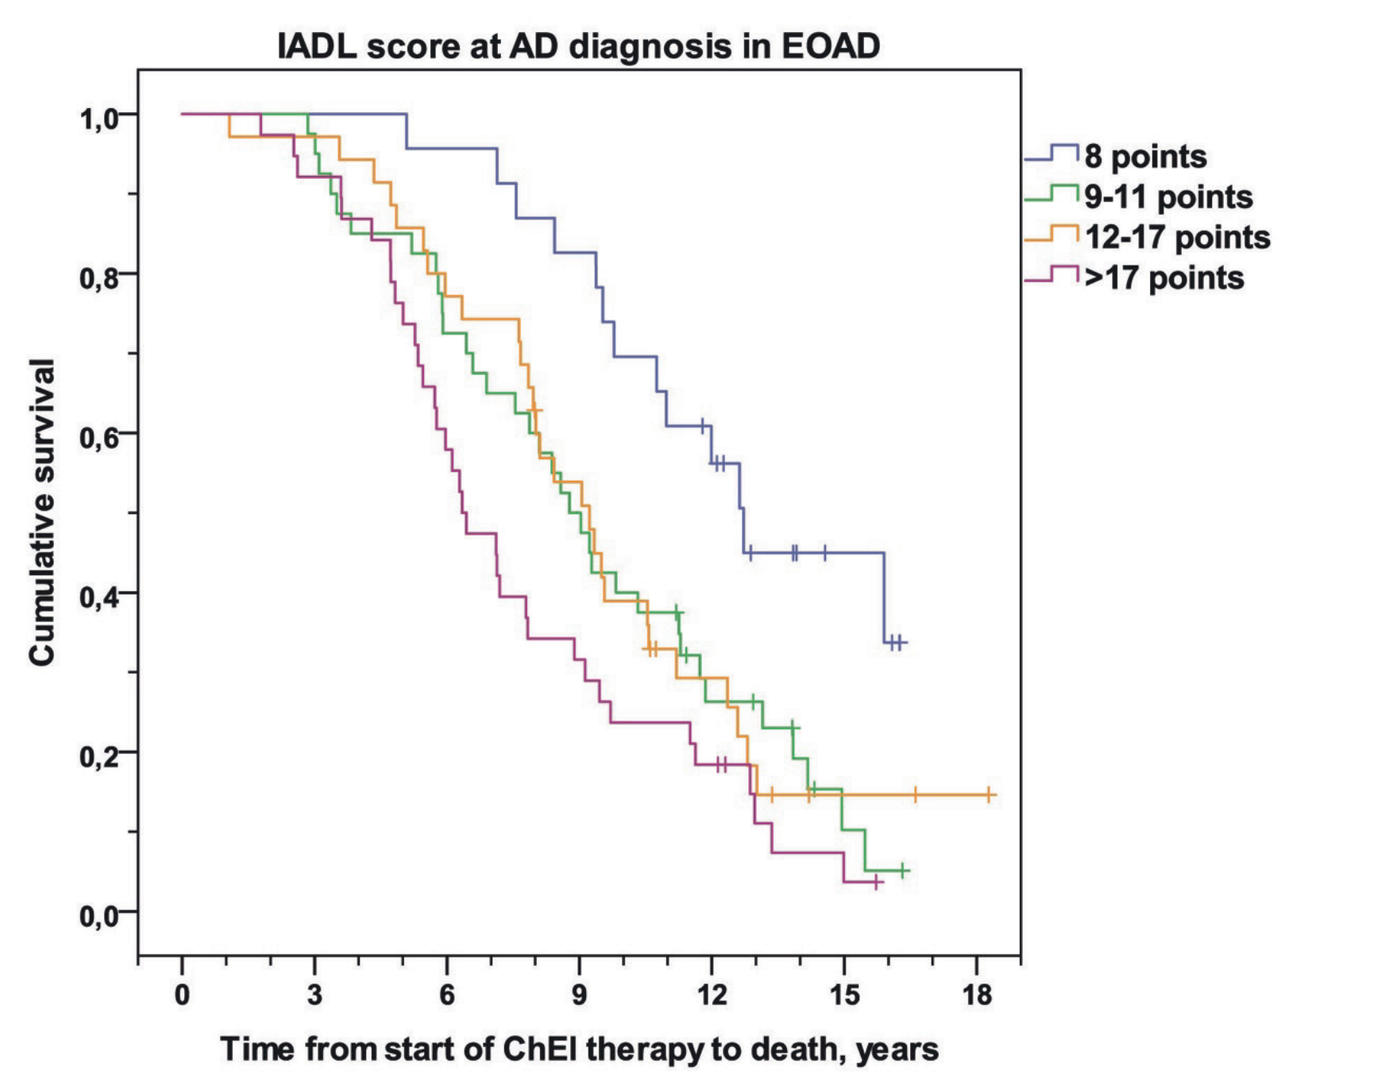

IADL score at AD diagnosis in EOAD
8 points
9-11 points
12-17 points
>17 points
Cumulative survival
Time from start of ChEI therapy to death, years

Kaplan–Meier graph of the distribution of time from the initiation of ChEI therapy (time of AD diagnosis) to death for four EOAD groups based on IADL score at baseline. A log–rank test found significant differences between the groups (P = 0.001).
For the deceased EOAD patients, the mean ± SD time from AD diagnosis to death differed between individuals with varying IADL score at baseline (five had missing data): 8 points (no impairment), (n = 13), 10.1 ± 2.8 years; 9–11 points (n = 34), 8.3 ± 3.6 years; 12–17 points (n = 28), 8.1 ± 3.0 years; and >17 points (n = 35), 7.1 ± 3.3 years, (P = 0.041).

AD, Alzheimer's disease; ChEI, cholinesterase inhibitor; EOAD, early-onset Alzheimer's disease; IADL, Instrumental Activities of Daily Living scale; SD, standard deviation.

Figure 3.

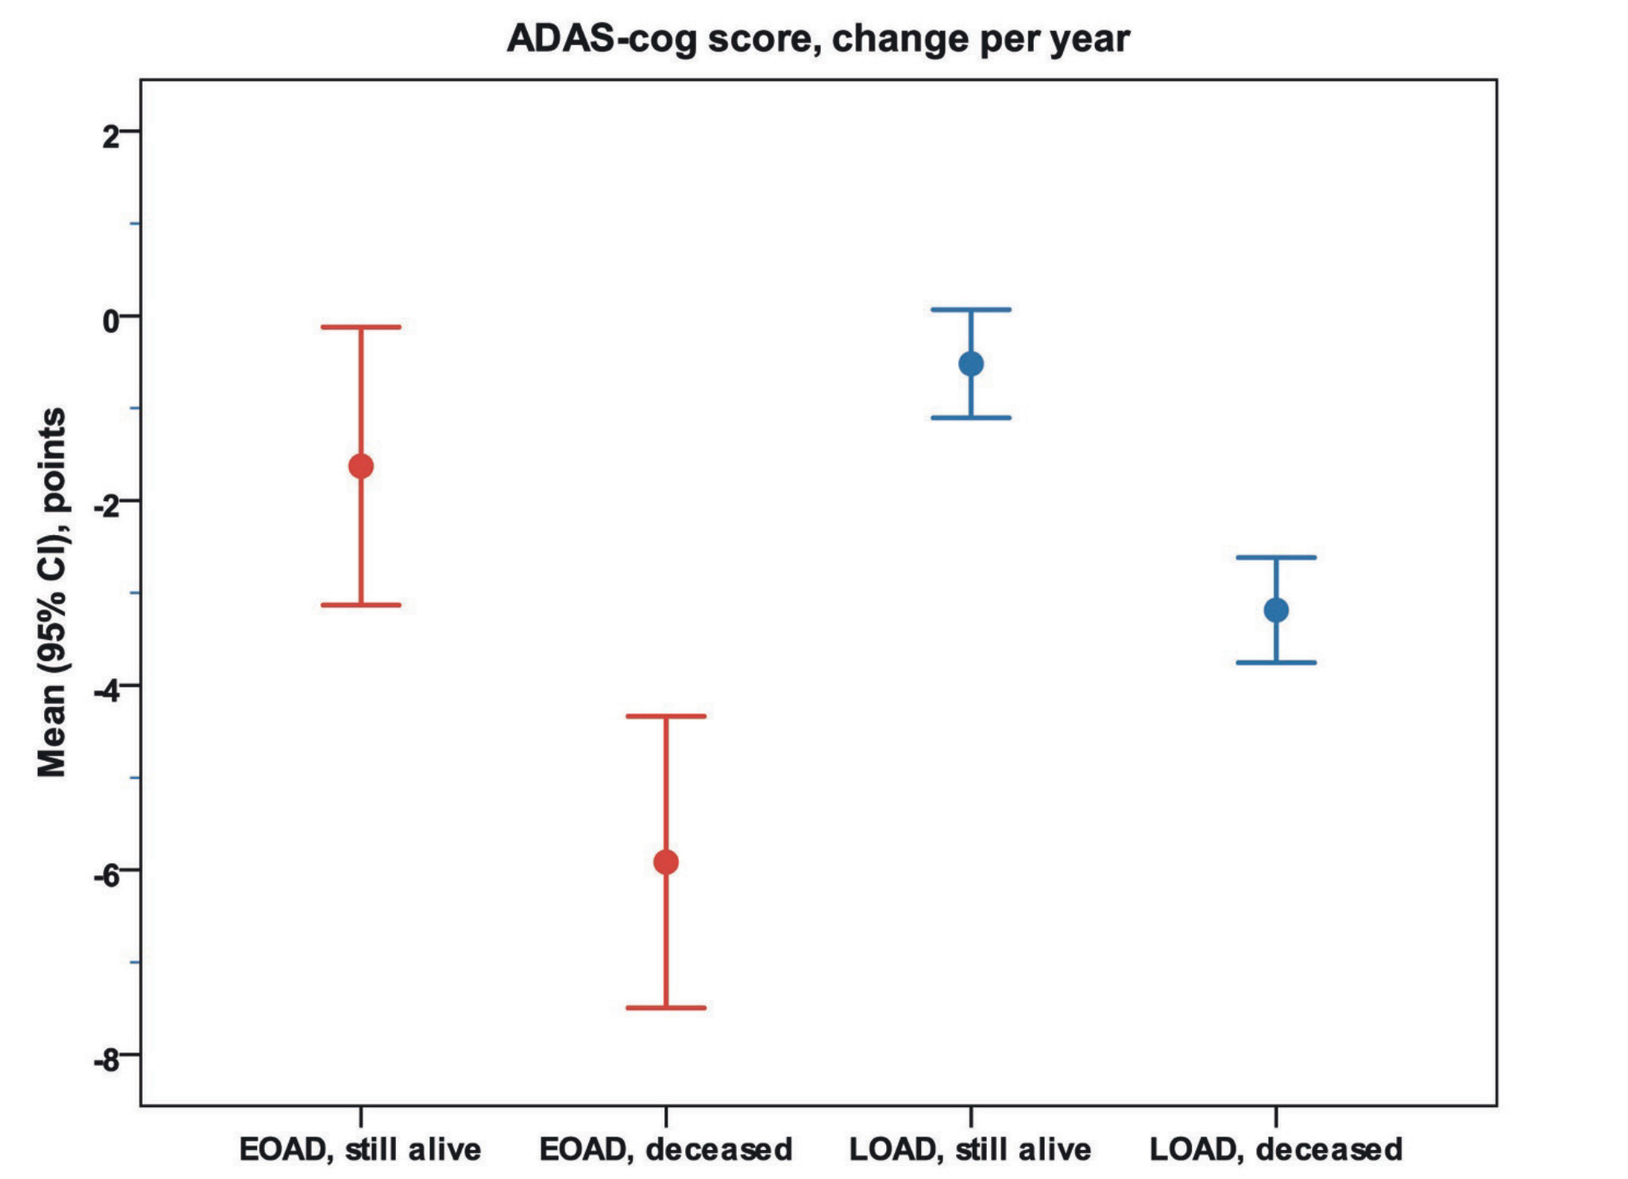

ADAS-cog score, change per year
Mean (95% CI), points
EOAD, still alive
EOAD, deceased
LOAD, still alive
LOAD, deceased

The deceased EOAD patients showed a faster mean (95% CI) annual decline in cognitive ability (ADAS-cog score), −5.9 (−7.5, −4.3) points, compared with the other groups, EOAD still alive, −1.6 (−3.1, −0.1) points; LOAD deceased, −3.2 (−3.8, −2.6) points; and LOAD still alive, −0.5 (−1.1, 0.1) points, (P < 0.001).

ADAS-cog, Alzheimer's Disease Assessment Scale-cognitive subscale; CI, confidence interval; EOAD, early-onset Alzheimer's disease; LOAD, late-onset Alzheimer's disease.

Figure 4.

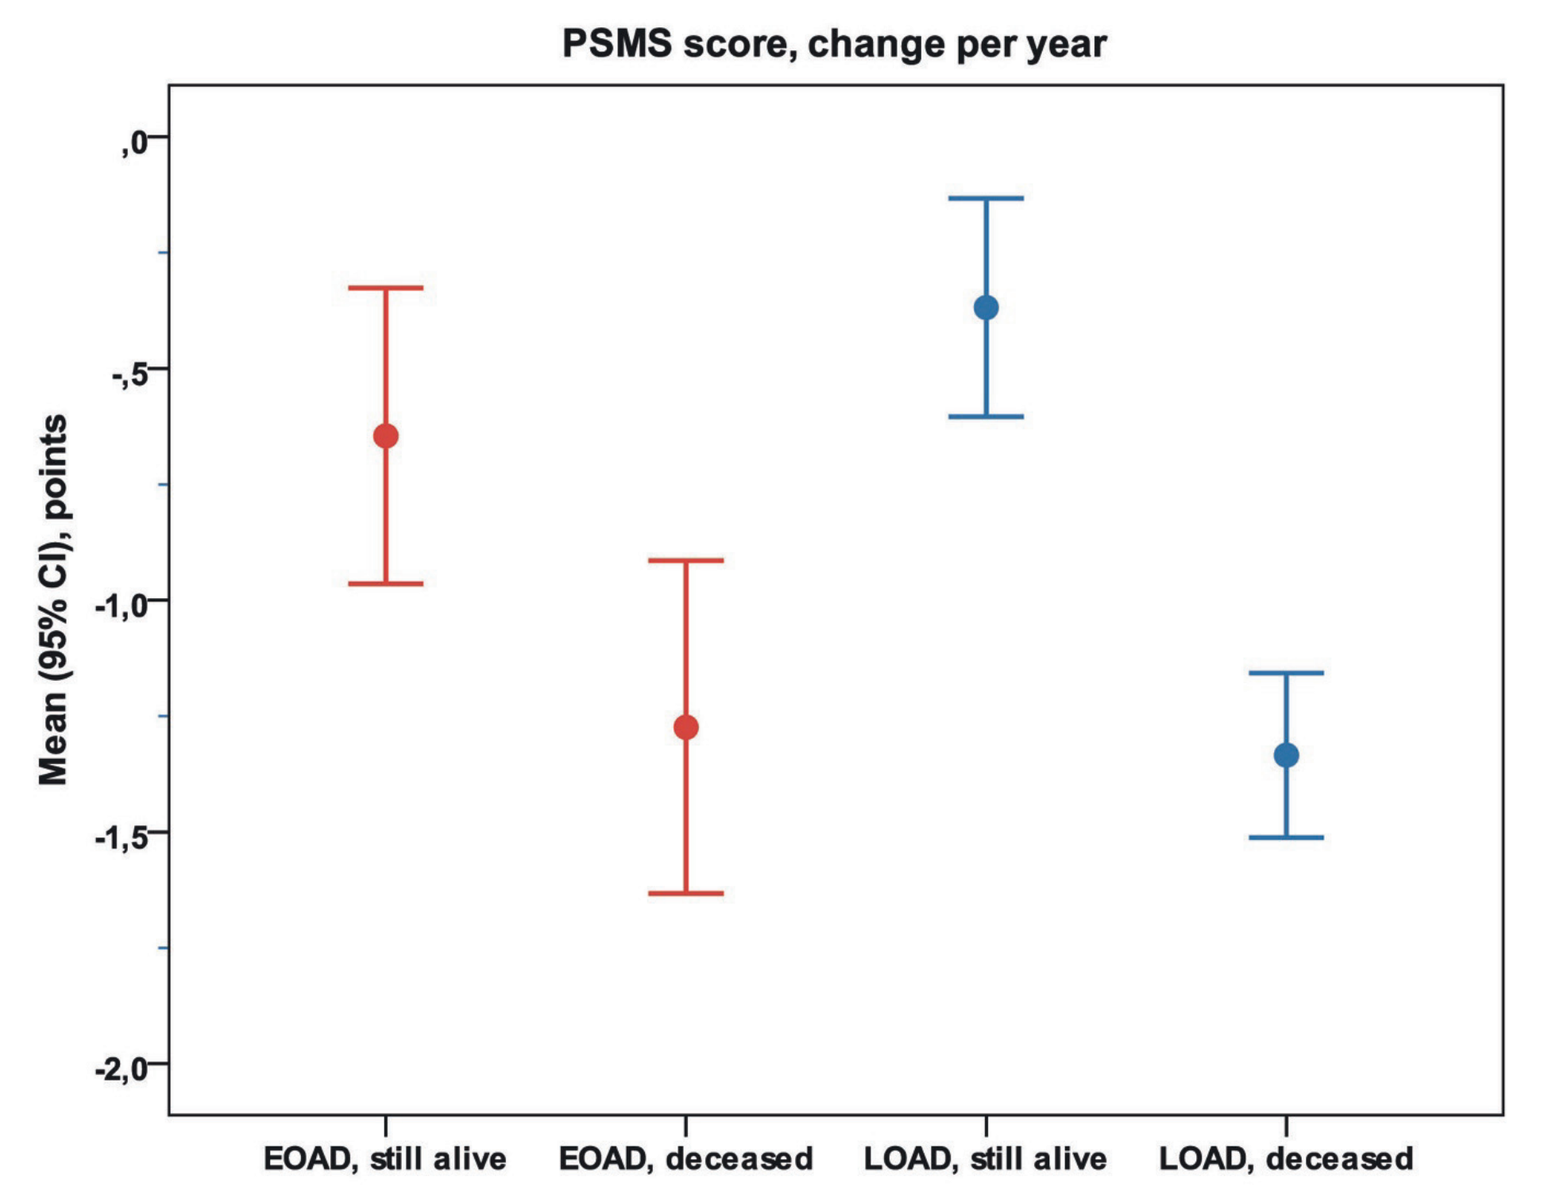

PSMS score, change per year
Mean (95% CI), points
EOAD, still alive
EOAD, deceased
LOAD, still alive
LOAD, deceased

No significant difference in mean (95% CI) basic ADL capacity (PSMS score) change/year was observed in EOAD between the deceased participants −1.3 (−1.6, −0.9) points and those still alive −0.6 (−1.0, −0.3) points. In LOAD, the deceased group exhibited a more rapid mean annual deterioration in basic ADL, −1.3 (−1.5, −1.2) points, compared with the patients still alive, −0.4 (−0.6, −0.1) points, (P = 0.004).

ADL, activities of daily living; CI, confidence interval; EOAD, early-onset Alzheimer's disease; LOAD, late-onset Alzheimer's disease; PSMS, Physical Self-Maintenance Scale.

Layout by Aletia Design, www.aletiadesign.com